618

High HBV and HIV Suppression With Treatment of HIV/HBV Coinfection in B/F/TAF Studies

Jürgen K. Rockstroh,[1] Paul E. Sax,[2] Eric S. Daar,[3] Sharon Walmsley,[4] Kim Workowski,[5] Chloe Orkin,[6] Jose R. Arribas,[7] Edwin DeJesus,[8] David Wohl,[9] Jean-Michel Molina,[10] David Piontkowsky,[11] Xuelian Wei,[11] Hal Martin,[11] Andrew Cheng,[11] Erin Quirk[11]

[1]Universität Bonn, Germany; [2]Brigham and Women's Hospital, Boston, MA; [3]Los Angeles Biomedical Research Institute at Harbor-UCLA Medical Center, Torrance, CA; [4]Toronto General Hospital, Toronto, Ontario, Canada; [5]Emory University, Atlanta, GA; [6]Barts Health NHS Trust, The Royal London Hospital, Ambrose King Centre, London, UK; [7]Hospital Universitario la Paz, Madrid, Spain; [8]Orlando Immunology Center, Orlando, FL; [9]University of North Carolina at Chapel Hill; [10]Hôpital Saint Louis, Paris, France; [11]Gilead Sciences, Inc., Foster City, CA

Gilead Sciences, Inc.
333 Lakeside Drive
Foster City, CA 94404
800-445-3235

Introduction

- Hepatitis B virus (HBV) is a common coinfection in HIV patients
 - Estimates range from 3 to 6 million individuals coinfected with HIV and HBV, and coinfection rates are as high as 25% in areas where both viruses are endemic[1]
- Coinfection worsens morbidity and mortality synergistically
 - HIV/HBV-coinfected patients have higher HBV DNA levels, progress to chronic HBV 5 times more quickly than HBV mono-infected patients, and have higher risks of cirrhosis and hepatocellular carcinoma
- Tenofovir alafenamide (TAF) is active against HBV and is approved for treatment of HBV as a single agent[2,3]
- Current HIV guidelines recommend TAF or tenofovir disoproxil fumarate (TDF) as components of regimens for treatment of patients coinfected with HIV and HBV[4-6]
- We report HBV and HIV outcomes in antiretroviral treatment (ART)-naïve and -experienced HIV/HBV-coinfected patients enrolled in 4 studies of coformulated bictegravir/emtricitabine/TAF (B/F/TAF)

Objectives

- To report HBV and HIV outcomes in ART-naïve and -experienced HIV/HBV-coinfected patients enrolled in 4 studies of coformulated B/F/TAF

Methods

HBV Assessments and Definition of Active HBV Infection

- All participants enrolled in the 4 studies were tested for HBV serologies at screening and Week 48
 - Active HBV infection at study entry (prevalent HBV) was defined as HBV surface antigen (HBsAg) positive on or prior to 1st dose date, or HBsAg negative, HBV surface antibody (HBsAb) negative, HBV core antibody (HBcAb) positive, and quantifiable HBV DNA (ie, HBV DNA ≥20 IU/mL) on or prior to 1st dose date
- Participants with active HBV infection at screening were excluded from Studies 1489 and 1844
 - Comparator arms did not include guideline-recommended nucleotide reverse transcriptase inhibitors (NRTIs) for treatment of HBV in HIV-infected participants

HBV Outcomes for Participants With Active HIV/HBV Coinfection at Study Entry: Studies 1490 and 1878

- For participants with active HBV infection on entry into Studies 1490 and 1878, the proportion with HBV DNA <29 IU/mL at Week 48 was assessed using a missing=excluded data imputation method to assess HBV efficacy of B/F/TAF, dolutegravir; (DTG) + F/TAF, and F/TDF + boosted protease inhibitor (PI) for participants with HIV/HBV coinfection

Identification of Potential Incident HBV Cases: Acquired HBV Infection in Studies 1489, 1490, 1844, and 1878

- Potential incident HIV/HBV coinfection was defined as participants enrolled in any of the 4 studies who were not HIV/HBV-coinfected at baseline (BL) and met the following criteria:
 - HBsAg positive after 1st dose date;
 - HBsAg negative, HBsAb negative, HBcAb positive, and quantifiable HBV DNA (ie, ≥20 IU/mL) after 1st dose date; or
 - Experienced adverse events (AEs) consistent with HBV infection after 1st dose date (eg, acute HBV, HBV infection, and HBV test positive)
- HBV serology and DNA data for participants who met criteria for potential incident HIV/HBV coinfection were reviewed to determine whether incident HBV infection occurred

Study 1490 Results: HIV/HBV-Coinfected Treatment-Naïve Participants

Treatment-Naïve Study Designs[7,8]

