FIBROSIS REGRESSION OBSERVED IN TREATMENT WITH FGF19 VARIANTS IN A DIET INDUCED MOUSE MODEL OF NASH IS PREDOMINANTLY MEDIATED BY FGFR4 ACTIVITY AND INDEPENDENT OF WEIGHT LOSS OR DECREASES IN HEPATIC STEATOSIS

Maria Deato, Brian Ko, Emily Snyder, Bernard Allan and Hui Tian
NGM Biopharmaceuticals, Inc., South San Francisco, CA 94080, USA

BACKGROUND

- Fibroblast growth factor 19 (FGF19) is an endocrine hormone produced in the ileum in an Farnesoid X receptor (FXR)-dependent manner[1,2]
- Previously, our group has demonstrated hepatoprotective effects of FGF19 analog NGM282 in murine models of NASH[3-5]
- Similarly, M52 is also an engineered analog of FGF19 which retains bile acid regulation via FGFR4-β-Klotho binding, retains insulin-sensitizing and body weight-reduction metabolic activities via FGFR1c-β-Klotho, but lacks the potential tumorigenic activity of FGF19[6]
- DEL30 is a variant of FGF19 with a truncated carboxyl terminus which is required for β-Klotho interaction and results in complete ablation of FGFR1c signaling. However, DEL30 is still capable of β-Klotho-independent FGFR4 signaling[7]

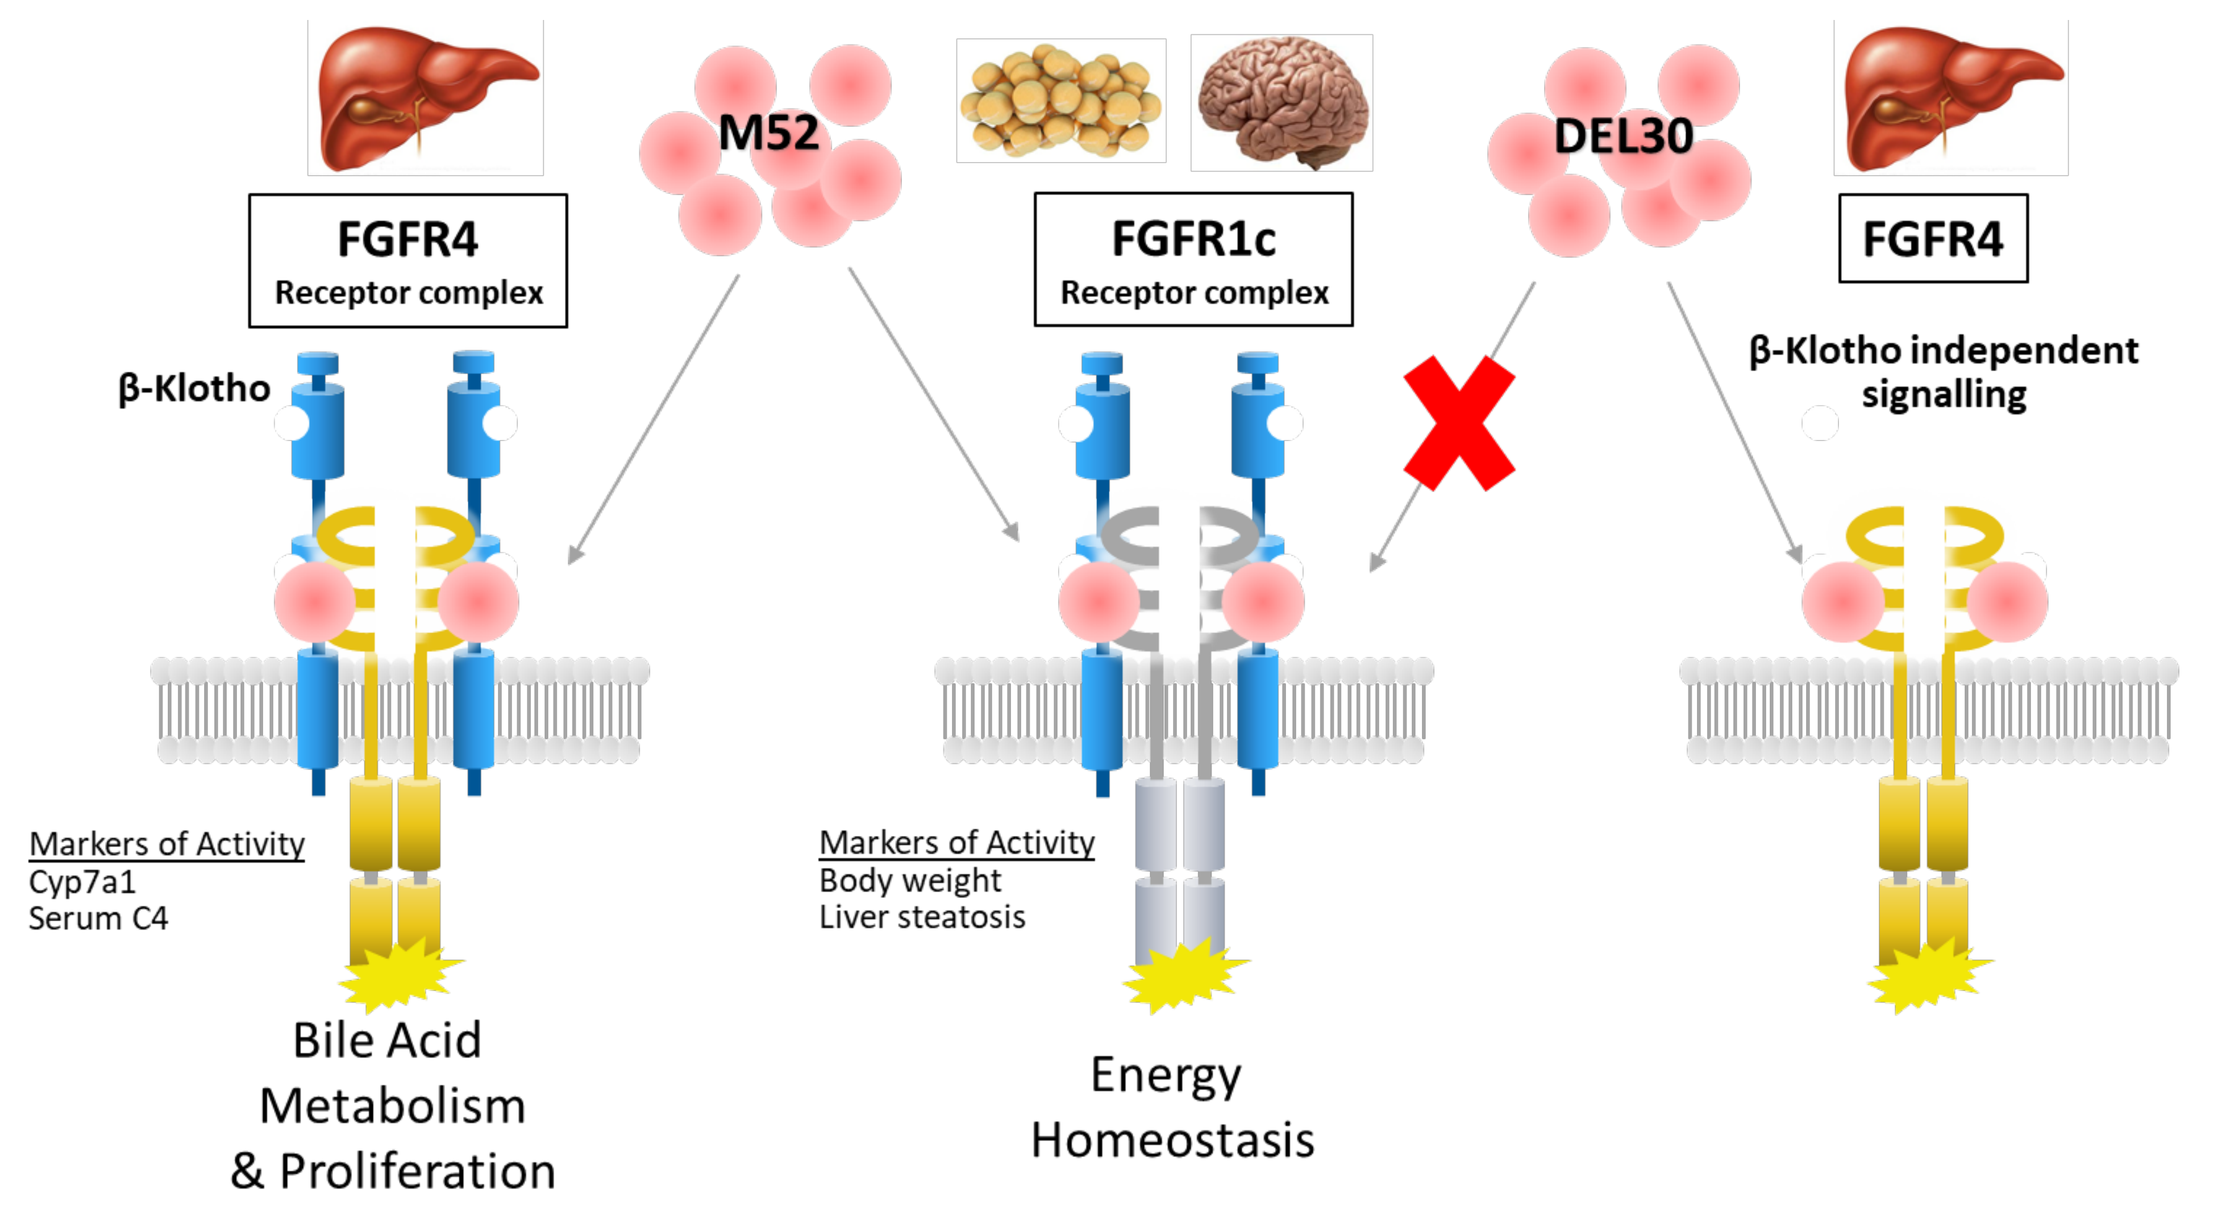
M52
DEL30
FGFR4
Receptor complex
FGFR1c
Receptor complex
FGFR4
β-Klotho
β-Klotho independent signalling
Markers of Activity
Cyp7a1
Serum C4
Markers of Activity
Body weight
Liver steatosis
Bile Acid Metabolism & Proliferation
Energy Homeostasis

