STARTVerso4 PHASE III TRIAL OF FALDAPREVIR PLUS PEGYLATED INTERFERON α-2a AND RIBAVIRIN IN PATIENTS WITH HIV AND HCV GENOTYPE-I CO-INFECTION

1099

JK Rockstroh[1], M Nelson[2], V Soriano[3], K Arastéh[4], J Guardiola[5], S Bhagani[6], J Mallolas[7], C Tural[8], M Puoti[9], P Ingiliz[10], M Battegay[11], MK Jain[12], M Núñez[13], K Marks[14], J Kort[15], JO Stern[15], R Vinisko[15], M Manero[16], D Dieterich[17], on behalf of the STARTVerso4 Study Group

[1]University of Bonn, Bonn, Germany; [2]Chelsea and Westminster Hospital, London, UK; [3]Hospital Carlos III, Madrid, Spain; [4]EPIMED, Vivantes Auguste-Viktoria Hospital, Berlin, Germany; [5]Hospital de la Santa Creu i Sant Pau, Barcelona, Spain; [6]Royal Free Hospital, London, UK; [7]Hospital Clínic, Barcelona, Spain; [8]Hospital Universitari Germans Trias i Pujol, Barcelona, Spain; [9]AO Ospedale Niguarda Cà Granda, Milan, Italy; [10]Medizinisches Infektiologiezentrum Berlin (MIB), Berlin, Germany; [11]Division of Infectious Diseases and Hospital Epidemiology, Basel, Switzerland; [12]UT Southwestern Medical Center, Dallas, TX, USA; [13]Wake Forest University, Winston-Salem, NC, USA; [14]Weill Cornell Medical College, New York, NY, USA; [15]Boehringer Ingelheim Pharmaceuticals, Inc., Ridgefield, CT, USA; [16]Boehringer Ingelheim España S.A., Barcelona, Spain; [17]Mount Sinai School of Medicine, New York, NY, USA

Jürgen K. Rockstroh
juergen.rockstroh@ukb.uni-bonn.de

BACKGROUND

- Faldaprevir (FDV) is a potent inhibitor of HCV NS3/4A.[1]
- FDV has antiviral activity against HCV genotypes (GT) 1, 4, 5, and 6 in vitro.[1]
- FDV pharmacokinetics support oral, once-daily administration.[2]
- Three Phase III trials of FDV + pegylated interferon α-2a and ribavirin (PR) in HCV GT-1 are complete.
 - In STARTVerso1, FDV + PR resulted in sustained virologic response 12 weeks after completion of treatment (SVR12) rates of 79% (FDV 120 mg QD) and 80% (FDV 240 mg QD) in treatment-naïve patients with chronic HCV GT-1 infection.[3]
- FDV is also being investigated in Phase III interferon-free trials.
- STARTVerso4 (NCT01399619) is an ongoing Phase III trial evaluating the safety and efficacy of FDV + PR in patients co-infected with HCV and HIV.

METHODS

STUDY DESIGN

- Multicenter, open-label, sponsor-blinded, Phase III study in patients co-infected with HCV GT-1 and HIV-1 (N=310; **Figures 1a** and **1b**).
- Patients randomized (1:1) or assigned to:
 - FDV 120 mg QD combined with PR for 24 weeks
 - FDV 240 mg QD + PR for 12 weeks followed by re-randomization to continue to week 24 or stop FDV and continue PR alone through week 24.
- At week 24, patients who achieved early treatment success (ETS; HCV RNA <25 IU/mL at week 4 and undetectable at week 8) were randomized to an additional 24 weeks of PR or to stop all treatment.
 - Patients who did not achieve ETS received PR through week 48.
- Randomizations (day 1, week 12, and week 24) were stratified by HCV GT-1 subtype (1a, 1b, and non 1a/1b).

FIGURE 1a. STARTVerso4 study design

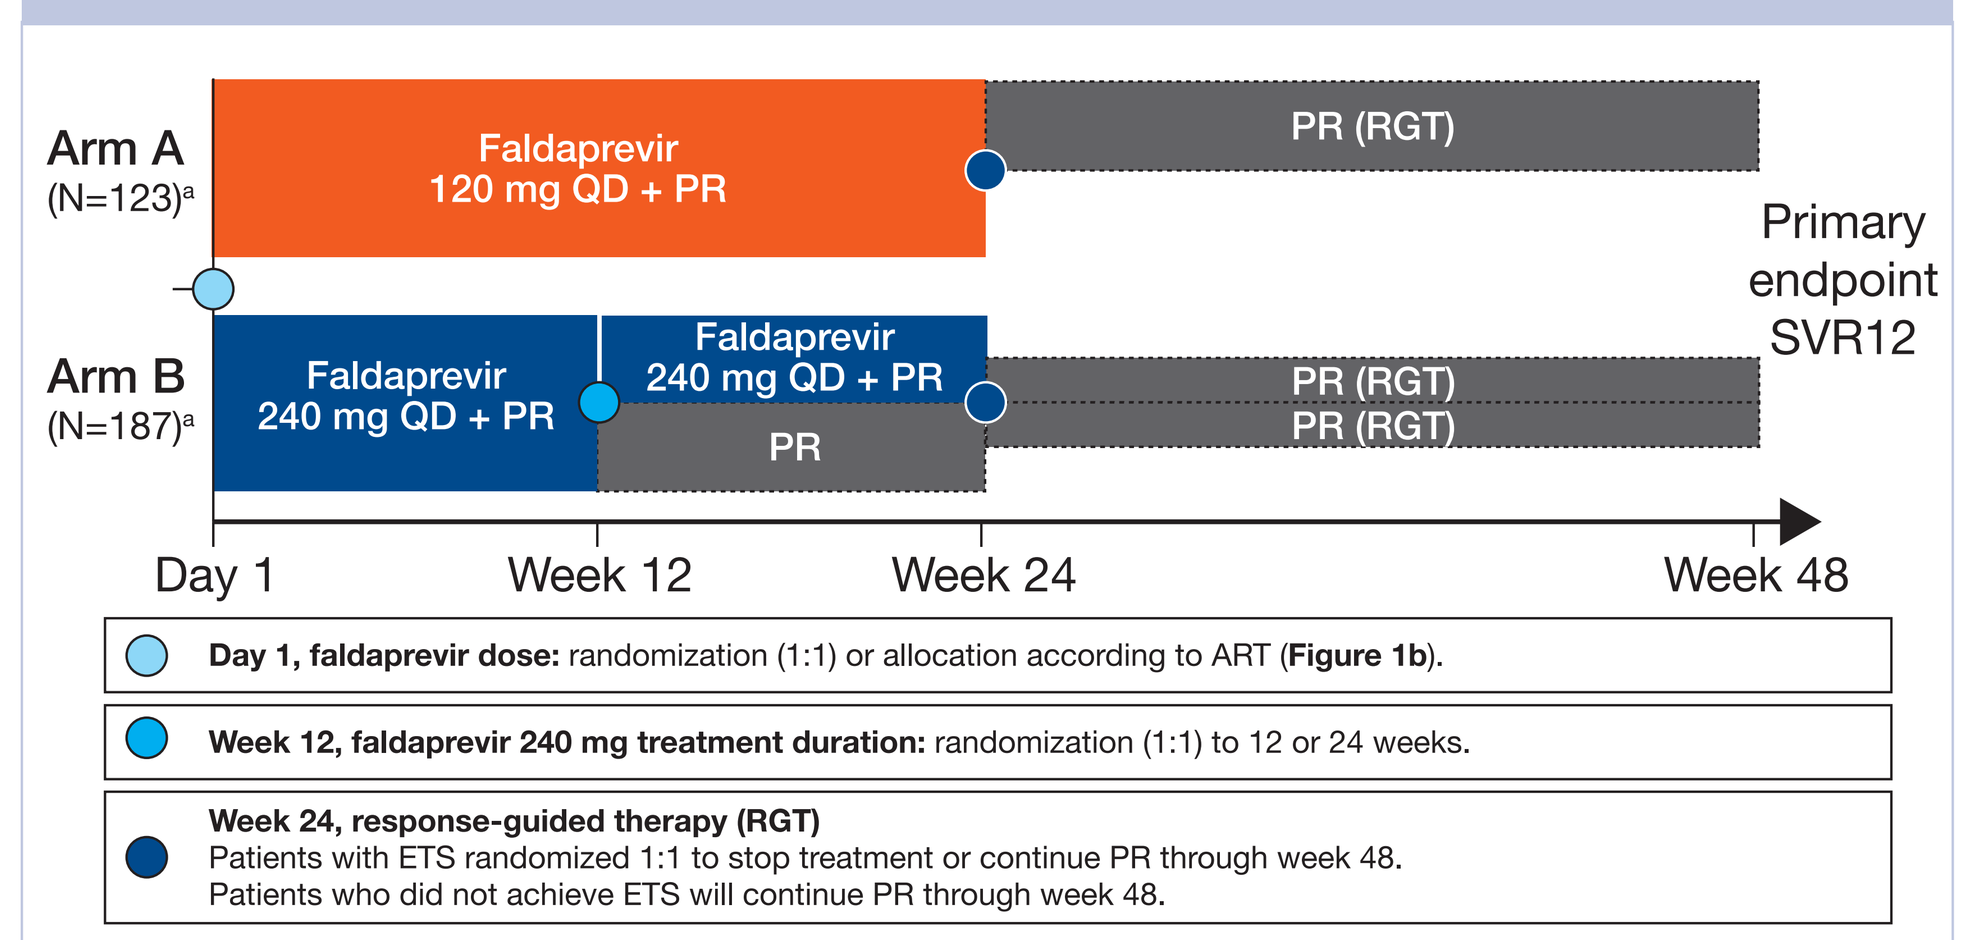

Arm A
(N=123)a
Faldaprevir 120 mg QD + PR
PR (RGT)
Arm B
(N=187)a
Faldaprevir 240 mg QD + PR
Faldaprevir 240 mg QD + PR
PR
PR (RGT)
PR (RGT)
Primary endpoint SVR12
Day 1
Week 12
Week 24
Week 48
Day 1, faldaprevir dose: randomization (1:1) or allocation according to ART (Figure 1b).
Week 12, faldaprevir 240 mg treatment duration: randomization (1:1) to 12 or 24 weeks.
Week 24, response-guided therapy (RGT)
Patients with ETS randomized 1:1 to stop treatment or continue PR through week 48.
Patients who did not achieve ETS will continue PR through week 48.