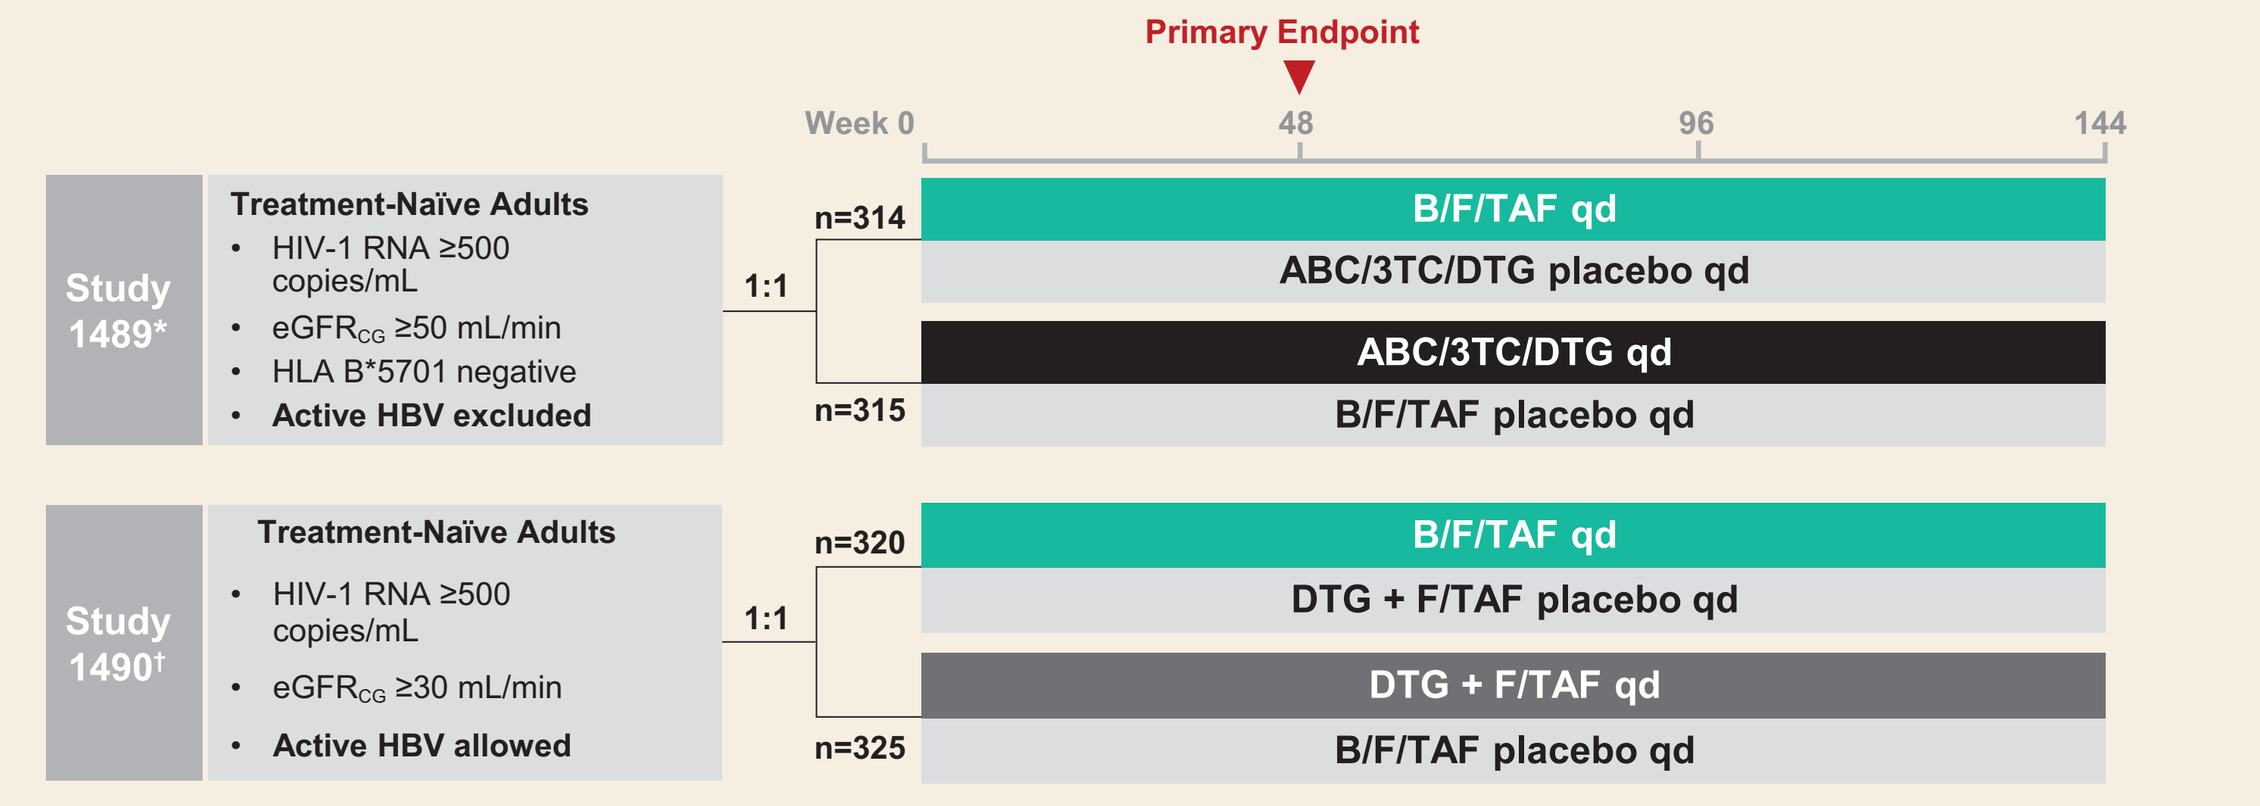
Primary Endpoint
Week 0
48
96
144
Study 1489*
Treatment-Naïve Adults
HIV-1 RNA ≥500 copies/mL
eGFR$_{CG}$ ≥50 mL/min
HLA B*5701 negative
Active HBV excluded
1:1
n=314
B/F/TAF qd
ABC/3TC/DTG placebo qd
ABC/3TC/DTG qd
B/F/TAF placebo qd
n=315
Study 1490†
Treatment-Naïve Adults
HIV-1 RNA ≥500 copies/mL
eGFR$_{CG}$ ≥30 mL/min
Active HBV allowed
1:1
n=320
B/F/TAF qd
DTG + F/TAF placebo qd
DTG + F/TAF qd
B/F/TAF placebo qd
n=325