NASH and HFFCD Model

- NASH is a common chronic liver disease associated with obesity, diabetes, and metabolic syndrome and has limited treatment options[8]
- There is also an increased risk of HCC in NASH patients, independent of the presence of cirrhosis[8]
- Mice fed a high-fat, high-fructose, high-cholesterol diet (HFFCD) display significantly increased steatosis, hepatocyte ballooning degeneration and lobular inflammation, and marked fibrosis – all histological features resembling NASH pathology[9]. These histological features including liver fibrosis are reversible with chow diet replacement
- HFFCD mice develop concomitant metabolic dysfunction such as obesity and insulin resistance, and are considered a model relevant to NASH clinical disease progression[9]
- Compounds with effects on lowering body weight in HFFCD model have concomitant effect in reducing hepatic steatosis and inflammation leading to no worsening of fibrosis[11]
- It is unknown if improvements in inflammation and fibrosis can be achieved independent of body weight or liver fat reduction

STUDY METHODS and DESIGN

- C57BL6/J mice (Jackson Laboratories, #000664, 9-week old) were placed on a high-fat, high-fructose, high-cholesterol diet (HFFCD, 40% fat, 22% fructose, 2% cholesterol) for 30 weeks to establish extensive liver fibrosis and NASH phenotype
- The following treatment groups (n=9 animals per group) received stable long-term transgene expression of GFP, M52 or DEL30 using an adeno-associated viral vector (AAV) at 3.3 x10 [11] genome copies per mouse
- Livers were stained for H&E, osmium tetrachloride (for liver fat), and picro-sirius red (for collagen)
- Quantitative-RT-PCR was conducted using pre-made Taqman Gene Expression Assay primers from Life Technologies
- Picro-sirius red tissue stain was quantified by Aperio ImageScope positive pixel count algorithm

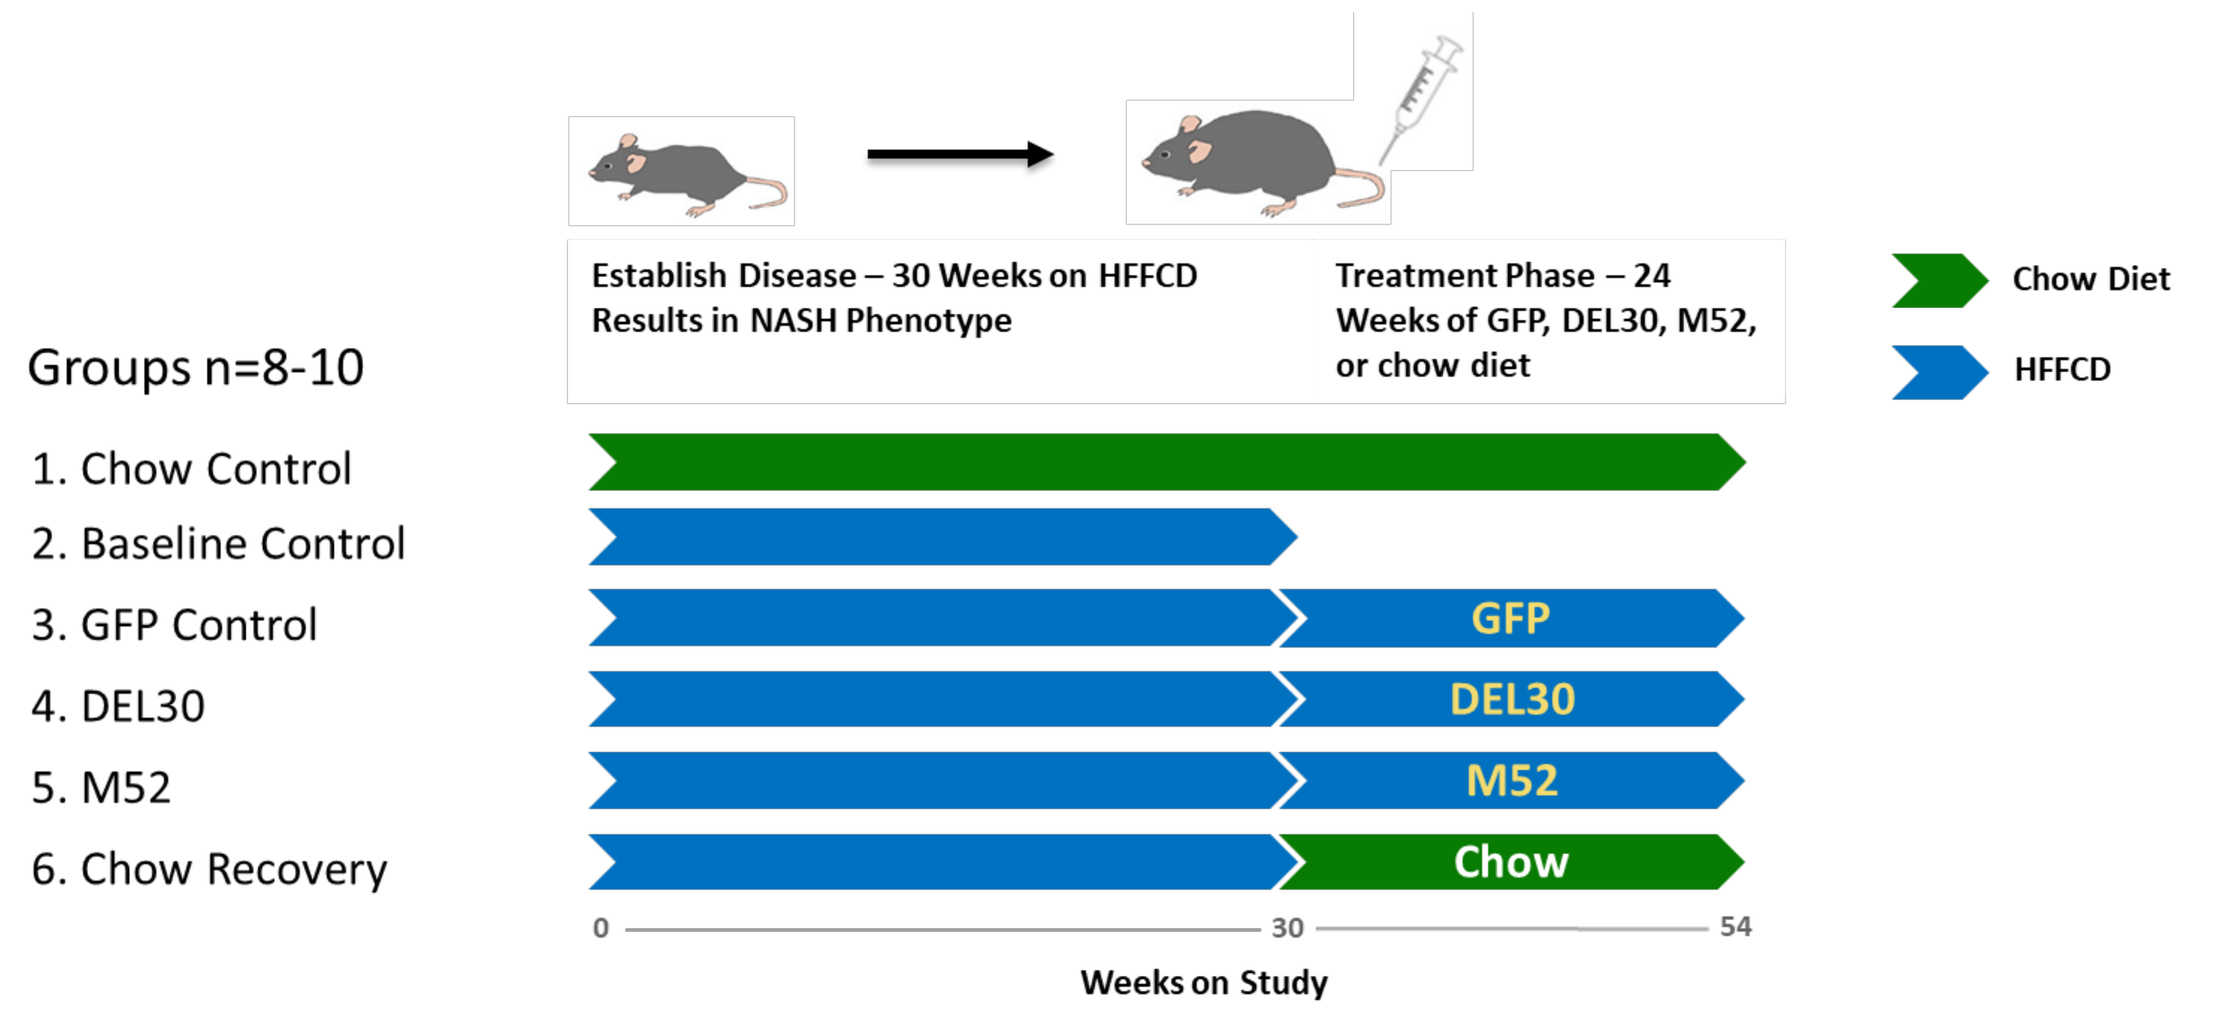
Establish Disease – 30 Weeks on HFFCD Results in NASH Phenotype
Treatment Phase – 24 Weeks of GFP, DEL30, M52, or chow diet
Chow Diet
HFFCD
Groups n=8-10
1. Chow Control
2. Baseline Control
3. GFP Control
4. DEL30
5. M52
6. Chow Recovery
GFP
DEL30
M52
Chow
0
30
54
Weeks on Study

RESULTS

M52 is a Potent FGFR1c/FGFR4 Dual Agonist and DEL30 is FGFR4 Specific

- DEL30 and M52 suppress bile acid synthesis (M52>DEL30)
- Exposure levels of DEL30 and M52 were comparable