[a]Number randomized/allocated.
ART, antiretroviral therapy; ETS, early treatment success (HCV RNA <25 IU/mL at week 4 and undetectable at week 8); PR, pegylated interferon α-2a and ribavirin; QD, once daily; SVR12, sustained virologic response (HCV RNA <25 IU/mL, not detected) 12 weeks after the planned end of treatment.

FIGURE 1b. Randomization/allocation to treatment arms according to antiretroviral therapy (ART)

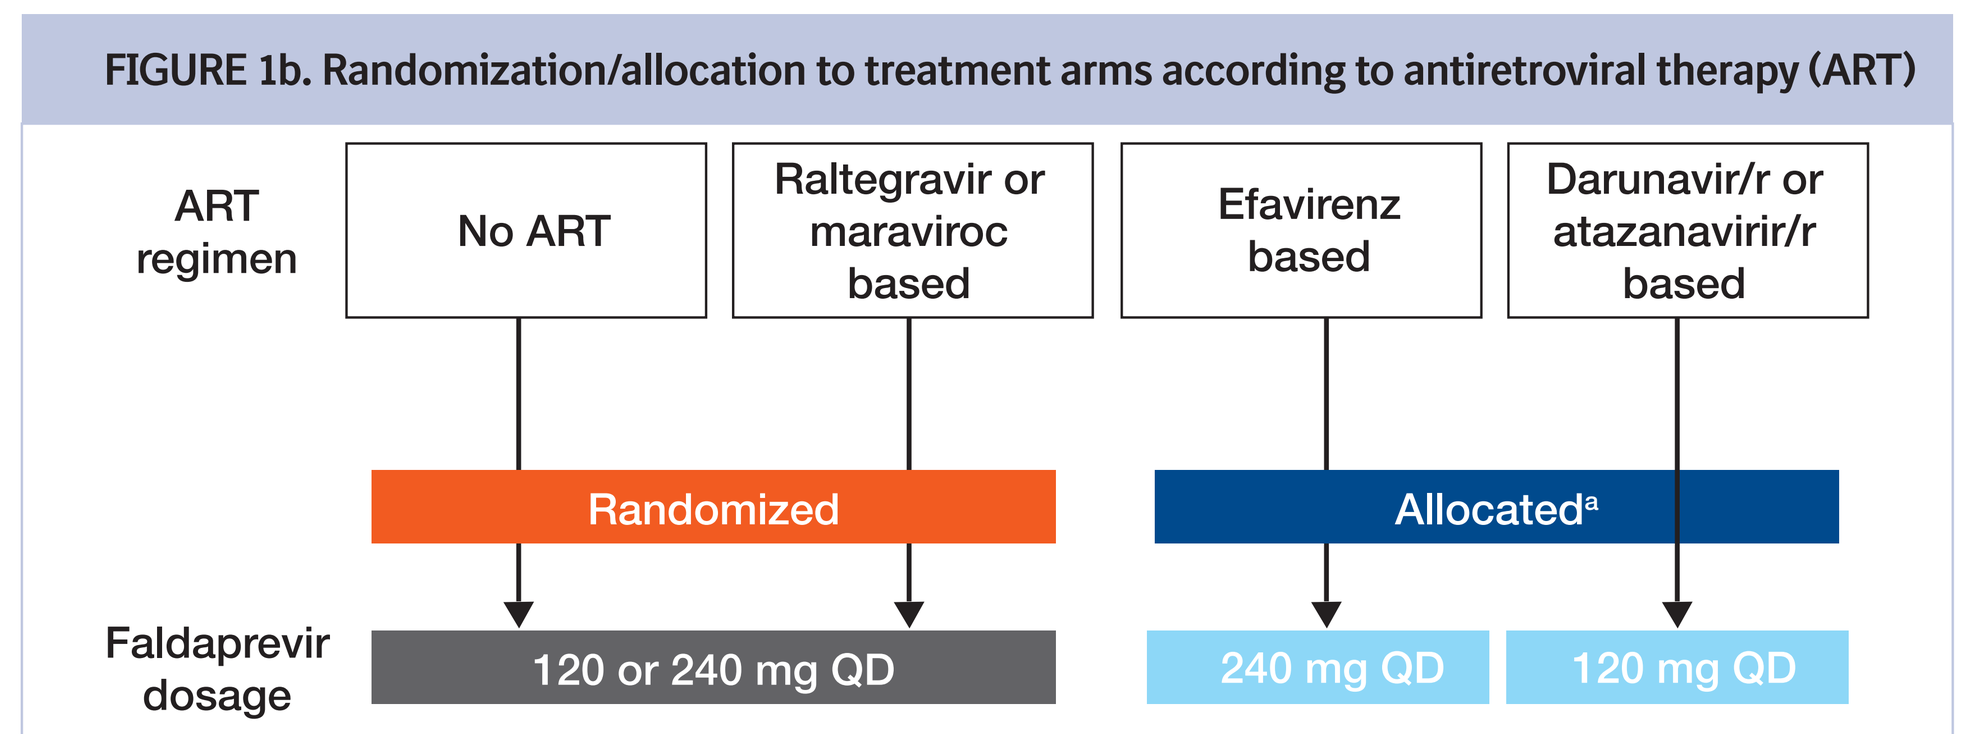

ART regimen
No ART
Raltegravir or maraviroc based
Efavirenz based
Darunavir/r or atazanavirir/r based
Randomized
Allocateda
Faldaprevir dosage
120 or 240 mg QD
240 mg QD
120 mg QD

[a]Based on results of drug interaction studies.[4]
ART, antiretroviral therapy; QD, once daily; r, ritonavir.

KEY INCLUSION CRITERIA

Chronic infection with HCV GT-1 at screening

- HCV RNA ≥1,000 IU/mL at screening.
- Naïve to treatment with interferon and PR, or prior relapser.
 - Prior relapse: Undetectable HCV RNA at the end of treatment with a PR-based regimen, but HCV RNA detectable within 24 weeks of treatment follow-up.
- Liver biopsy <3 years or Fibroscan <6 months prior to randomization.
- Patients with HCV-related compensated cirrhosis.

HIV-1 infection

- Antiretroviral therapy (ART)-naïve or stable on ART.
- For ART-naïve patients: Peripheral CD4 T-cell count ≥500 cells/mm^3 at screening, and HIV-1 plasma RNA <100,000 c/mL.
- For patients on ART: On the same ART regimen for ≥6 weeks prior to randomization, HIV RNA <40 c/mL at screening, and <50 c/mL for ≥6 months prior to screening.
- Karnofsky score >70.
- No AIDS-defining illness during 6 months prior to screening.

ANALYSES

- This is an interim analysis of a fully enrolled ongoing study.
- Efficacy analysis:
 - ETS (HCV RNA <25 IU/mL at week 4 and undetectable at week 8)
 - SVR (undetectable HCV RNA) 4 weeks after the planned end of treatment (SVR4).
- Safety assessment:
 - Occurrence of adverse events (AEs) overall and by NIH Division of AIDS (DAIDS) grades, AEs leading to treatment discontinuation and serious AEs (SAEs)
 - HIV virologic failure (HIV RNA >200 copies/mL on 2 consecutive visits) from <40 c/mL or undetectable HIV RNA.

RESULTS

PATIENTS

- Of 310 randomized patients, 308 received at least 1 dose of study medication and were evaluable for SVR4 (**Figure 2**).
- Overall, demographic and baseline disease characteristics were balanced across the treatment groups (**Tables 1–3**).