*NCT02607930; †NCT02607956. 3TC, lamivudine; ABC, abacavir; eGFR$_{CG}$, estimated glomerular filtration rate by Cockcroft-Gault; HLA, human leukocyte antigen.

- Primary endpoint: proportion with HIV-1 RNA <50 copies/mL at Week 48
 - Noninferiority margin of 12% prespecified based on FDA-defined snapshot algorithm
- Participants with active HBV infection were excluded from Study 1489 and were allowed to enroll in Study 1490

Disposition

- 14/645 participants (2%) had HIV/HBV coinfection at screening, and were randomized and treated (n=8 with B/F/TAF; n=6 with DTG + F/TAF)
 - Male: n=12 (86%); nonwhite: n=8 (57%)
 - n=5 (36%) had HBV DNA >170,000,000 IU/mL: 4 were hepatitis B e antigen (HBeAg) positive at BL (1 had missing HBe serologies)
 - HBsAg positive: n=12 (86%)
 - 4/10 (40%) with BL HBe serologies were HBeAg positive
- HIV/HBV participant disposition:
 - 1/14 did not have Week 48 HBV DNA assessments: randomized to B/F/TAF group and discontinued by investigator discretion at Day 68 (moved out of state)
 - n=13 had post-BL HBV DNA assessments

HBV and HIV Assessments at Baseline and Week 48

		Baseline					Week 48			
	Participant	ALT, U/L	HBV DNA, IU/mL	HBsAg	HBeAg	HIV-1 RNA, Copies/mL	HBV DNA, IU/mL	HBsAg	HBeAg	HIV-1 RNA, Copies/mL
B/F/TAF	1	30	>170,000,000	+	N/A	47,000	28	+	–	Not detected
	2	118	>170,000,000	+	+	97,500	<20	+	–	Not detected
	3*	25	>170,000,000	+	+	719,000	N/A	N/A	N/A	N/A
	4	16	354,000	+	N/A	13,700	Not detected	–	–	Not detected
	5	12	1930	+	–	2220	Not detected	+	N/A	Not detected
	6	18	259	+	–	40,200	Not detected	+	–	Not detected
	7	12	120	–	–	57,200	Not detected	N/A	N/A	<20
	8	38	76	–	–	30,700	Not detected	–	–	22
DTG + F/TAF	9	47	>170,000,000	+	+	284,000	303	+	+	37
	10	67	>170,000,000	+	+	721	80	+	+	Not detected
	11	40	<20	+	–	227,000	<20	–	–	<20
	12	23	5210	+	N/A	20,700	<20	+	–	<20
	13	20	1050	+	N/A	39,500	Not detected	+	N/A	Not detected
	14	26	980	+	–	150,000	Not detected	+	–	<20

*On study, but had no data in Week 48 window. ALT, alanine aminotransferase; N/A, not available.