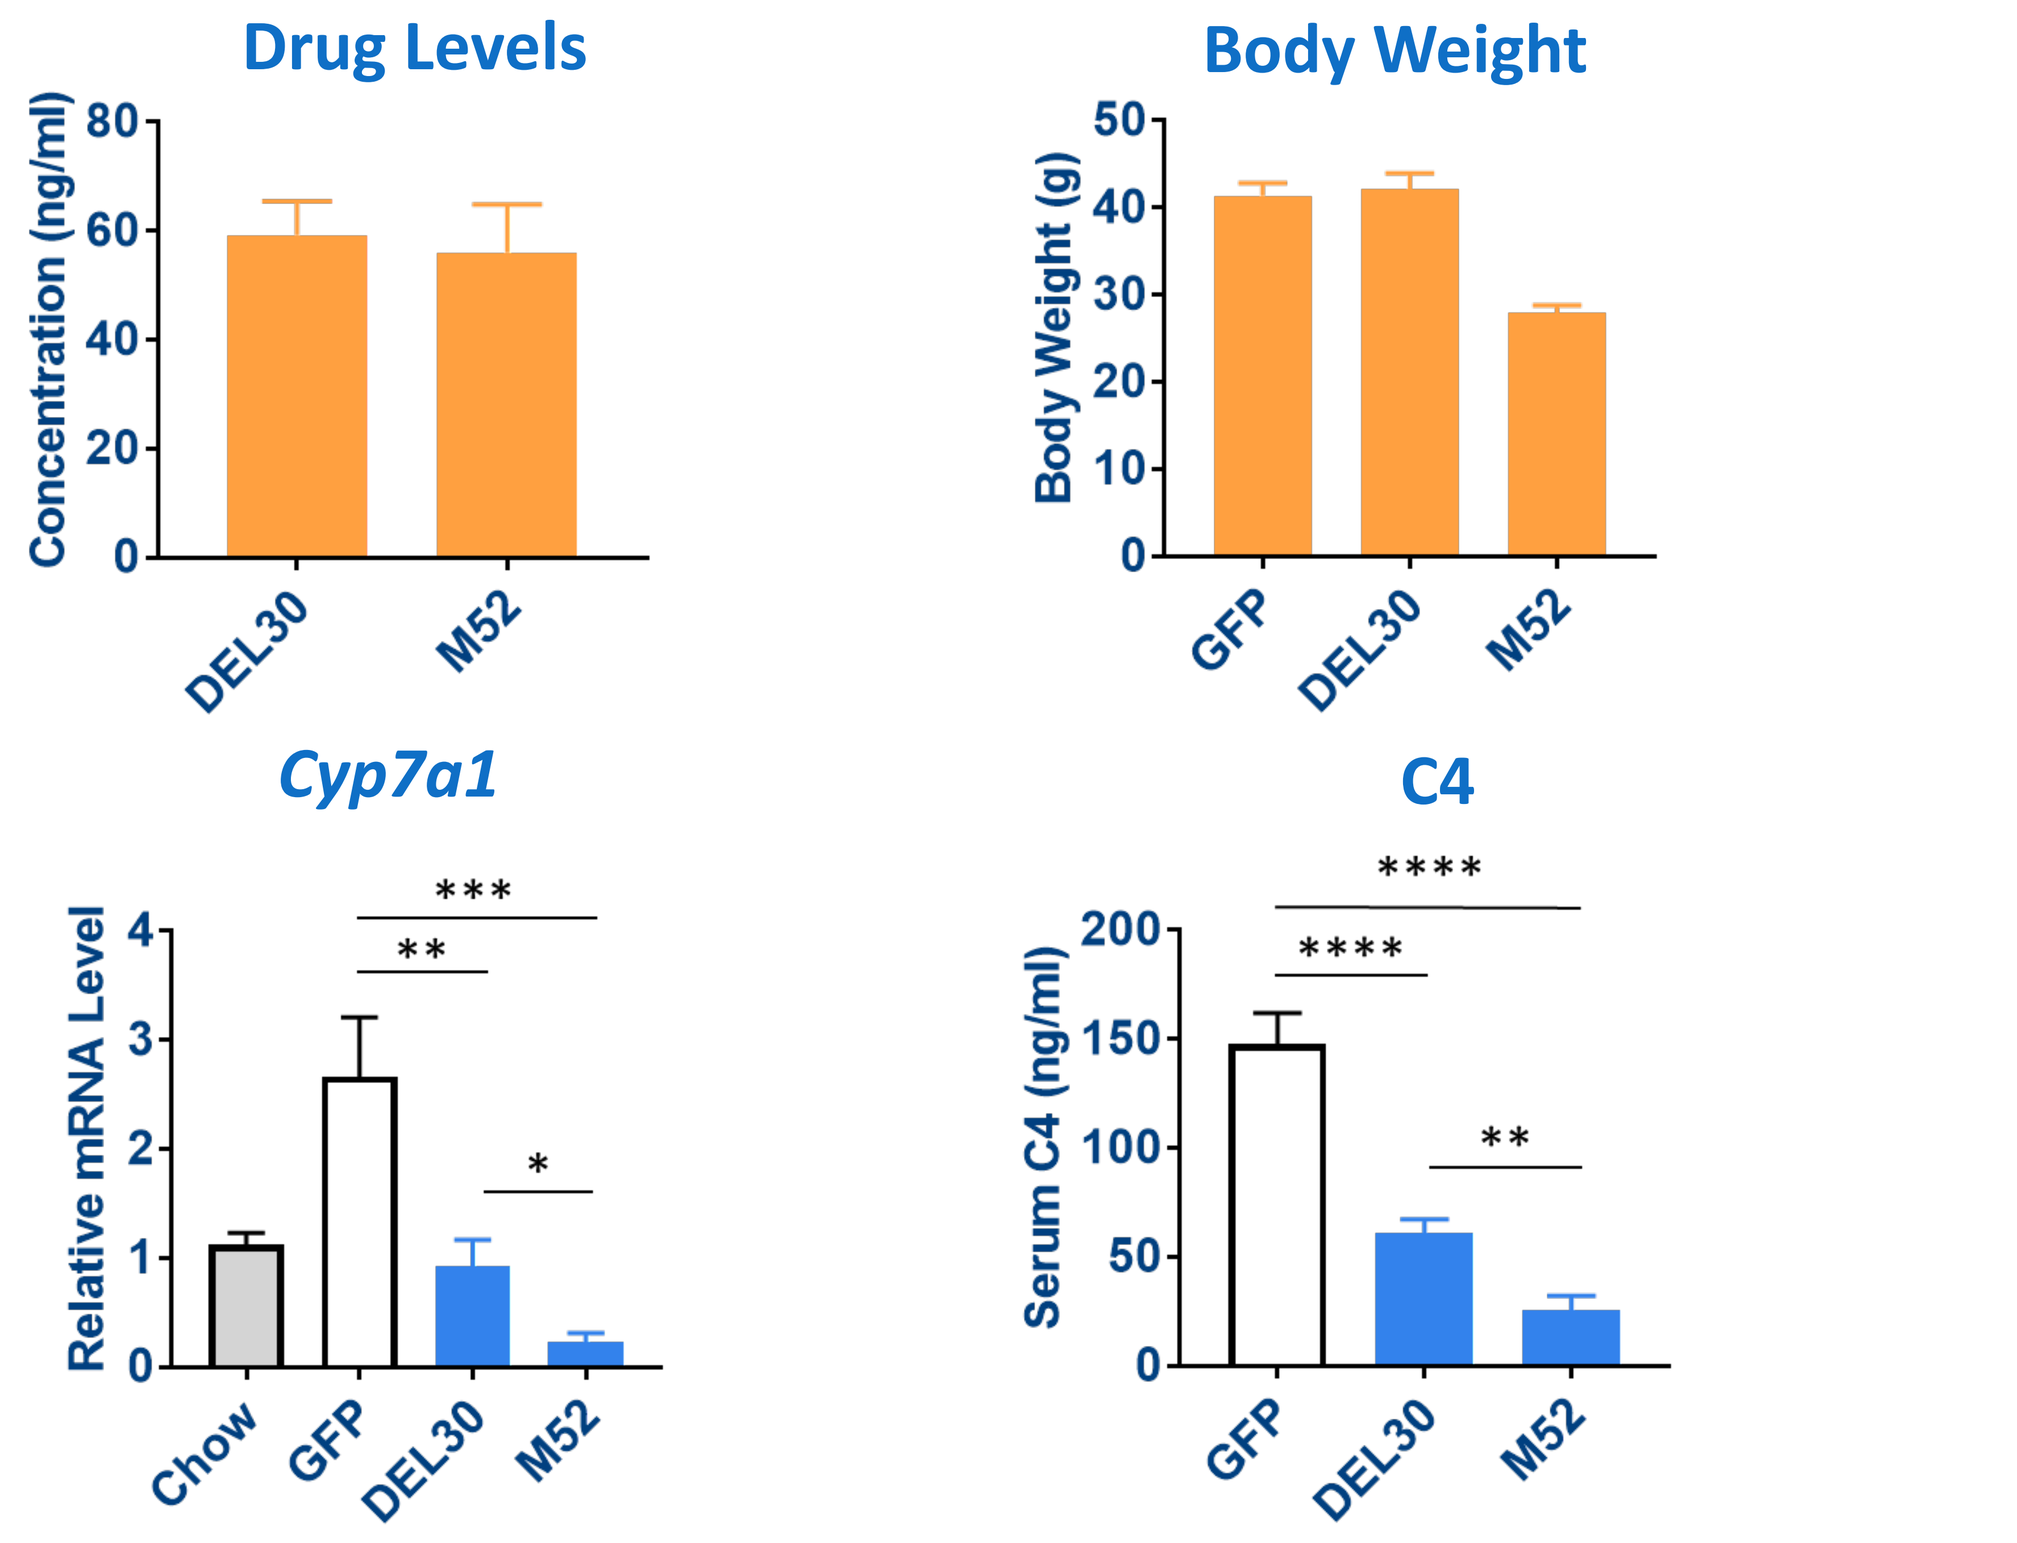
Drug Levels
Concentration (ng/ml)
DEL30
M52
Body Weight
Body Weight (g)
GFP
DEL30
M52
Cyp7a1
Relative mRNA Level
Chow
GFP
DEL30
M52
C4
Serum C4 (ng/ml)
GFP
DEL30
M52

(**** p<0.0001, *** p<0.001, p<0.01, p<0.05)

M52 Normalizes Liver Weight While DEL30 Has No Effect on Liver Weight

- DEL30 does not effect liver weight or body weight
- Chow Recovery and M52 normalize liver weight