FIGURE 2. Patient disposition

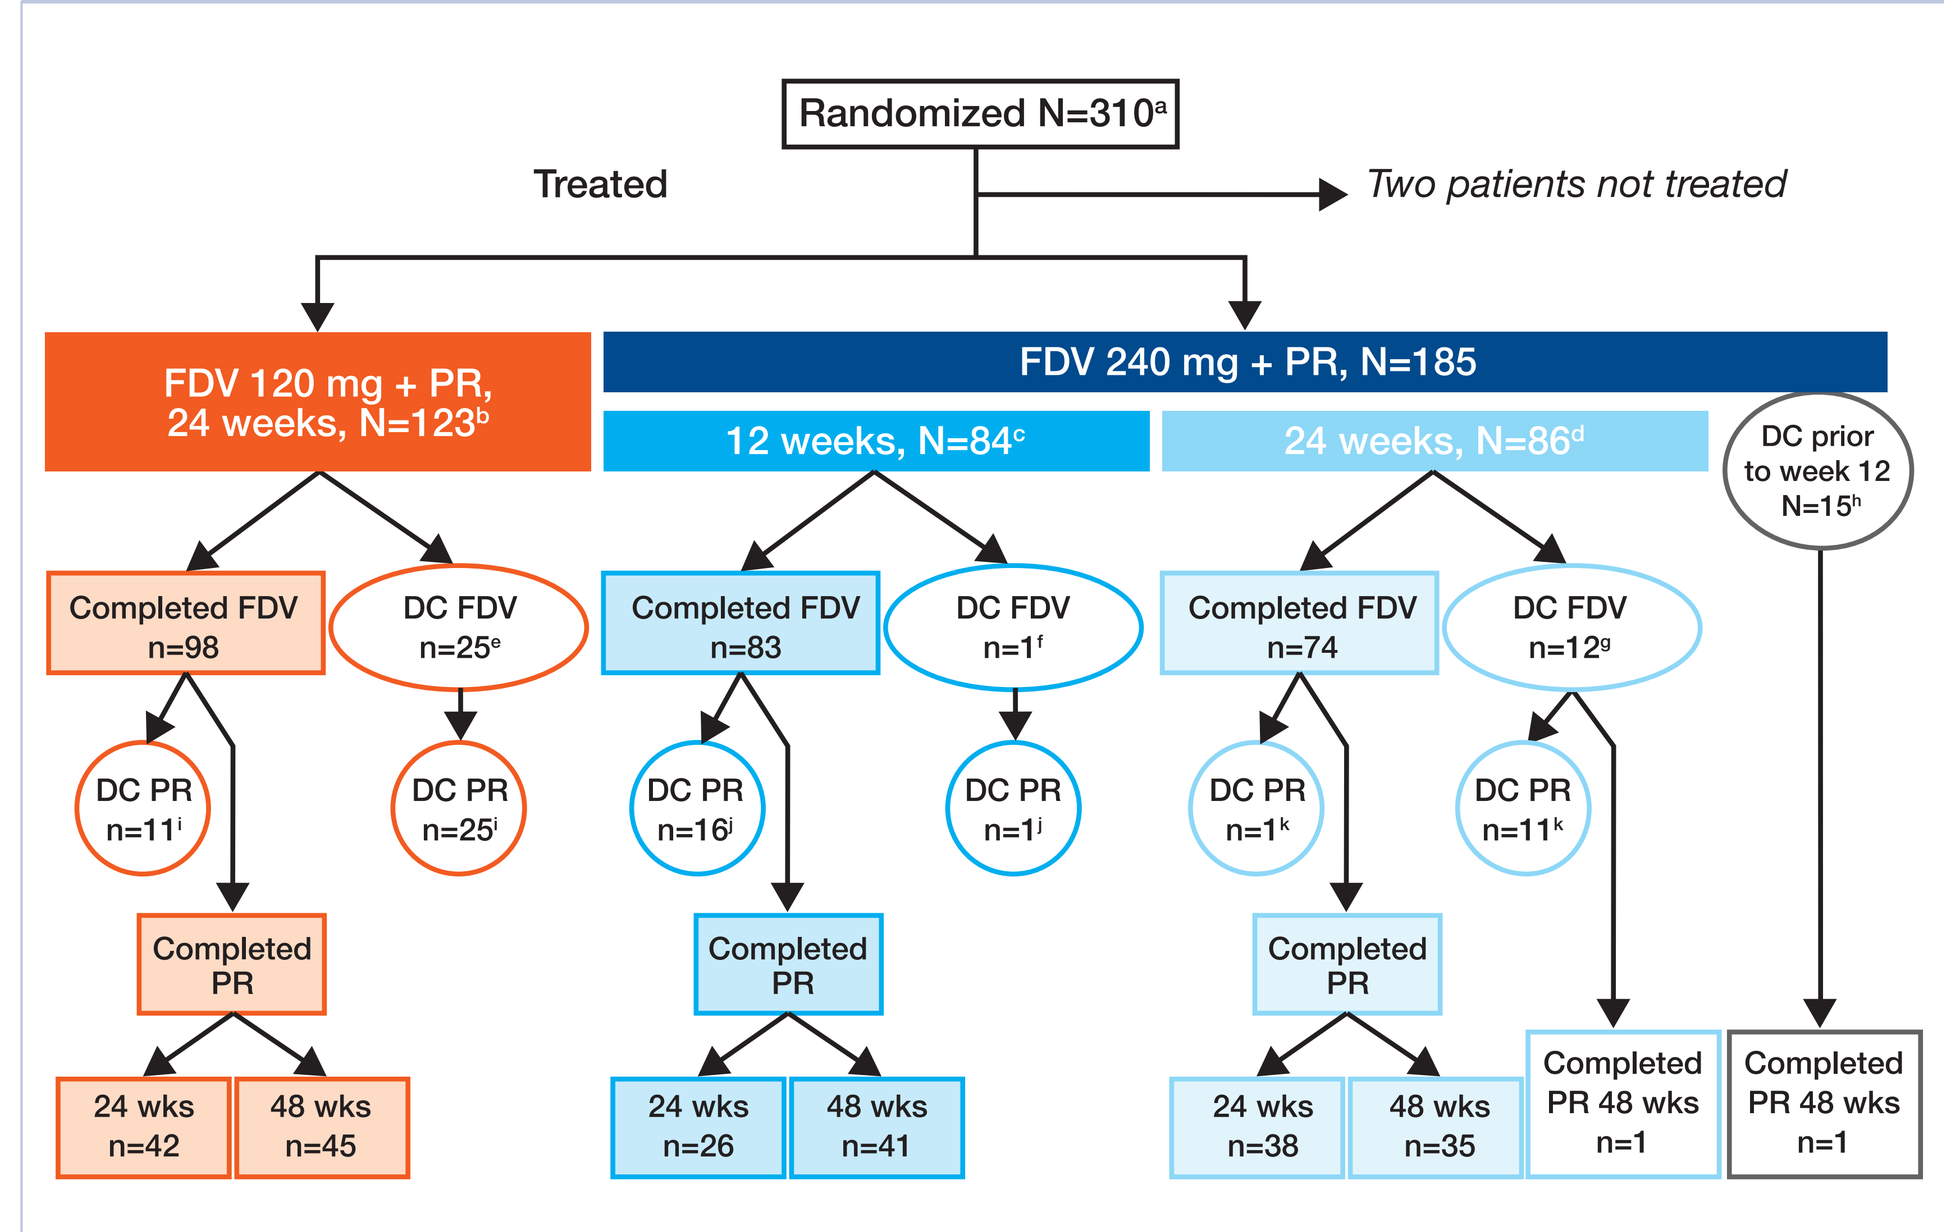

Randomized N=310a
Treated
Two patients not treated
FDV 120 mg + PR, 24 weeks, N=123b
FDV 240 mg + PR, N=185
12 weeks, N=84c
24 weeks, N=86d
DC prior to week 12 N=15h
Completed FDV n=98
DC FDV n=25e
DC PR n=11i
DC PR n=25i
Completed PR
24 wks n=42
48 wks n=45
Completed FDV n=83
DC FDV n=1f
DC PR n=16j
DC PR n=1j
Completed PR
24 wks n=26
48 wks n=41
Completed FDV n=74
DC FDV n=12g
DC PR n=1k
DC PR n=11k
Completed PR
24 wks n=38
48 wks n=35
Completed PR 48 wks n=1
Completed PR 48 wks n=1

DC, discontinued; FDV, faldaprevir; PR, pegylated interferon α-2a and ribavirin; wks, weeks.
[a]Disposition as of 7[th] October 2013; [b]Treatment-naïve (100), relapser (23); [c]Treatment-naïve (62), relapser (22); [d]Treatment-naïve (63), relapser (23); [e]Reasons: adverse event (10), lack of efficacy (9), withdrawal by subject (6); [f]Reasons: lack of efficacy (1); [g]Reasons: adverse event (4), lack of efficacy (5), withdrawal by subject (3); [h]Reasons: adverse event (10), protocol violation (1), withdrawal by subject (4); [i]Reasons: adverse event (9), lack of efficacy (13), lost to follow-up (1), withdrawal by subject (12), reason not stated (1); [j]Reasons: adverse event (3), lack of efficacy (8), protocol violation (1), withdrawal by subject (4), other (1); [k]Reasons: adverse event (4), lack of efficacy (5), withdrawal by subject (2), other (1).

TABLE 1. Baseline demographics

	FDV 120 mg + PR 24 weeks (N=123)	FDV 240 mg + PR 12 weeks (N=84)	FDV 240 mg + PR 24 weeks (N=86)	FDV 240 mg + PR Total (N=185)[a]	Total (N=308)
Male, n (%)	103 (84)	66 (79)	68 (79)	145 (78)	248 (81)
Race, n (%)					
White	106 (86)	69 (82)	75 (87)	150 (81)	256 (83)
Black or African American	12 (10)	12 (14)	11 (13)	30 (16)	42 (14)
Asian	3 (2)	2 (2)	0	4 (2)	7 (2)
Other[b]	2 (2)	1 (1)	0	1 (1)	3 (1)
Region, n (%)					
Europe	98 (80)	58 (69)	61 (71)	127 (69)	225 (73)
North America	18 (15)	24 (29)	20 (23)	51 (28)	69 (22)
Brazil	7 (6)	2 (2)	5 (6)	7 (4)	14 (5)
Mean age, years (SD)	47.6 (7.63)	46.1 (8.64)	46.0 (7.97)	46.5 (8.36)	46.9 (8.08)
Mean BMI, kg/m^2 (SD)	24.5 (4.32)	25.2 (4.23)	24.9 (3.94)	24.9 (4.03)	24.8 (4.15)
IL28B (rs12979860), n (%)					
CC	47 (38)	29 (35)	19 (22)	53 (29)	100 (32)
CT	57 (46)	43 (51)	44 (51)	94 (51)	151 (49)
TT	17 (14)	11 (13)	20 (23)	34 (18)	51 (17)
Missing	2 (2)	1 (1)	3 (3)	4 (2)	6 (2)

[a]Includes 15 patients from 240 mg treatment group who discontinued prior to week 12.
[b]Other race includes American Indian or Alaska Native, Native Hawaiian or other Pacific Islander, and missing data.

TABLE 2. HCV-specific baseline data

	FDV 120 mg + PR 24 weeks (N=123)	FDV 240 mg + PR 12 weeks (N=84)	FDV 240 mg + PR 24 weeks (N=86)	FDV 240 mg + PR Total (N=185)[a]	Total (N=308)
Fibrosis stage[b], n (%)					
F0–F2	76 (62)	62 (74)	63 (73)	138 (75)	214 (69)
F3/F4	47 (38)	20 (24)	22 (26)	43 (23)	90 (29)
Missing	0	2 (2)	1 (1)	4 (2)	4 (1)
Cirrhosis[c], n (%)					
Yes	24 (20)	10 (12)	11 (13)	21 (11)	45 (15)
No	99 (80)	73 (87)	75 (87)	162 (88)	261 (85)
HCV GT-1 subtype[d], n (%)					
1a	94 (76)	68 (81)	72 (84)	148 (80)	242 (79)
1b	29 (24)	16 (19)	14 (16)	37 (20)	66 (21)
Mean baseline HCV RNA, $\log_{10}$ IU/mL (SD)	6.5 (0.68)	6.4 (0.65)	6.3 (0.67)	6.4 (0.66)	6.4 (0.67)
Baseline HCV RNA, n (%) ≥800,000 IU/mL	104 (85)	67 (80)	68 (79)	148 (80)	252 (82)
Previous treatment, n (%)					
Treatment-naïve	100 (81)	62 (74)	63 (73)	139 (75)	239 (78)
Relapser	23 (19)	22 (27)	23 (27)	46 (25)	69 (22)

[a]Includes 15 patients from 240 mg treatment group who discontinued prior to week 12. [b]If no Metavir, then Fibroscan result was used (<9.5 F0–F2, ≥9.5 F3/F4). If none was available and patient was indicated to have cirrhosis, then F3/F4 was recorded. [c]Cirrhosis was determined by the investigator based on Fibroscan, biopsy, and/or other clinical parameters. [d]HCV GT-1 subtype analyses by sequencing of NS3.