HBV Outcomes and HBV DNA Results at Week 48

Participants, n/n (%)	Total Randomized to B/F/TAF or DTG + F/TAF With HBV DNA Results at Week 48 n=13
HBV DNA <29 IU/mL	11/13 (85)
HBV DNA ≥29 IU/mL	2/13 (15)
HBsAg conversion	2/12 (17)
HBeAg conversion	1/4 (25)

HBV and HIV Outcomes

- Of 13 treatment-naïve participants with HIV/HBV at BL and HBV DNA results at Week 48, 11 (85%) achieved HBV DNA <29 IU/mL at Week 48
- 2/13 participants (17%) had HBV DNA ≥29 IU/mL at Week 48
 - Both had HBV DNA >170,000,000 IU/mL at BL and were HBeAg positive
 - Week 48 HBV DNA results were 303 and 80 IU/mL (study drug adherence 100% and 99%, respectively)
- 2/12 participants (15%) experienced HBsAg loss: both seroconverted to HBsAb-positive status at Week 48
 - 1 was HBsAg and HBcAb positive, but had HBV DNA <20 IU/mL at BL, and was HBsAg negative and HBsAb positive at Week 48, indicating HBsAg loss and seroconversion during the study
- Of 4 HBeAg-positive participants at BL, 1 (25%) experienced HBeAg loss at Week 48 (HBe antibody positive at Week 48)
- All 13 participants had HIV-1 RNA <50 copies/mL at Week 48

Study 1878 Results: HIV/HBV-Coinfected Treatment-Experienced Participants

Switch Study Designs

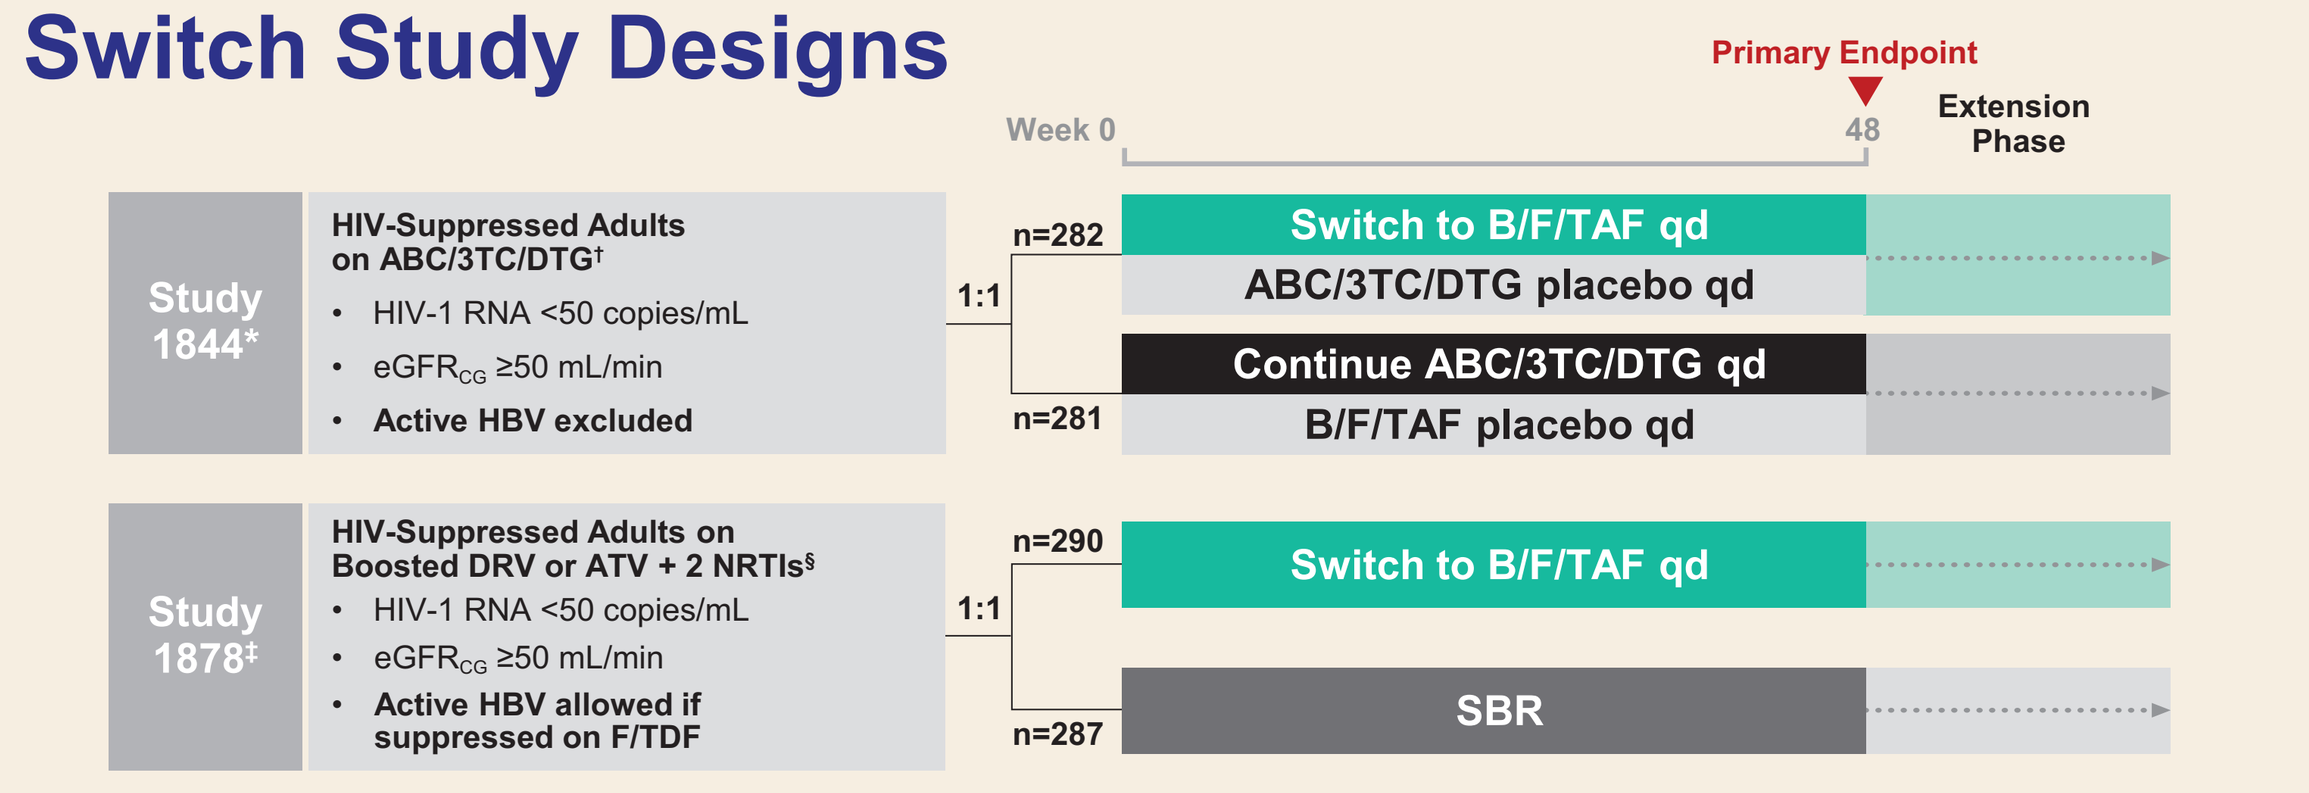
Primary Endpoint
Week 0
48
Extension Phase
Study 1844*
HIV-Suppressed Adults on ABC/3TC/DTG†
HIV-1 RNA <50 copies/mL
eGFR$_{CG}$ ≥50 mL/min
Active HBV excluded
1:1
n=282
Switch to B/F/TAF qd
ABC/3TC/DTG placebo qd
Continue ABC/3TC/DTG qd
B/F/TAF placebo qd
n=281
Study 1878‡
HIV-Suppressed Adults on Boosted DRV or ATV + 2 NRTIs§
HIV-1 RNA <50 copies/mL
eGFR$_{CG}$ ≥50 mL/min
Active HBV allowed if suppressed on F/TDF
1:1
n=290
Switch to B/F/TAF qd
SBR
n=287