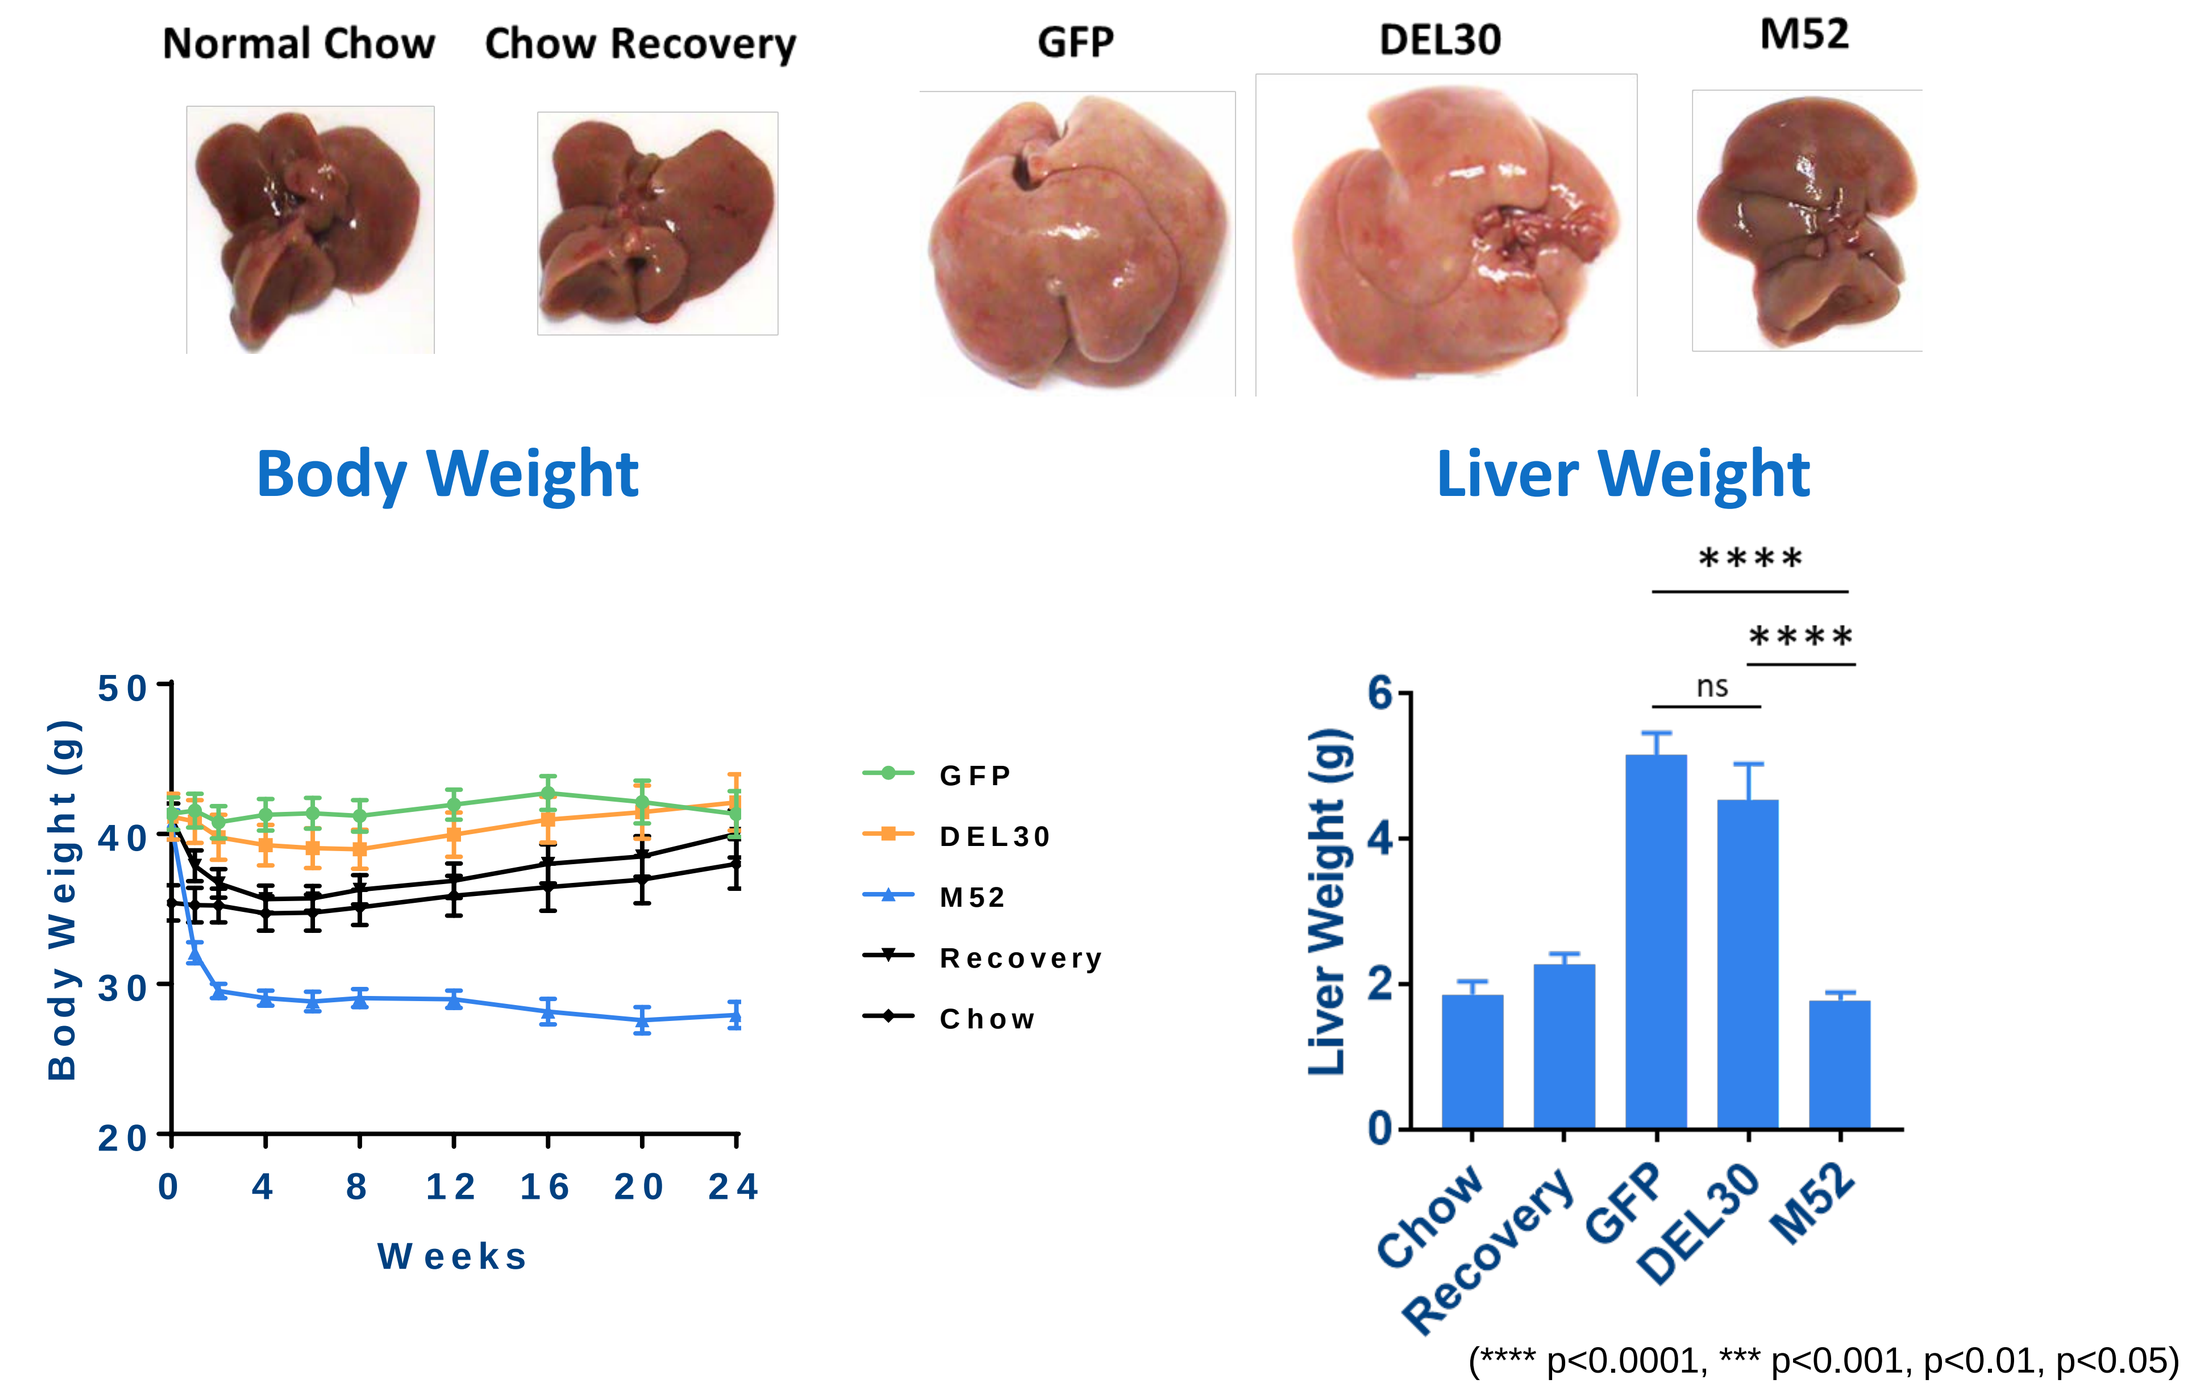
Normal Chow
Chow Recovery
GFP
DEL30
M52
Body Weight
Body Weight (g)
Weeks
GFP
DEL30
M52
Recovery
Chow
Liver Weight
Liver Weight (g)
Chow
Recovery
GFP
DEL30
M52

(**** p<0.0001, *** p<0.001, p<0.01, p<0.05)

M52 Normalizes ALT and AST and Reduces Markers of Liver Inflammation

- FGFR4 activation also improves liver enzymes and reduces gene expression of inflammation markers