TABLE 3. HIV-specific baseline data

	FDV 120 mg + PR 24 weeks (N=123)	FDV 240 mg + PR 12 weeks (N=84)	FDV 240 mg + PR 24 weeks (N=86)	FDV 240 mg + PR Total (N=185)[a]	Total (N=308)
ART regimen, n (%)					
ATV/r-based	12 (10)	0	0	0	12 (4)
DRV/r-based	54 (44)	1 (1)[b]	0	1 (1)	55 (18)
EFV-based	2 (2)	37 (44)	39 (45)	82 (44)	84 (27)
Raltegravir-based	49 (40)	43 (51)	42 (49)	94 (51)	143 (46)
Other ART regimen	0	2 (2)	1 (1)	3 (2)	3 (1)
ART-naïve	6 (5)	1 (1)	4 (5)	5 (3)	11 (4)
Prior AIDS-defined illness, n (%)					
0	94 (76)	71 (85)	73 (85)	157 (85)	251 (81)
1	17 (14)	12 (14)	10 (12)	23 (12)	40 (13)
>1	12 (10)	1 (1)	3 (3)	5 (3)	17 (6)
HIV RNA (copies/mL), n (%)					
Not detected	82 (67)	69 (82)	62 (72)	145 (78)	227 (74)
<40	33 (27)	11 (13)	17 (20)	29 (16)	62 (20)
≥40	8 (7)	4 (5)	7 (8)	11 (6)	19 (6)
Mean CD4+ cell count, cells/mm^3 (SD)	520 (234.7)	522 (211.5)	587 (248.6)	549 (231.7)	537 (233.0)

[a]Includes additional patients from 240 mg treatment group who discontinued prior to week 12.
[b]One patient taking DRV/r was randomized in error and received FDV 240 mg for 12 weeks before being randomized to stop FDV at week 12.

EFFICACY

- SVR4 was achieved by 74% of patients overall (**Figure 3**).
 - SVR4 rates were comparable across FDV doses and durations of treatment.
- The most frequent reason for not achieving SVR4 was on-treatment breakthrough (**Table 4**).
- SVR4 rates were high in all patient subgroups, regardless of HCV genotype, IL28B genotype, presence of compensated cirrhosis, and ART regimen (**Figures 4a–e**).
- ETS was achieved by 80% of patients (**Figure 5a**) and 88% of these patients achieved SVR4 (**Figure 5b**).
 - Among patients with ETS, SVR4 rates were comparable at both FDV doses and in patients randomized to 24 or 48 weeks of PR (**Figure 5c**).

FIGURE 3. SVR4 overall population

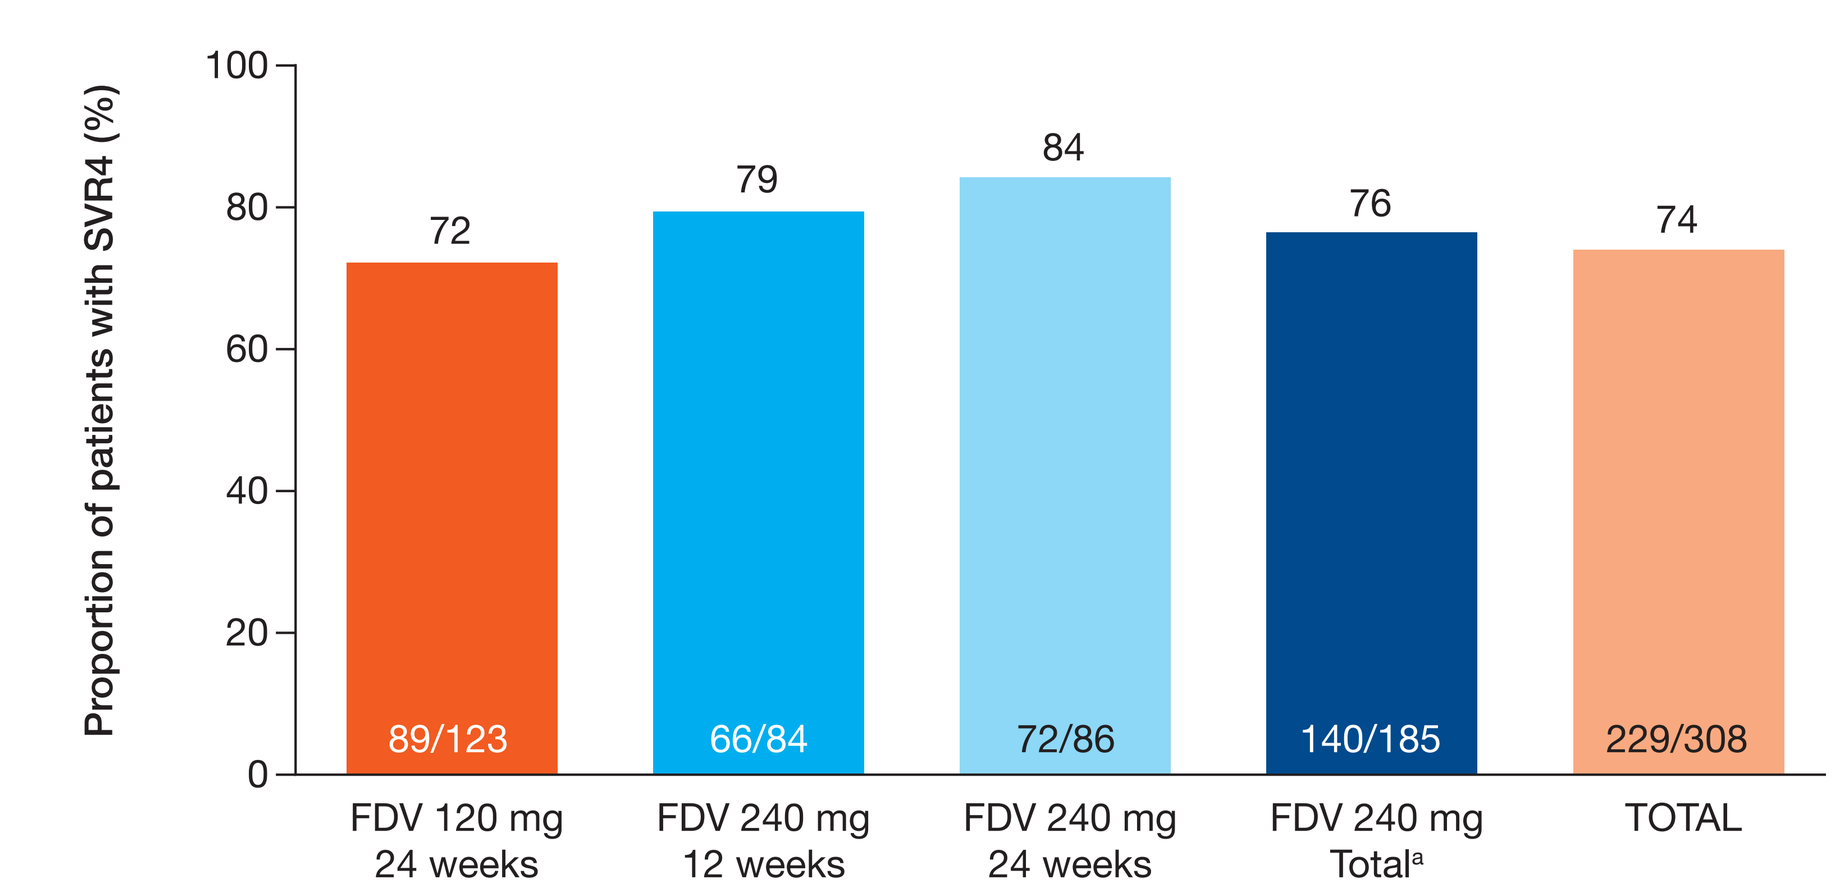

Proportion of patients with SVR4 (%)
100
80
60
40
20
0
72
79
84
76
74
89/123
66/84
72/86
140/185
229/308
FDV 120 mg 24 weeks
FDV 240 mg 12 weeks
FDV 240 mg 24 weeks
FDV 240 mg Totala
TOTAL

[a]Includes additional patients from 240 mg treatment group who discontinued prior to week 12.

TABLE 4. Summary of reasons for not achieving SVR4

n (%)	FDV 120 mg + PR 24 weeks (N=123)	FDV 240 mg + PR 12 weeks (N=84)	FDV 240 mg + PR 24 weeks (N=86)	FDV 240 mg + PR Total (N=185)[a]	Total (N=308)
Without SVR4	34 (28)	18 (21)	14 (16)	45 (24)	79 (26)
Non-response[b]	10 (8)	2 (2)	2 (2)	11 (6)	21 (7)
Breakthrough[c]	11 (9)	8 (10)	7 (8)	15 (8)	26 (8)
FDV breakthrough	9 (7)	1 (1)	5 (6)	6 (3)	15 (5)
0–12 weeks	2 (2)	1 (1)	3 (3)	4 (2)	6 (2)
>12–24 weeks	7 (6)	0	2 (2)	2 (1)	9 (3)
PR breakthrough	2 (2)	7 (8)	2 (2)	9 (5)	11 (4)
0–12 weeks	0	1 (1)	0	1 (<1)	1 (<1)
>12–24 weeks	0	6 (7)	0	6 (3)	6 (2)
>24 weeks	2 (2)	0	2 (2)	2 (1)	4 (1)
Relapse[d]	4 (3)	6 (7)	2 (2)	8 (4)	12 (4)
Discontinued at nadir (responders)	9 (7)	2 (2)	2 (2)	9 (5)	18 (6)
Other	0	0	1 (1)	2 (1)	2 (1)

[a]Includes 15 patients from 240 mg treatment group who discontinued prior to week 12.
[b]Non-response, failure to achieve undetectable HCV RNA over 24 weeks of treatment.
[c]Breakthrough, confirmed ≥1 $\log_{10}$ rebound at any time during FDV or PR treatment.
[d]Relapse, HCV RNA rebound after the end of treatment.