*NCT02603120; †Suppressed on regimen for ≥3 mo; ‡NCT02603107; §Suppressed on regimen for ≥6 mo (NRTIs: ABC/3TC or F/TDF). ATV, atazanavir; DRV, darunavir; SBR, stay on BL regimen.

- Primary endpoint: proportion with HIV-1 RNA ≥50 copies/mL at Week 48
- Participants with active HBV infection were excluded from Study 1844 and were allowed to enroll in Study 1878

Disposition

- 14/577 participants (2%) had HIV/HBV coinfection at BL, and were randomized and treated
 - Male: n=13 (93%); nonwhite: n=6 (43%)
 - HBsAg positive: 12/14 (86%)
 - HBsAg negative: 2/14 (14%); both were HBsAb negative and HBcAb positive, and had HBV DNA <1000 IU/mL at BL
 - HBeAg positive: n=4 (29%)
 - Randomized treatments:
 - B/F/TAF: 8/8 participants (100%) had HBV DNA <29 IU/mL at BL
 - SBR: 4/6 (67%) had HBV DNA <29 IU/mL at BL
 - 1 HBeAg-positive participant taking F/TDF + ritonavir-boosted ATV (ATV/r) had HBV DNA 8300 IU/mL and HIV-1 <50 copies/mL at BL
 - 1 participant taking ABC/3TC + ATV/r had HBV DNA >170,000,000 IU/mL and HIV-1 RNA <50 copies/mL at BL
- 2 participants did not have Week 48 HBV DNA assessments
 - 1 randomized to stay on ABC/3TC + ATV/r discontinued after Day 1 (lost to follow-up)
 - 1 randomized to stay on F/TDF + ATV/r discontinued study drug and switched to open-label B/F/TAF before Week 48 assessment
- 12 participants had Week 48 HBV DNA assessments

HBV and HIV Outcomes at Week 48

HBV DNA <29 IU/mL, n/n (%)	B/F/TAF, n=8	SBR, n=6
BL	8	4
Week 48	8/8 (100)	4/4 (100)