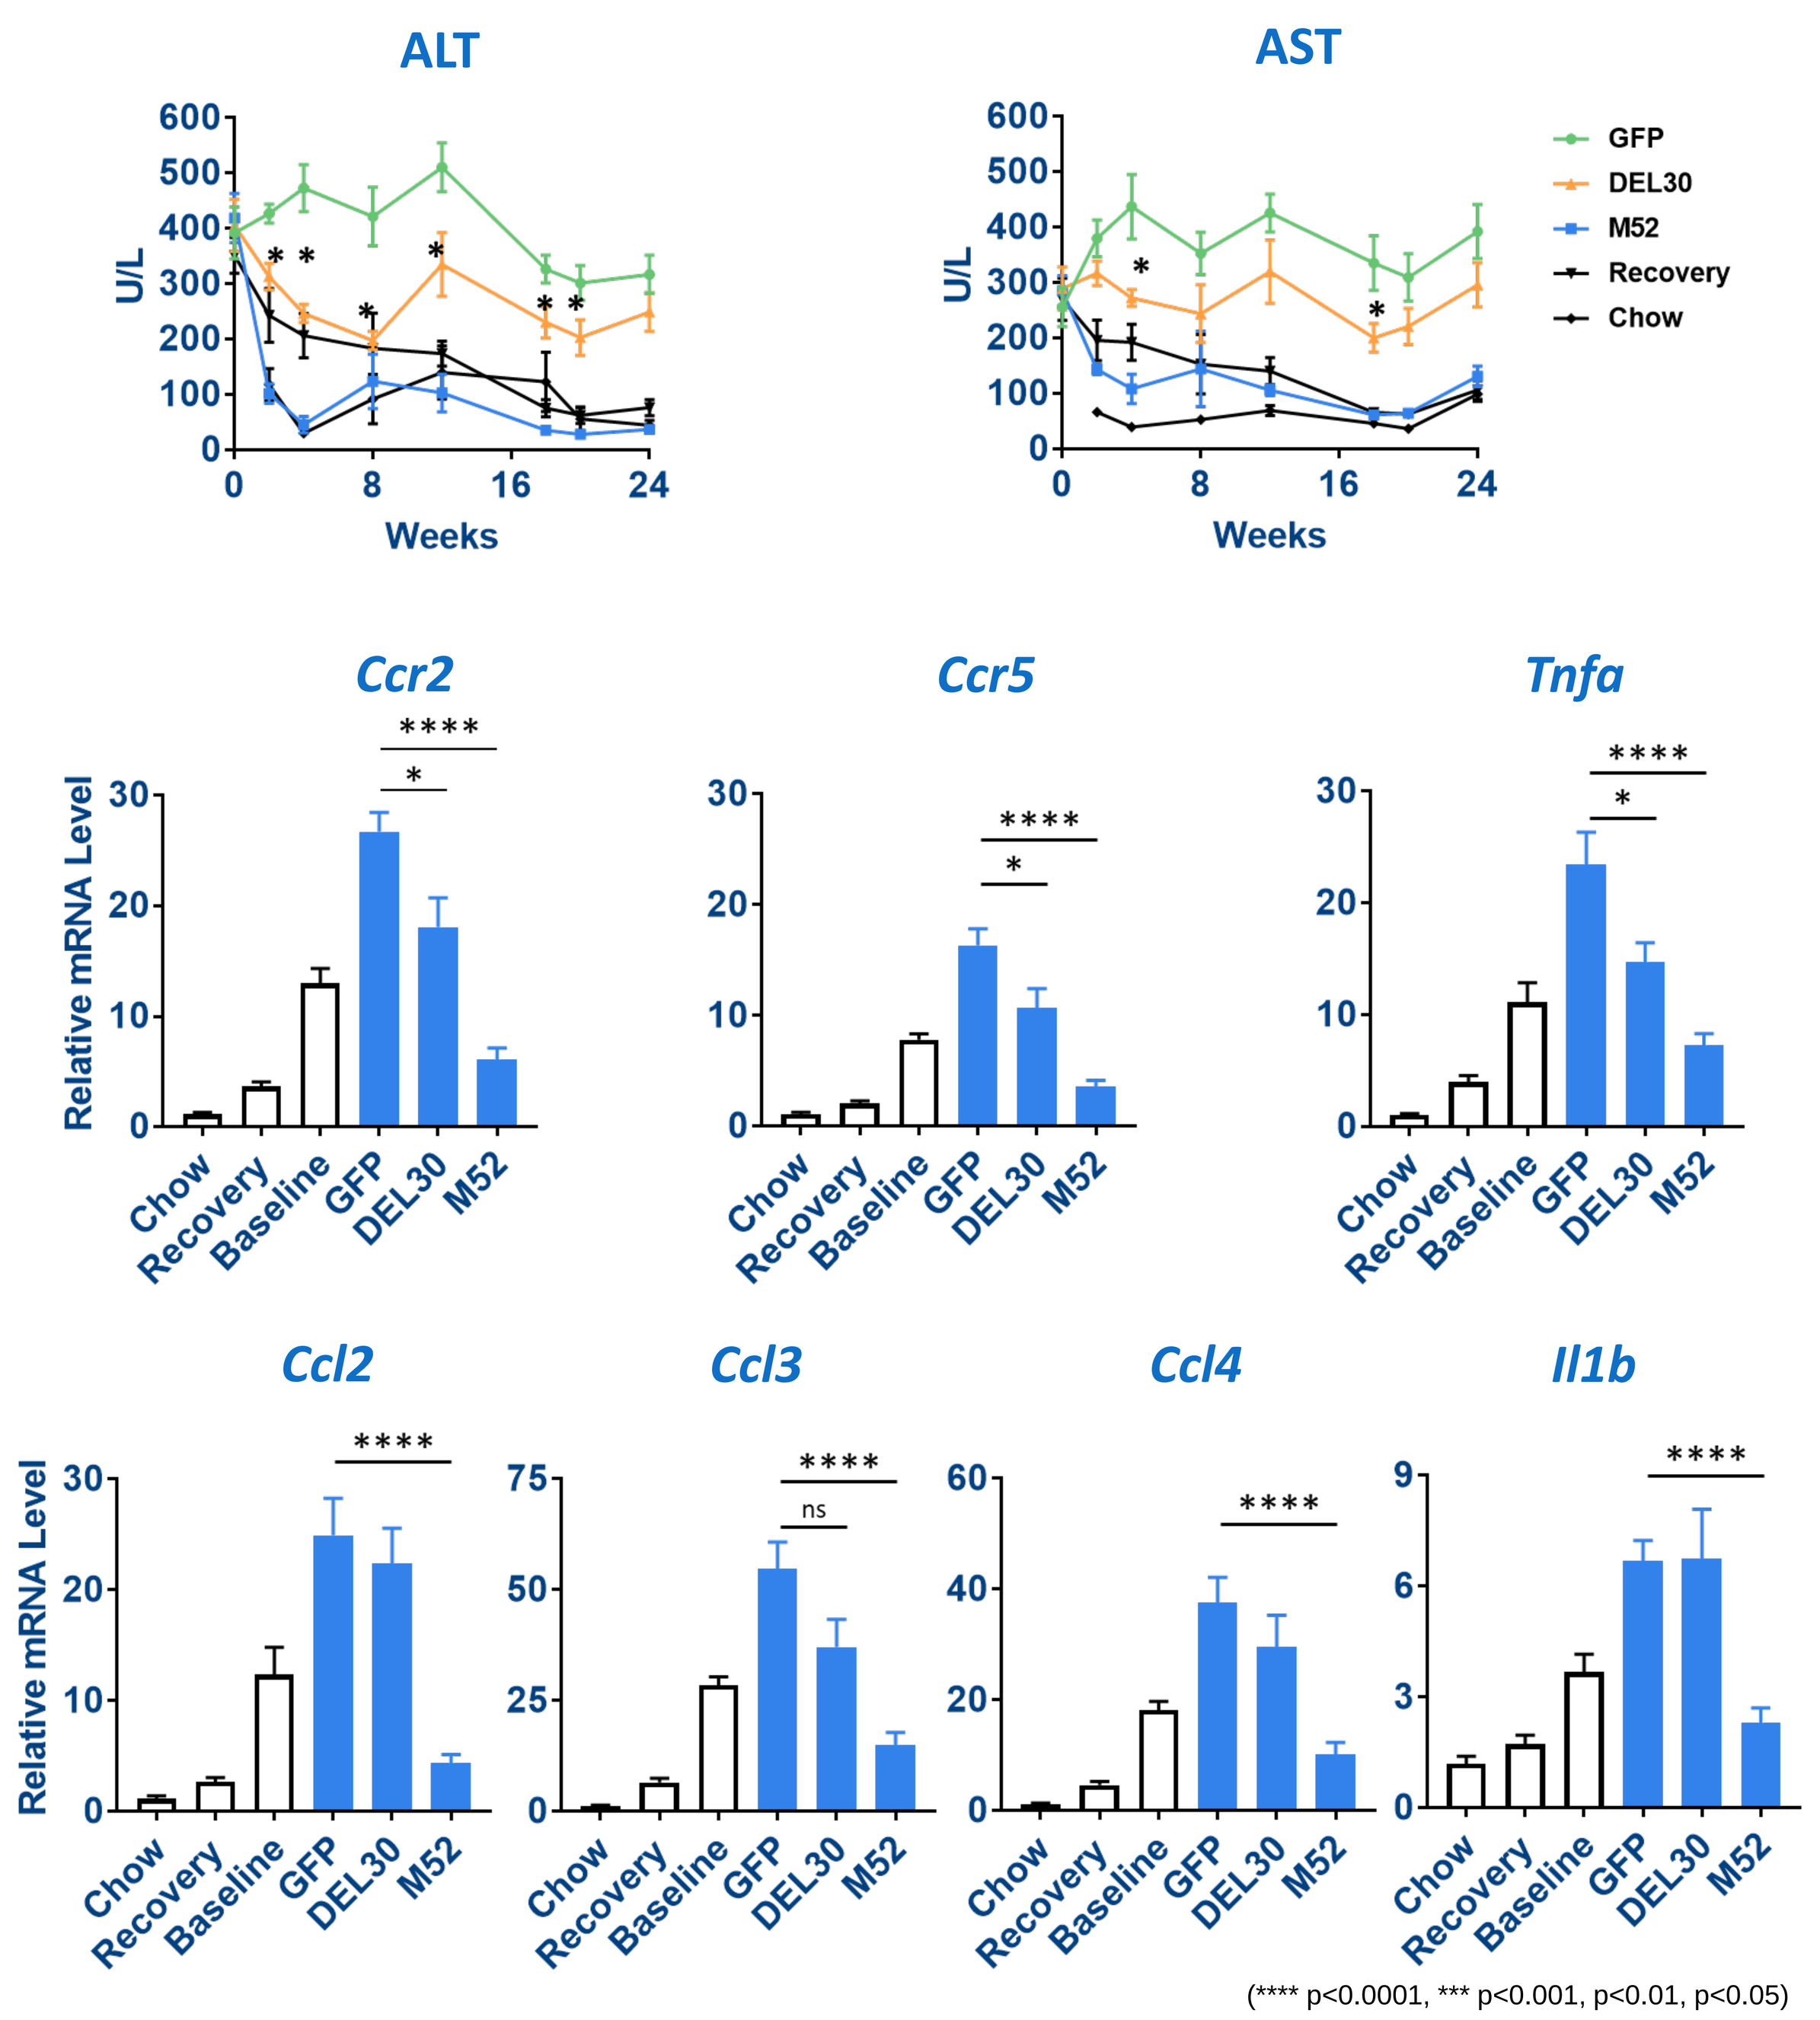
ALT
AST
U/L
Weeks
GFP
DEL30
M52
Recovery
Chow
Ccr2
Ccr5
Tnfa
Ccl2
Ccl3
Ccl4
Il1b
Relative mRNA Level
Chow
Recovery
Baseline
GFP
DEL30
M52