FIGURE 4a. SVR4 by HCV genotype (total population)

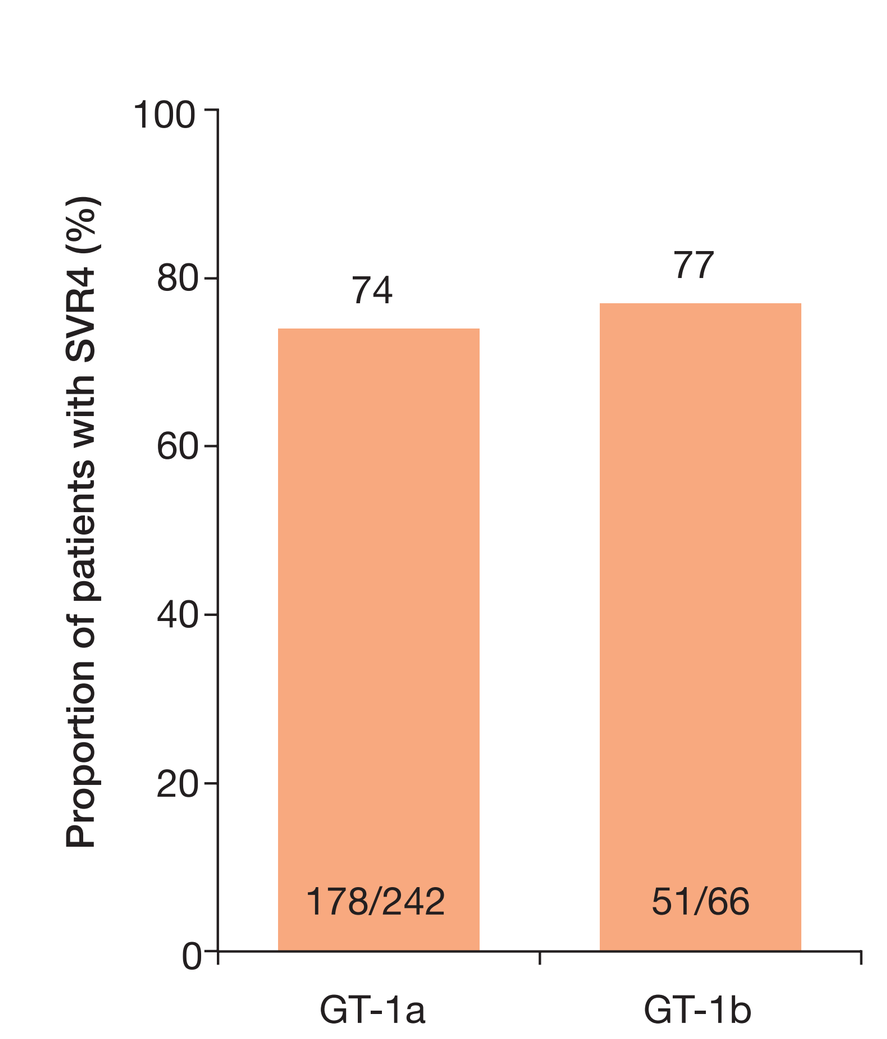

Proportion of patients with SVR4 (%)
100
80
60
40
20
0
74
77
178/242
51/66
GT-1a
GT-1b

FIGURE 4b. SVR4 by IL28B (total population)

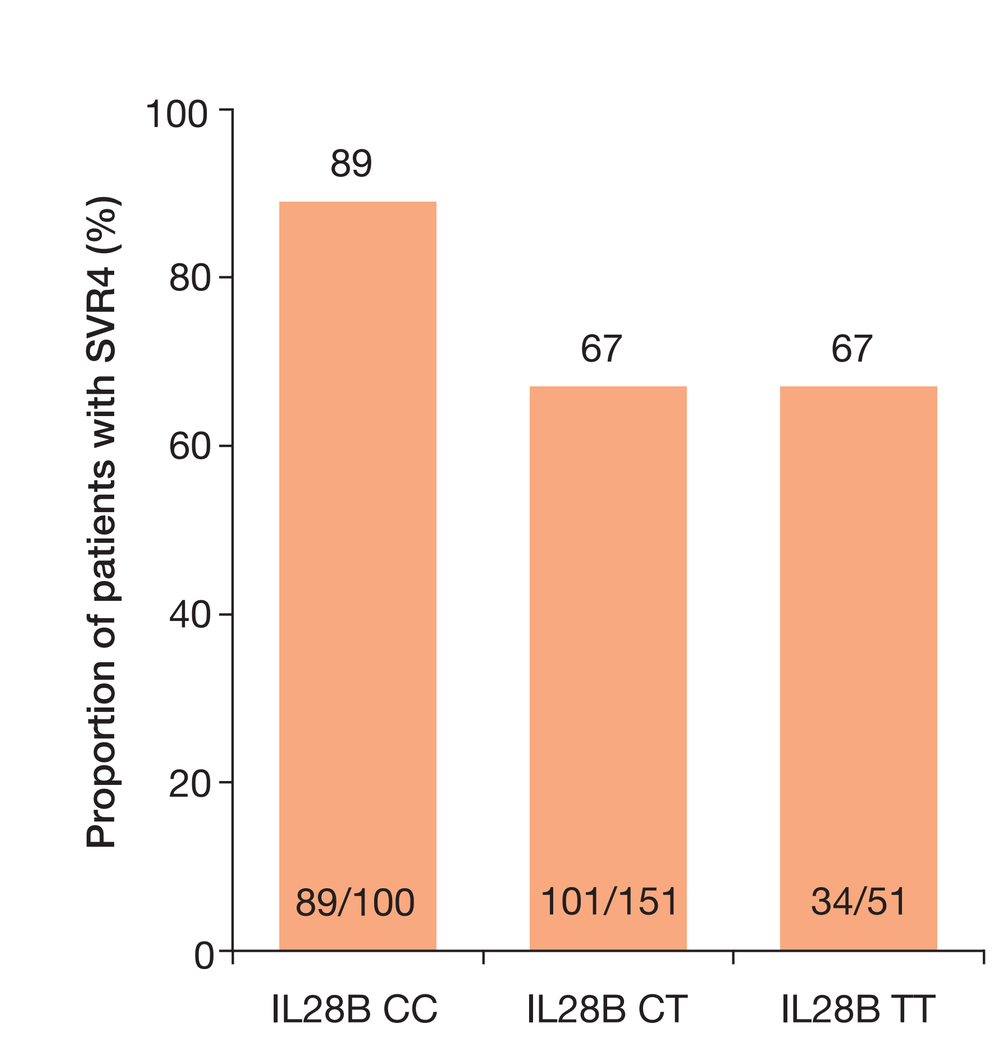

Proportion of patients with SVR4 (%)
100
80
60
40
20
0
89
67
67
89/100
101/151
34/51
IL28B CC
IL28B CT
IL28B TT

FIGURE 4c. SVR4 by cirrhosis (total population)

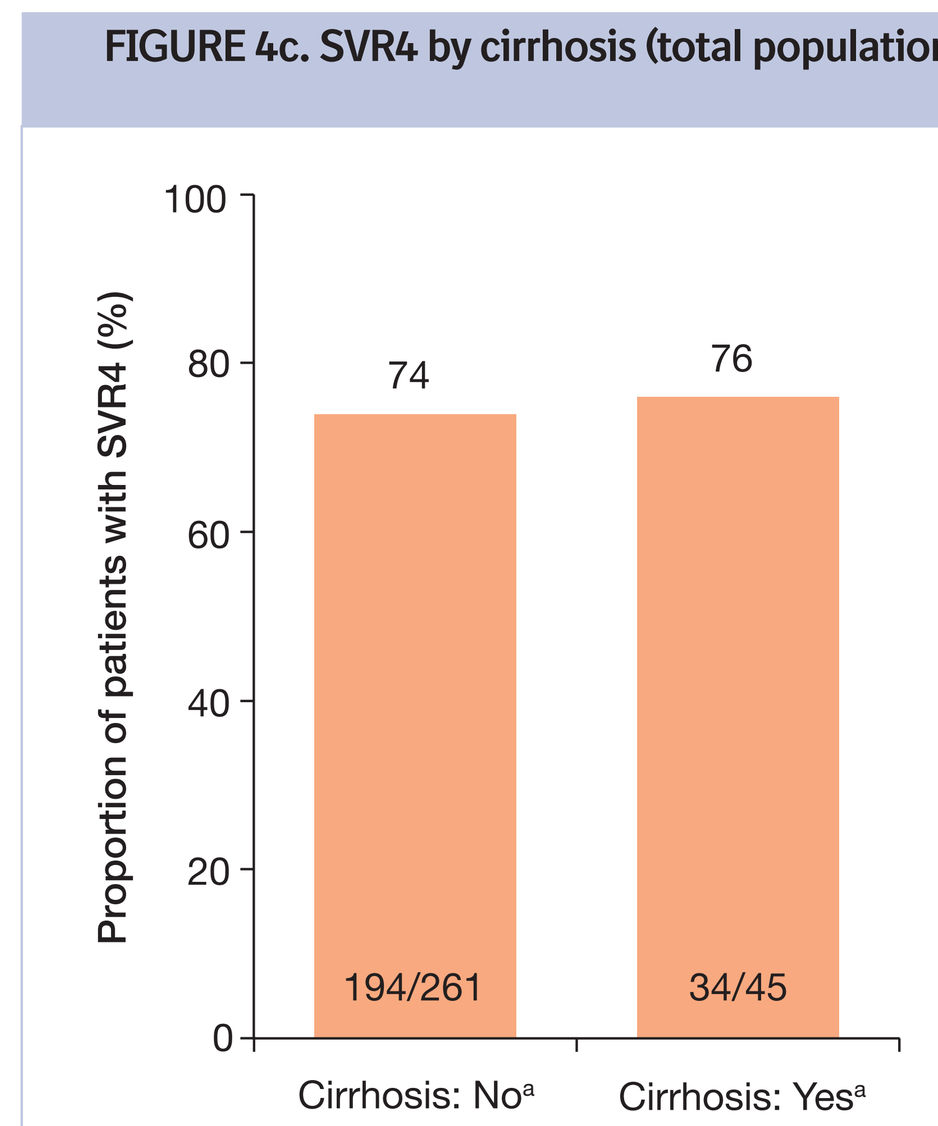

Proportion of patients with SVR4 (%)
100
80
60
40
20
0
74
76
194/261
34/45
Cirrhosis: Noa
Cirrhosis: Yesa

FIGURE 4d. SVR4 by previous PR treatment (total population)