- 2 participants in the SBR group had HBV DNA ≥29 IU/mL at BL
 - 1 randomized to continue ABC/3TC + ATV/r had HBV DNA >170,000,000 IU/mL and HIV-1 RNA <50 copies/mL (target not detected) at BL, and discontinued the study due to lost to follow-up on Day 1
 - Resistance analysis of BL HBV DNA sample showed resistance to adefovir (A181T/V, N236T), entecavir (T184ANY, S202ANY, M204I/V, M250ANY), and 3TC (A181T/V, M204I/S/V)
 - Other HBV resistance mutations detected: L80I/V, I169T, V173L, L180M, and A194T
 - 1 HBeAg-positive participant randomized to continue F/TDF + ATV/r had HBV DNA 8300 IU/mL and HIV-1 RNA <50 copies/mL at BL
 - At Week 48, HBV DNA was >170,000,000 IU/mL and HIV-1 RNA was 16,000 copies/mL when switching to B/F/TAF at Week 48
 - 9 days later, HBV DNA was 599,000 IU/mL and HIV-1 RNA was <50 copies/mL while continuing B/F/TAF
- All 12 participants with Week 48 data were HBsAg positive at BL and Week 48
- 4 participants were HBeAg positive at BL: 2 on B/F/TAF and 2 on SBR
 - 1 participant in the SBR group discontinued at Day 1, 1 participant in the B/F/TAF group was HBeAg negative at Week 48, and 2 participants remained HBeAg positive at Week 48 (1 on B/F/TAF and 1 on SBR)
- All 12 participants with Week 48 data had HIV-1 RNA <50 copies/mL at Week 48
- The overall incidence of confirmed HBV viremia in HBV-susceptible participants was 0.1% (0% in B/F/TAF, F/TAF, and F/TDF arms, and 0.4% in ABC/3TC/DTG arms)