(**** p<0.0001, *** p<0.001, p<0.01, p<0.05)

Fibrosis Regression with M52 and DEL30 Treatment with Ongoing Injury

- DEL30 reduces liver expression of collagen and markers of hepatic stellate cells

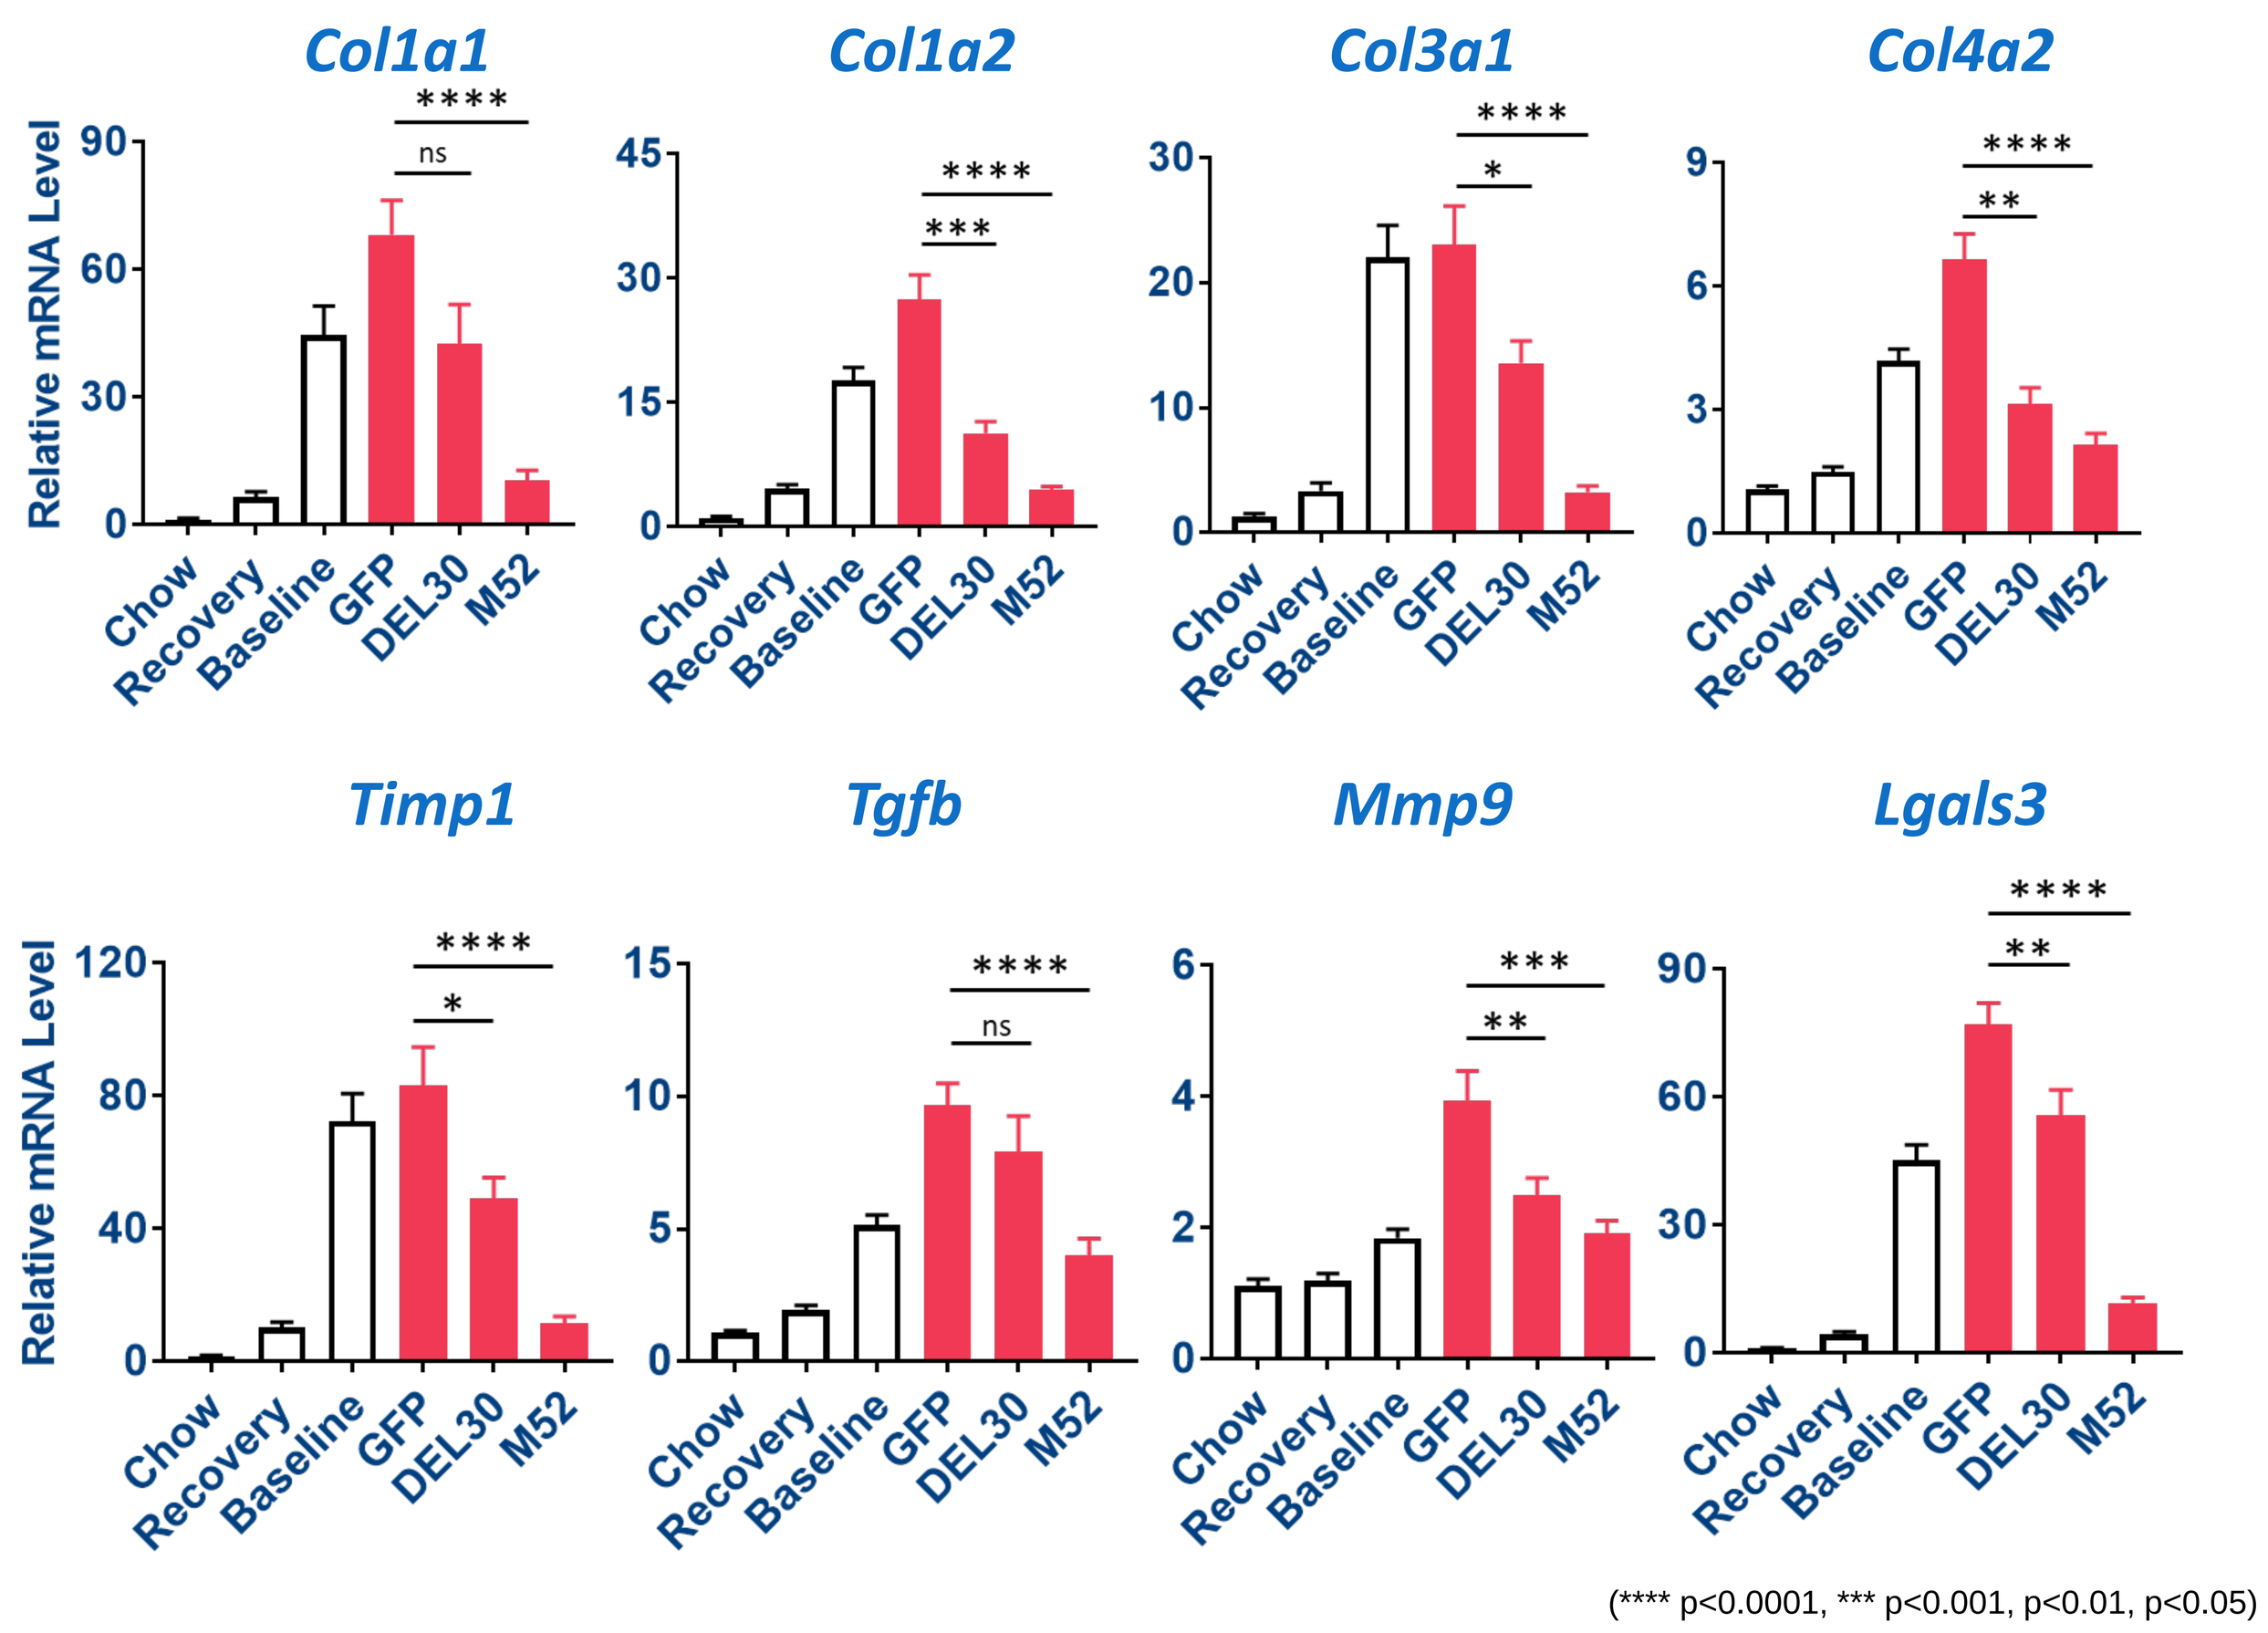
Col1a1
Col1a2
Col3a1
Col4a2
Timp1
Tgfb
Mmp9
Lgals3
Relative mRNA Level
Chow
Recovery
Baseline
GFP
DEL30
M52