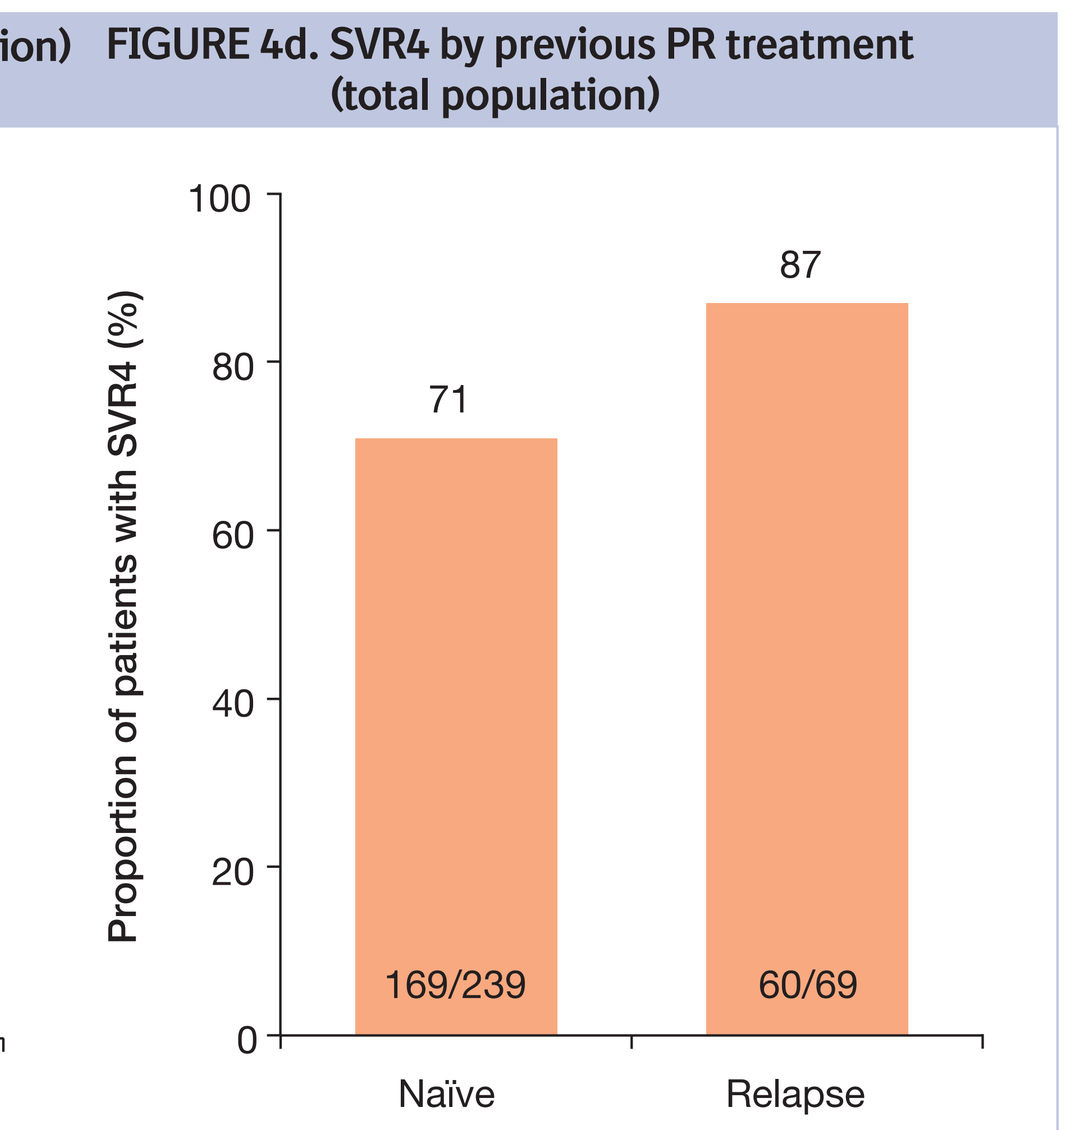

Proportion of patients with SVR4 (%)
100
80
60
40
20
0
71
87
169/239
60/69
Naïve
Relapse

[a]Cirrhosis was determined by the investigator based on Fibroscan, biopsy, and/or other clinical parameters.

FIGURE 4e. SVR4 by ART (total population)

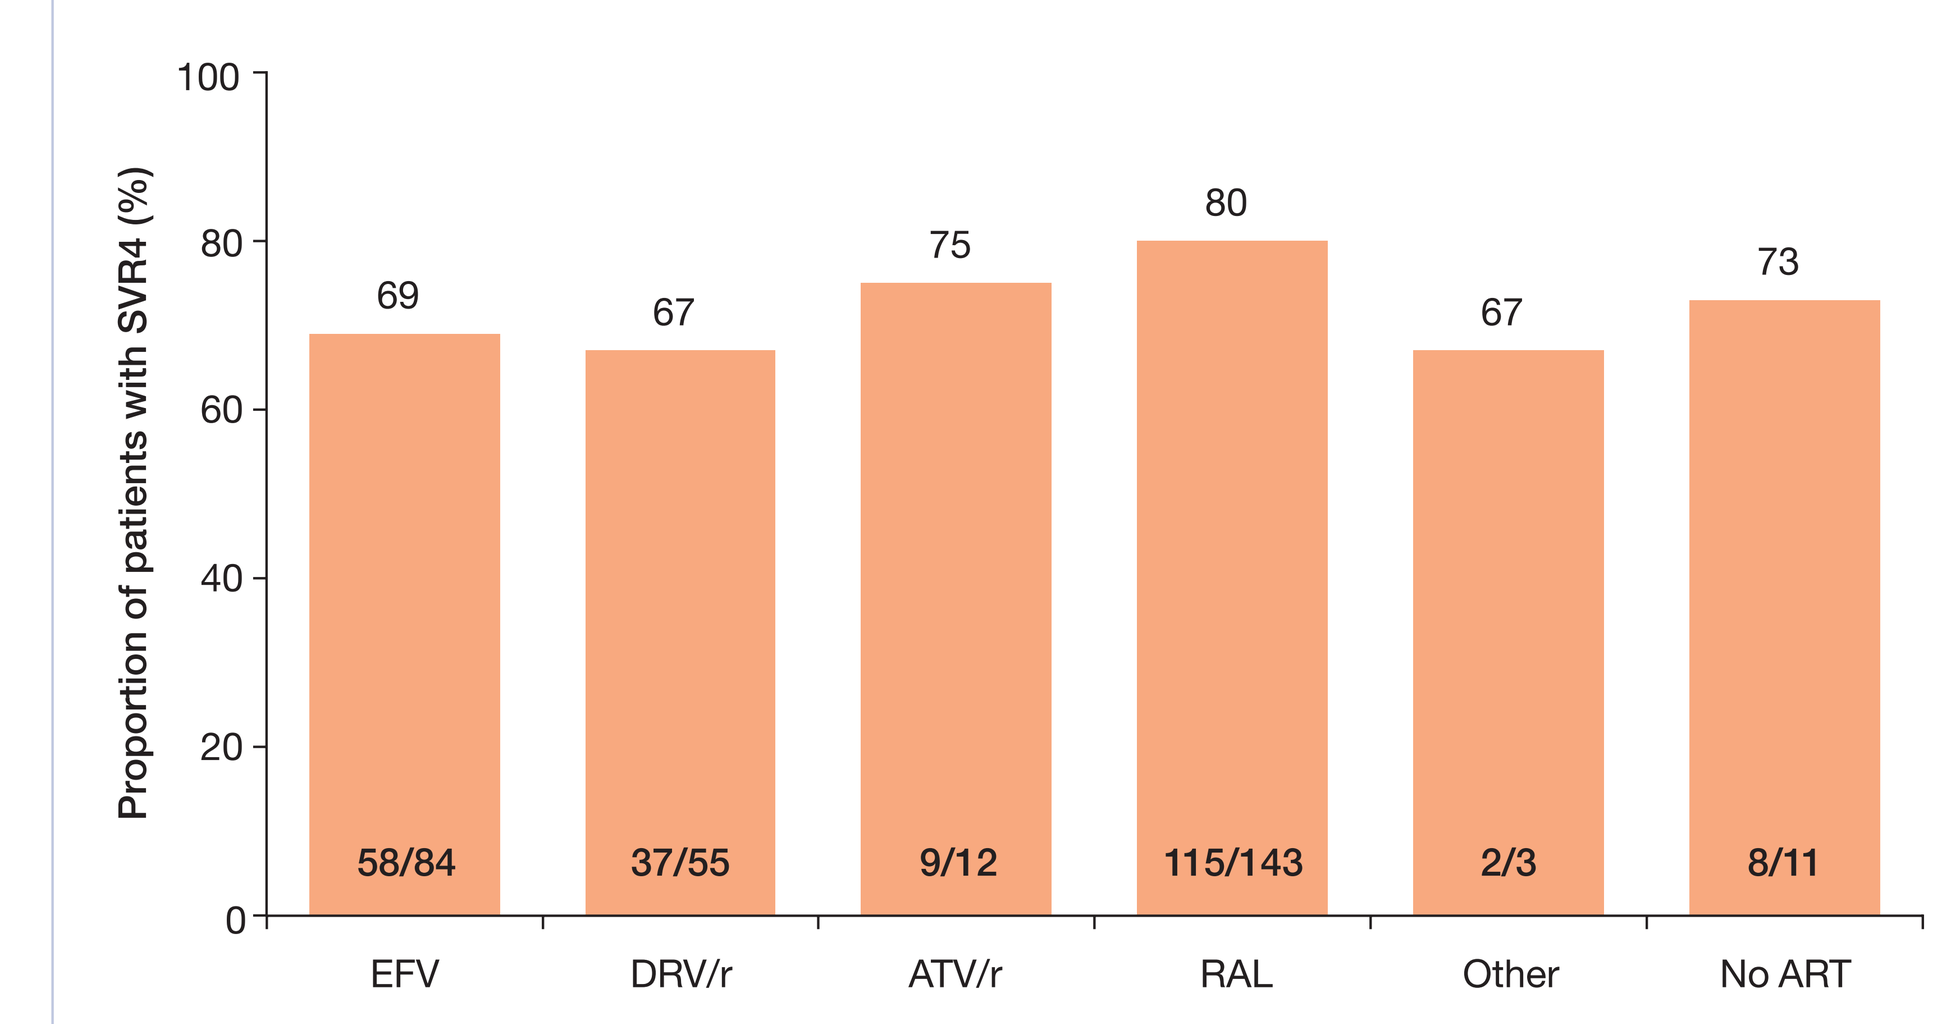

Proportion of patients with SVR4 (%)
100
80
60
40
20
0
69
67
75
80
67
73
58/84
37/55
9/12
115/143
2/3
8/11
EFV
DRV/r
ATV/r
RAL
Other
No ART

ART, antiretroviral therapy; ATV/r, atazanavir/ritonavir; DRV/r, darunavir/ritonavir; EFV, efavirenz; RAL, raltegravir.

FIGURE 5a. ETS

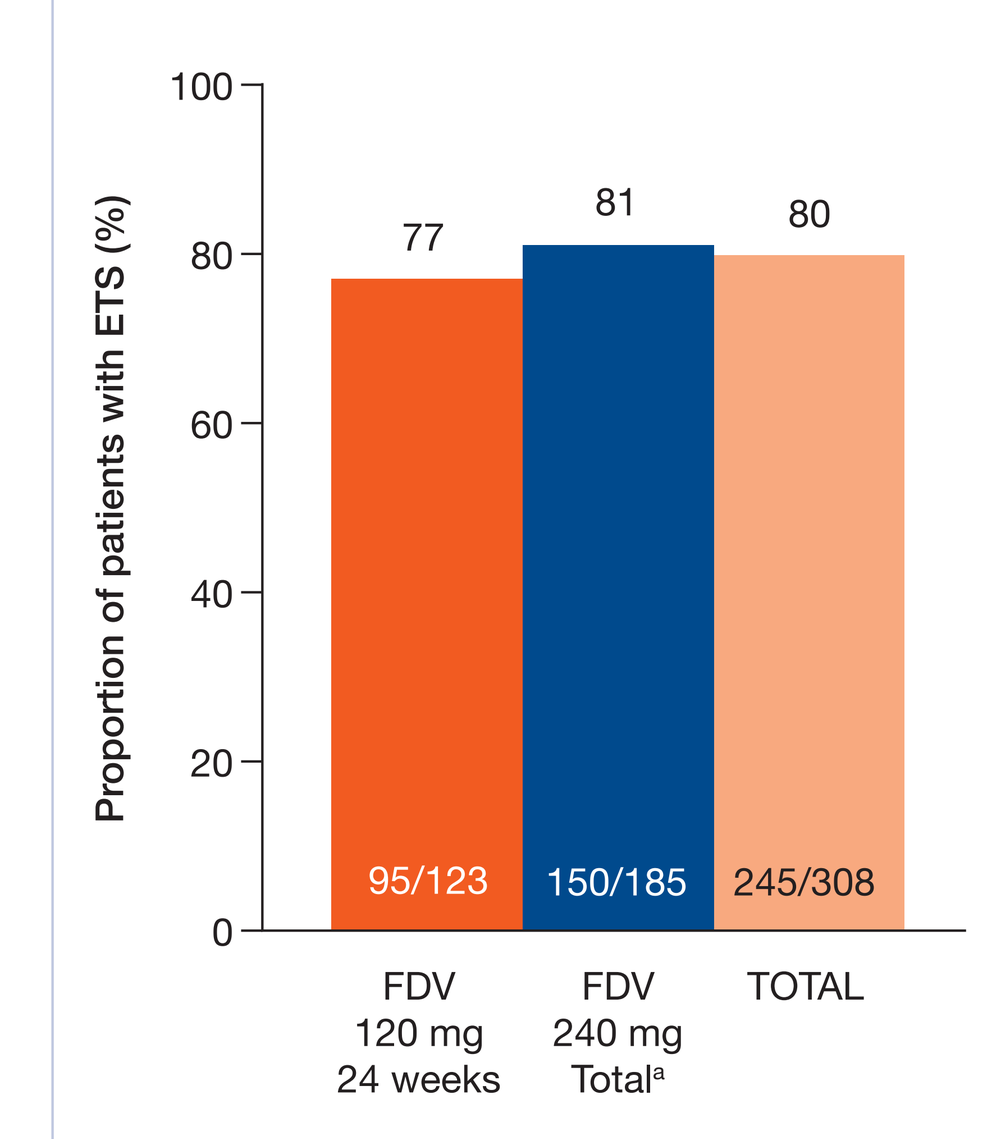

Proportion of patients with ETS (%)
100
80
60
40
20
0
77
81
80
95/123
150/185
245/308
FDV 120 mg 24 weeks
FDV 240 mg Totala
TOTAL

FIGURE 5b. SVR4 in patients who achieved ETS

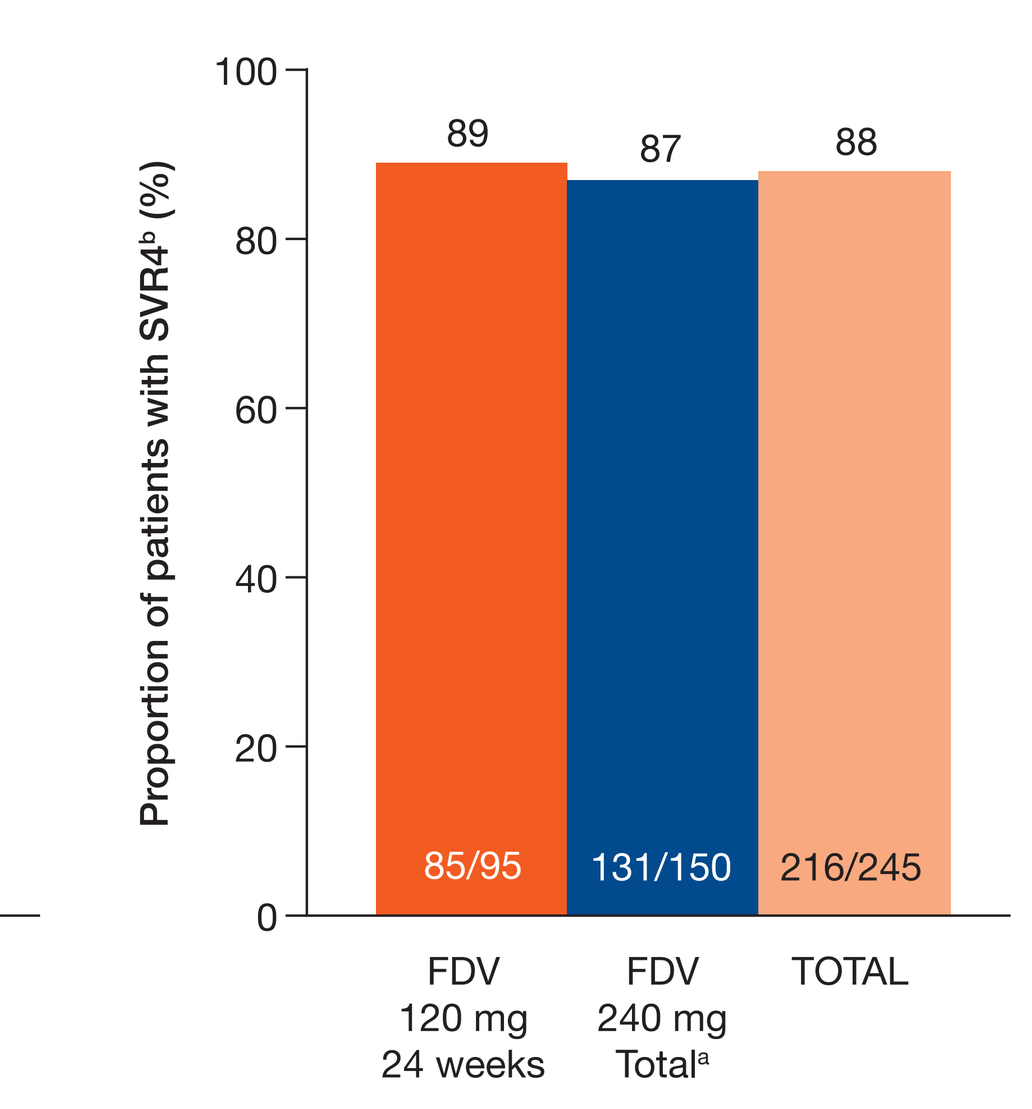

Proportion of patients with SVR4b (%)
100
80
60
40
20
0
89
87
88
85/95
131/150
216/245
FDV 120 mg 24 weeks
FDV 240 mg Totala
TOTAL

[a]Includes patients from 240 mg treatment group who discontinued prior to week 12.
[b]Denominator = patients with ETS regardless of randomization to 24 weeks or 48 weeks of PR.
ETS = HCV RNA <25 IU/mL at week 4 and undetectable at week 8.

FIGURE 5c. SVR4 in patients who achieved ETS according to treatment duration (Per Protocol)