(**** p<0.0001, *** p<0.001, p<0.01, p<0.05)

- DEL30 reduces fibrosis with no change in hepatic steatosis
- M52 normalizes hepatic steatosis and reduces fibrosis

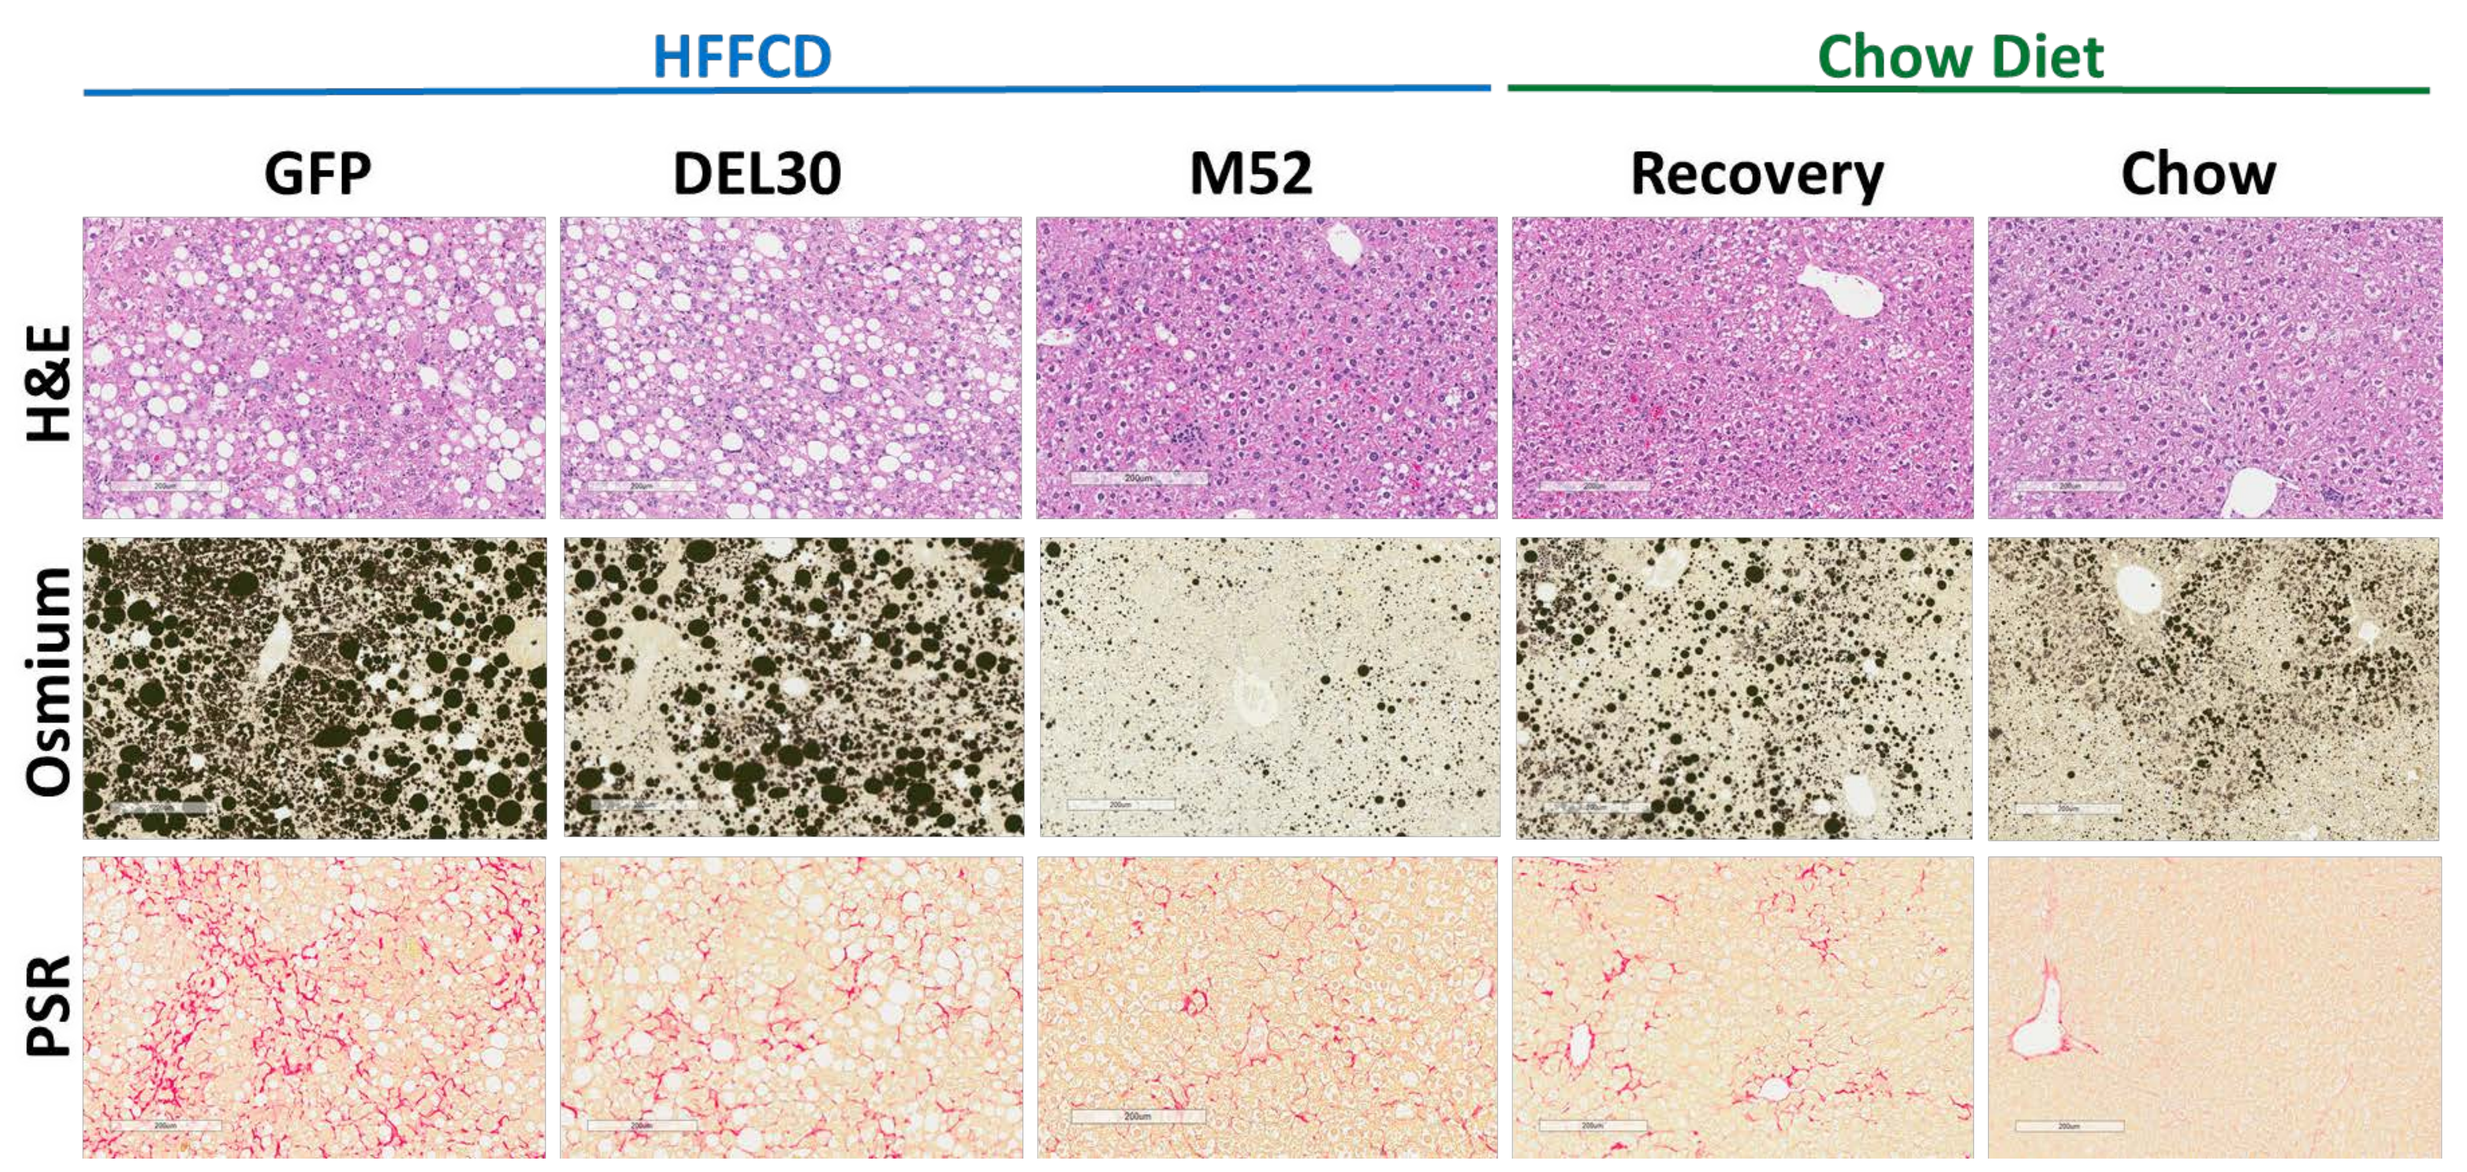
HFFCD
Chow Diet
GFP
DEL30
M52
Recovery
Chow
H&E
Osmium
PSR