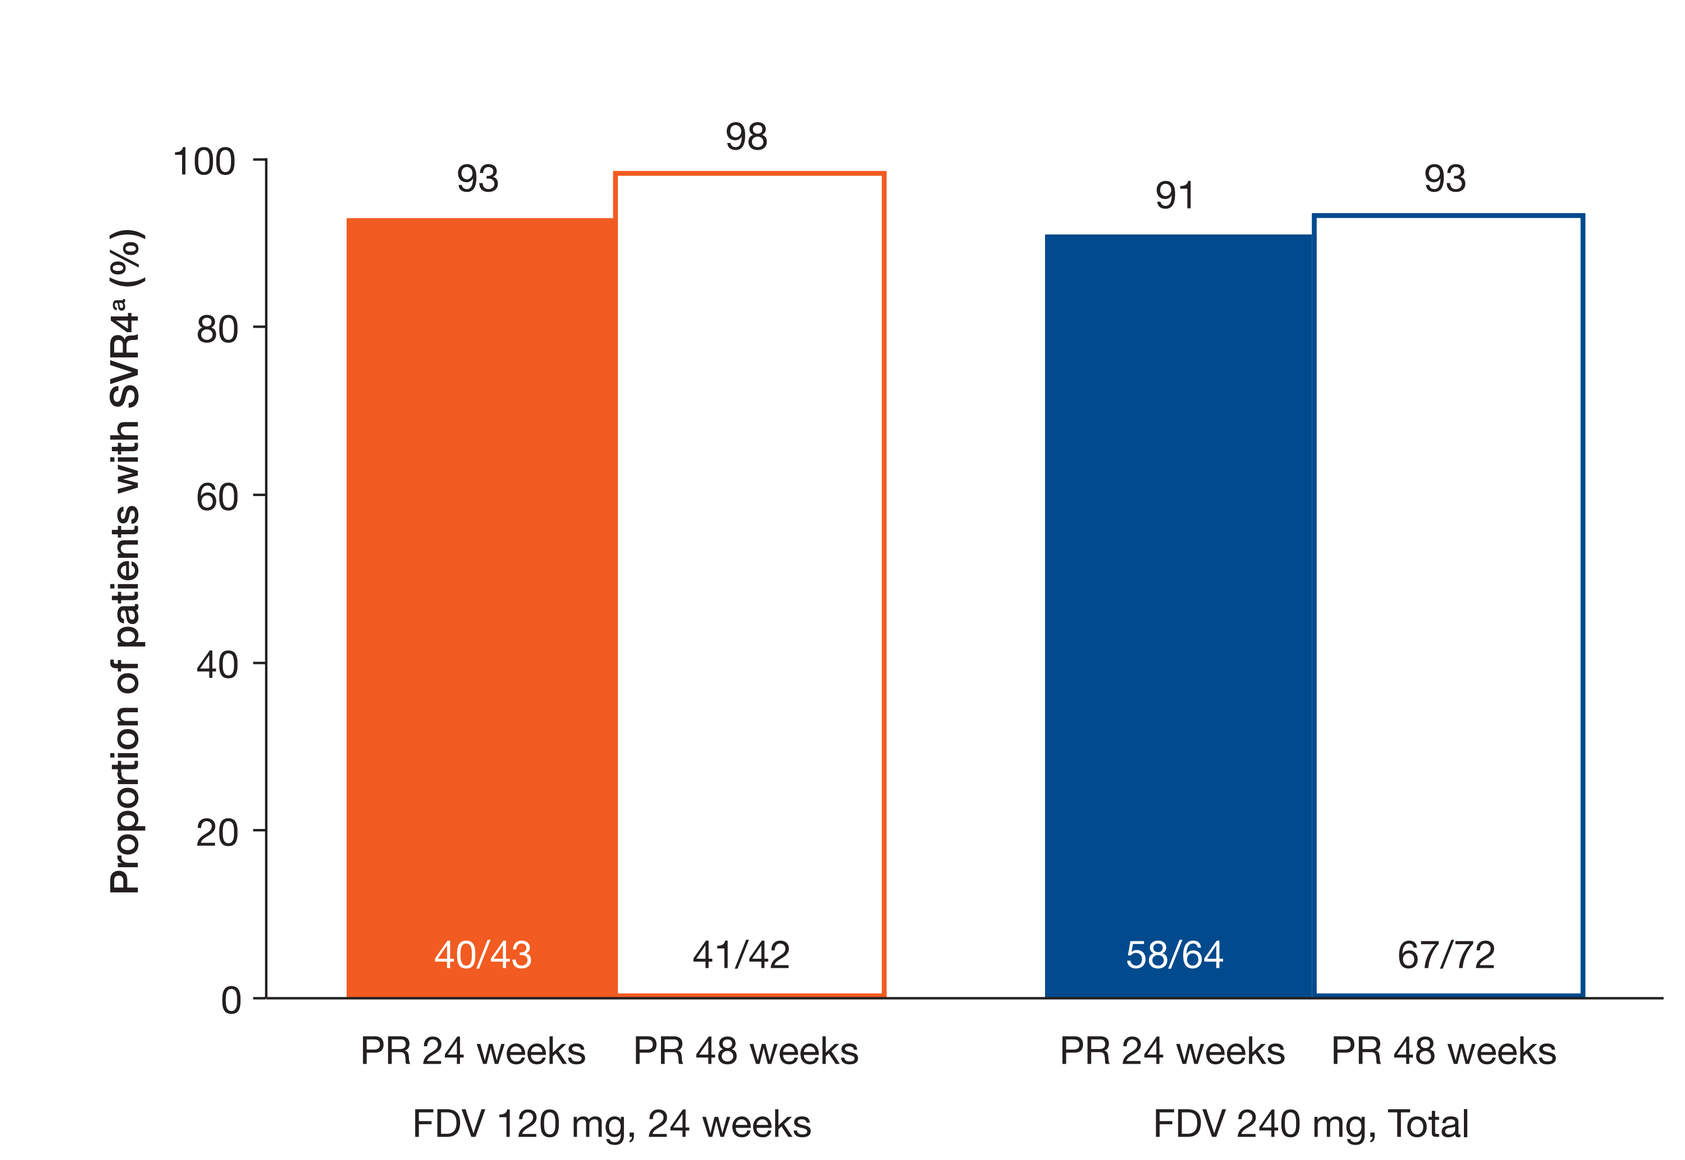

Proportion of patients with SVR4a (%)
100
80
60
40
20
0
93
98
91
93
40/43
41/42
58/64
67/72
PR 24 weeks
PR 48 weeks
PR 24 weeks
PR 48 weeks
FDV 120 mg, 24 weeks
FDV 240 mg, Total

[a]Denominator = patients with ETS who were randomized to stop PR at the indicated time point.

SAFETY

- Incidence of AEs, SAEs, and AEs leading to discontinuation of study treatment similar across FDV treatment groups (**Table 5**).
- Most frequent AEs were nausea and fatigue (**Table 6**).
- Grade ≥3 laboratory abnormalities occurred with similar frequency across all treatment groups (**Table 7**).
- Hemoglobin reduction was similar to historical data with PR-based regimen.
- Bilirubin elevations were:
 - Transient and resolved shortly after completion of FDV treatment
 - Characterized by a predominance of unconjugated bilirubin.
- Among patients with adequate suppression of HIV RNA (<40 c/mL) at baseline, one patient in the FDV 120 mg group lost HIV suppression (>200 copies/mL on 2 consecutive visits) and required a change of ART regimen.

TABLE 5. AE summary

n (%)	FDV 120 mg + PR 24 weeks (N=123)	FDV 240 mg + PR Total (N=185)[a]	Total FDV + PR (N=308)
Any AE	119 (97)	182 (98)	301 (98)
Drug-related AEs	114 (93)	172 (93)	286 (93)
DAIDS Grade 3 or 4 AEs	24 (20)	33 (18)	57 (19)
DAIDS Grade 4 AEs	5 (4)	5 (3)	10 (3)
AEs leading to discontinuation of FDV only	1 (1)	1 (1)	2 (1)
AEs leading to discontinuation of all medication	9 (7)	13 (7)	22 (7)
Serious AEs[c]	17 (14)	15 (8)	32 (10)
Resulting in death	0	1 (1)[b]	1 (<1)
Life threatening	1 (1)	1 (1)	2 (1)
Required hospitalization	15 (12)	12 (6)	27 (9)
Other[d]	4 (3)	4 (2)	8 (3)

[a]Includes additional patients from 240 mg treatment group who discontinued prior to week 12.
[b]Death from SJS with onset 145 days after discontinuing treatment while on systemic antibiotics. [c]A subject may be counted in more than one seriousness criterion. [d]Other serious AEs include those that persist or cause significant disability/incapacity, or other medically important serious events.
DAIDS, Division of AIDS table for grading the severity of adult and pediatric adverse events.

TABLE 6. Most frequent AEs (≥20% overall)

n (%)	FDV 120 mg + PR 24 weeks (N=123)	FDV 240 mg + PR Total (N=185)[a]	Total FDV + PR (N=308)
Nausea	34 (28)	81 (44)	115 (37)
Fatigue	39 (32)	65 (35)	104 (34)
Diarrhea	31 (25)	52 (28)	83 (27)
Headache	29 (24)	47 (25)	76 (25)
Asthenia	32 (26)	39 (21)	71 (23)
Decreased appetite	30 (24)	36 (20)	66 (21)

[a]Includes additional patients from 240 mg treatment group who discontinued prior to week 12.

TABLE 7. Grade ≥3 laboratory abnormalities

n (%)	FDV 120 mg + PR 24 weeks (N=123)	FDV 240 mg + PR Total (N=185)[a]	Total FDV + PR (N=308)
Total patients with Grade ≥3 laboratory abnormalities	50 (41)	87 (47)	137 (44)
Hemoglobin decreased	2 (2)	3 (2)	4 (2)
White blood cell count	8 (7)	11 (6)	19 (6)
Platelets decreased	13 (11)	13 (7)	23 (7)
Neutrophil, absolute	24 (20)	31 (17)	55 (18)
ALT increased	3 (3)	3 (2)	6 (2)
Creatinine increased	0	0	0
Total bilirubin[b]	17 (14)	43 (23)	60 (19)

[a]Includes additional patients from 240 mg treatment group who discontinued prior to week 12.
[b]Worst DAIDS grade on-treatment.
ALT, alanine transaminase.

SUMMARY

- In this interim analysis in patients co-infected with HIV and HCV GT-1, treatment with FDV resulted in a total SVR4 rate of 74% in all patients.
 - SVR4 rates were similar across FDV dose (120 mg vs 240 mg) and duration (12 weeks vs 24 weeks).
- High SVR4 rates were achieved regardless of:
 - HCV genotype
 - IL28B genotype
 - Presence of compensated cirrhosis
 - ART regimen, including efavirenz, raltegravir, and boosted PIs.
- A high proportion of patients (80%) achieved ETS and 88% of those patients achieved SVR4.
 - SVR4 rates were comparable among patients who received 24 or 48 weeks of PR.
- The safety profile of FDV in HIV and HCV GT-1 co-infected patients was similar to that observed in HCV GT-1 mono-infected patients.

CONCLUSIONS

- In this large, Phase III study in patients co-infected with HIV and HCV GT-1, FDV + PR was well tolerated and efficacious and did not have any impact on HIV RNA suppression by ART.
- The results confirm the value of using ETS to guide PR treatment duration of 24 weeks vs 48 weeks.
- These SVR4 results are encouraging given the difficult-to-treat population (79% GT-1a, 15% cirrhosis, 82% high baseline HCV RNA, 66% IL28B non-CC), and suggest that FDV + PR may become an important option for the treatment of chronic HCV GT-1 infection in patients co-infected with HIV with or without concomitant ART.

REFERENCES: 1. White PW, et al. *Antimicrob Agents Chemother* 2010;54:4611–4618. **2.** Manns MP, et al. *J Hepatol* 2011; 54:1114–1122. **3.** Ferenci P, et al. *J Hepatol* 2013;58:S569. **4.** Sabo J, et al. *CROI* 2013, Abstract 35.

ACKNOWLEDGEMENTS

- We thank the patients, the investigators and all of our colleagues at Boehringer Ingelheim who worked to provide the data reported here.
- Medical writing assistance, supported by Boehringer Ingelheim, was provided by Esther Race of Choice Healthcare Solutions during preparation of this poster.

DISCLOSURES

- The authors report the following disclosures: AbbVie (JR, SB, PI, MKJ, DD, MN), BI (JR, MN, VS, KA, SB, JM, MP, PI, MKJ, KM, JK, JOS, RV, MM, DD), Bionor (JR), BMS (JR, MN, VS, KA, SB, MP, PI, MKJ, KM, DD), Gilead (JR, MN, VS, SB, JM, MP, PI, MB, MKJ, MaN, KM, DD), GSK (MP), Hexal (KA), Idenix (DD), Medscape (JR), Merck (JR, VS, MKJ, DD), MSD (MN, KA, MP, PI, MB), Novartis (MP), Roche (SB, MP, PI), Tibotec/Janssen (JR, MN, VS, SB, JM, MP, PI, MB, MKJ, KM), Tobira (JR), Vertex (JR, MP, MKJ, KM), ViiV (JR, MN, VS, MP, MB), ViralEd (JR), and none (CT).
- This study was sponsored by Boehringer Ingelheim.
- This presentation includes discussion of investigational drugs not approved for use in humans.

STARTVerso™4
Committed to Cure

The 64[th] Annual Meeting of the American Association for the Study of Liver Diseases (AASLD), November 1–5, 2013, Washington, DC.