- Osmium tetroxide stains lipids
- Morphometric quantification of picro-sirius red staining

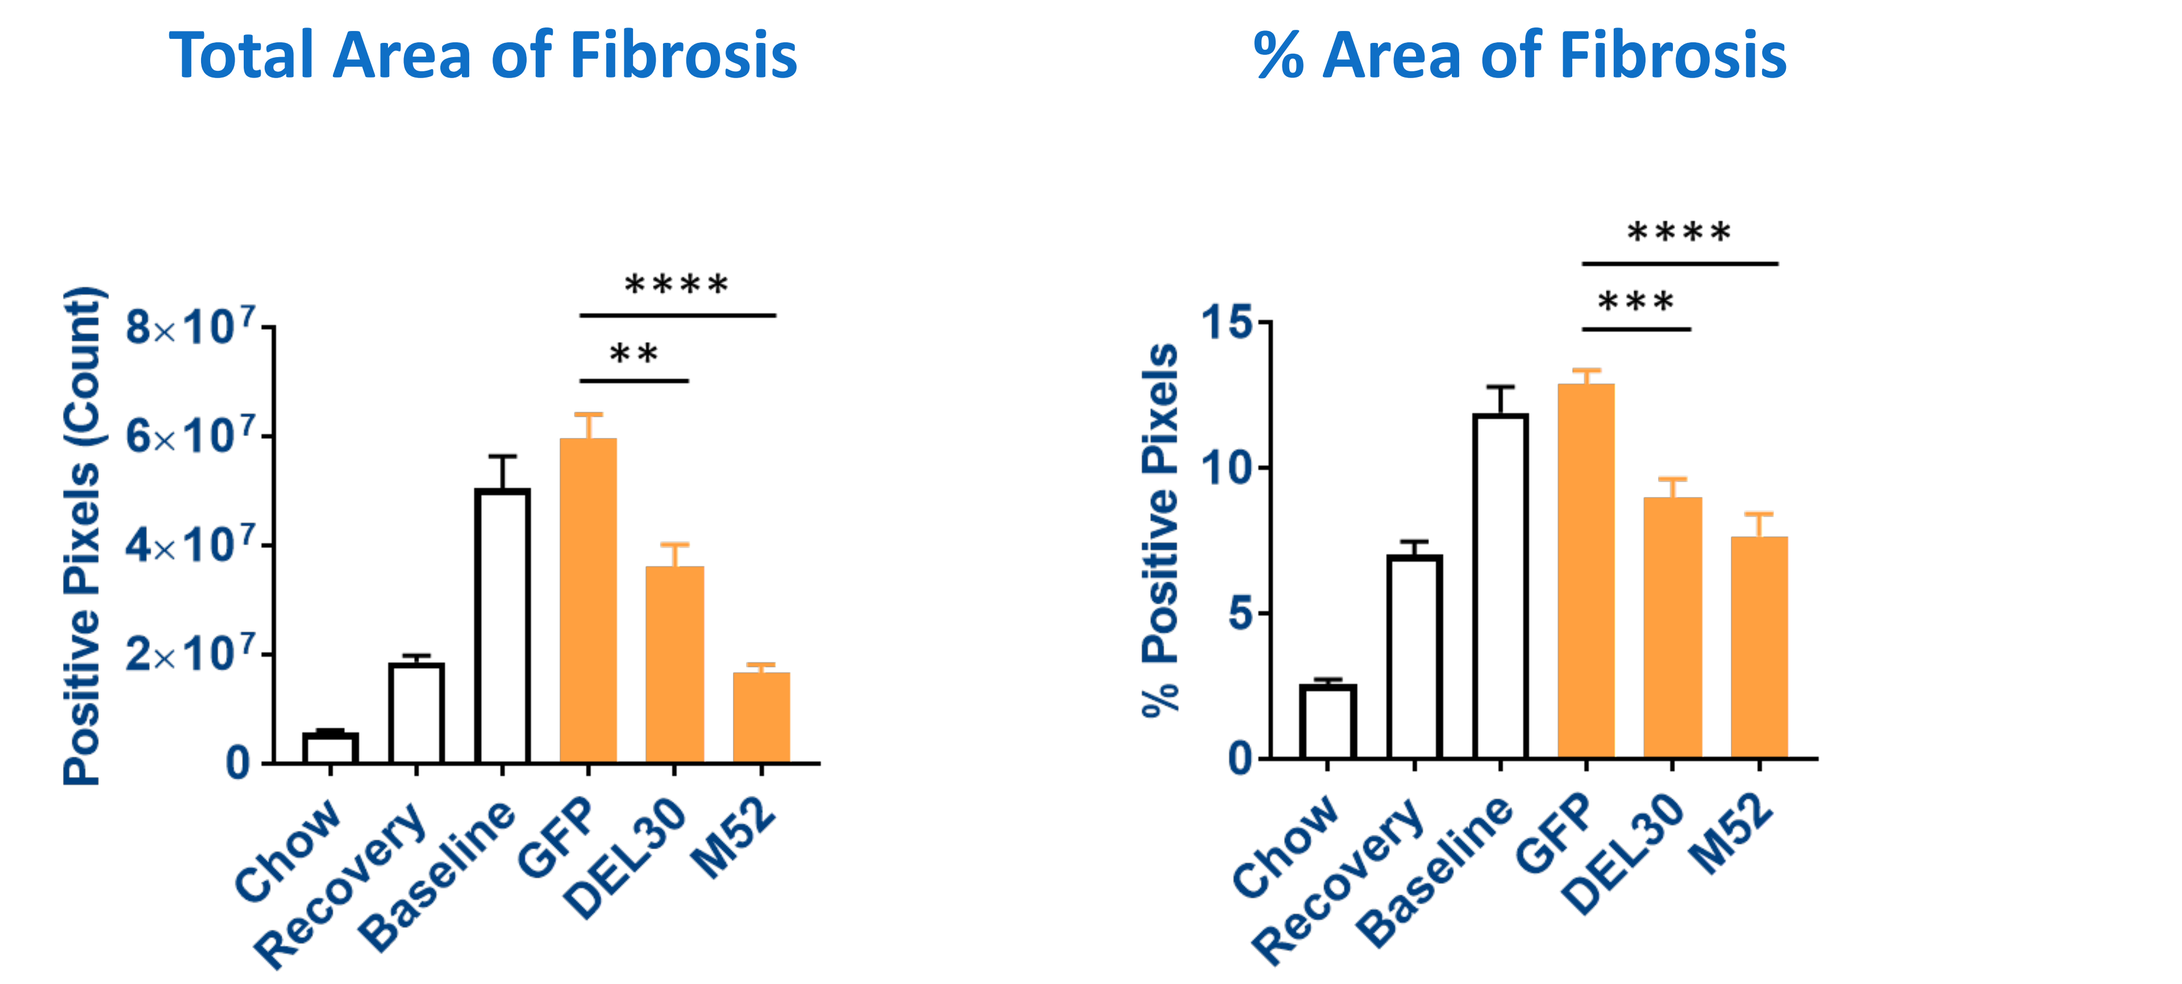
Total Area of Fibrosis
Positive Pixels (Count)
% Area of Fibrosis
% Positive Pixels
Chow
Recovery
Baseline
GFP
DEL30
M52

(**** p<0.0001, *** p<0.001, p<0.01, p<0.05)

Summary of Histopathology Findings

- Mice on HFFCD diet develop NASH phenotype –steatosis, inflammation and fibrosis
- Chow recovery induced by diet replacement reduces steatosis, inflammation and regression of fibrosis

Histological Evaluation (with Mean Group Severity Score)

	Steatosis (0-4)	Inflammation (0-4)	Fibrosis Score (0-6)
Chow Recovery	minimal to mild (1.6 ± 0.92)	minimal (1.1 ± 0.35)	1.3 ± 0.46
GFP	moderate (2.8 ± 0.44)	mild (2.2 ± 0.42)	1.8 ± 0.67
DEL30	marked (3.7 ± 0.5)	minimal (1.0 ± 0.0)	1.2 ± 0.67
M52	minimal (1.0 ± 0.5)	minimal (1.1 ± 0.33)	1.3 ± 0.50

CONCLUSIONS

- Treatment of M52 and DEL30 results in fibrosis regression in diet induced model of NASH (HFFCD model)
- FGFR4 signaling is a primary driver of FGF19-mediated anti-fibrotic activity that is independent of weight loss or decrease in hepatic steatosis
- Additional anti-fibrotic and anti-inflammatory activities of M52 support that other mechanisms (such as FGFR1c activity) further contribute to the improvements in NASH-related liver disease and fibrosis
- These data have important implications for understanding the multiple mechanism of action responsible for the improvements in histology observed with NGM282 patients

References:

1. *Kliewer et al., Dig Dis 2015; 33:327-331*
2. *Degirolamo et al., Nat Rev Drug Discov 2016; 15:51-69*
3. *Luo et al., EASL 2015*
4. *Ling et al., EASL 2015*
5. *Zhou et al., Hepatology Comm 2017; 1:1024-1042*
6. *Zhou et al., Cancer Res 2014; 74:3306-3316*
7. *Wu et al., Proc Natl Acad Sci U S A. 2009 Aug 25; 106(34): 14379–14384*
8. *Harrison et al., Am J Gastroenterol 2002; 97:2714-2724*
9. *Clapper et al., Am J Physiol Gastrointest Liver Physiol 2013; 305:G483-G495*
10. *Harrison et al., Hepatology. 2015;62:1652–1655*
11. *Tølbøl et al., World J Gastroenterol. 2018; 24(2): 179-194*

DISCLOSURES

- This study was funded by NGM Biopharmaceuticals, Inc.
- Maria Deato, Brian Ko, Emily Snyder, Bernard Allan, and Hui Tian are employees and stockholders of NGM Biopharmaceuticals, Inc.