Incident HBV Infections

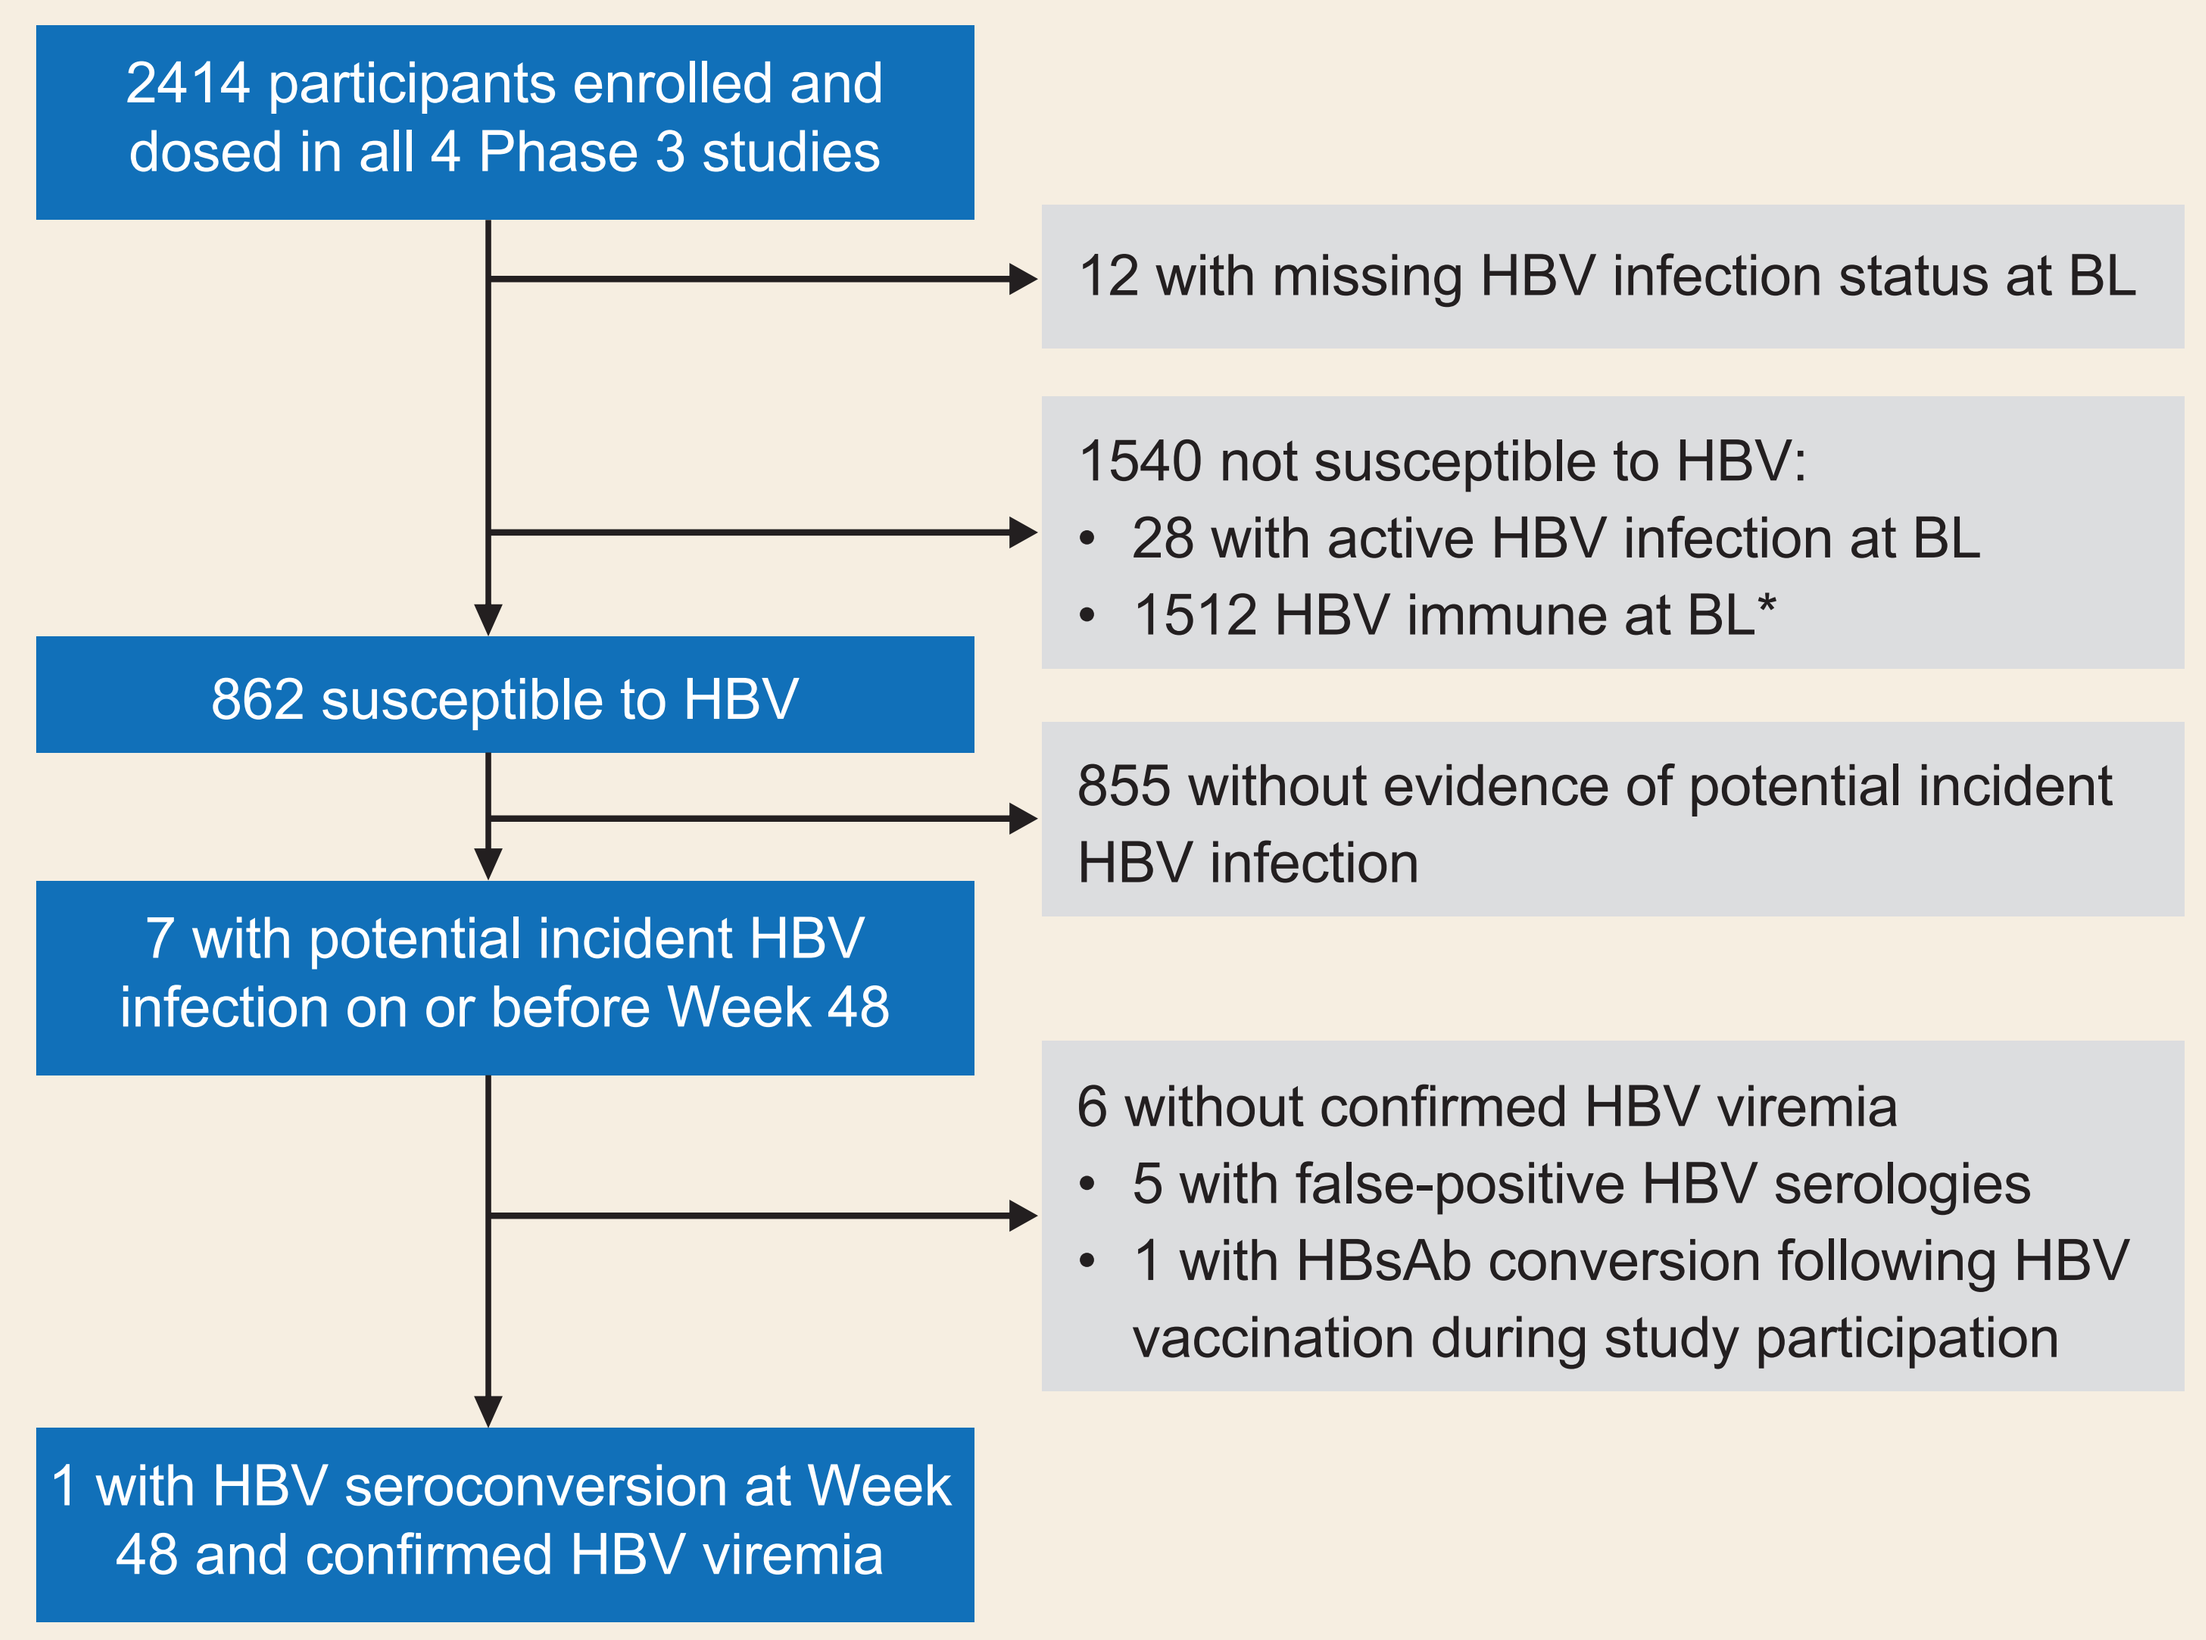
2414 participants enrolled and dosed in all 4 Phase 3 studies
12 with missing HBV infection status at BL
1540 not susceptible to HBV:
28 with active HBV infection at BL
1512 HBV immune at BL*
862 susceptible to HBV
855 without evidence of potential incident HBV infection
7 with potential incident HBV infection on or before Week 48
6 without confirmed HBV viremia
5 with false-positive HBV serologies
1 with HBsAb conversion following HBV vaccination during study participation
1 with HBV seroconversion at Week 48 and confirmed HBV viremia

*Defined as participants not HBV coinfected at BL who had positive BL HBsAb.

- 1 participant randomized to receive DTG/ABC/3TC developed incident HBV infection
 - At BL, HIV-1 RNA was 406,000 copies/mL, and HBsAg, HBsAb, and HBcAb were negative
 - At Week 48, HIV-1 RNA was <50 copies/mL (ie, 31) and HBsAg was positive (confirmed on repeat)
 - HBV DNA 78 IU/mL at Week 48 and repeated 16 days later at 97 IU/mL
 - HBsAb and HBcAb negative at Week 48 (confirmed on repeat)
 - Participant continued on DTG/ABC/3TC and HBV DNA was not detected at Week-60 visit

Incident HBV Infection Case: HIV RNA, HBV Serologies, and HBV DNA Results

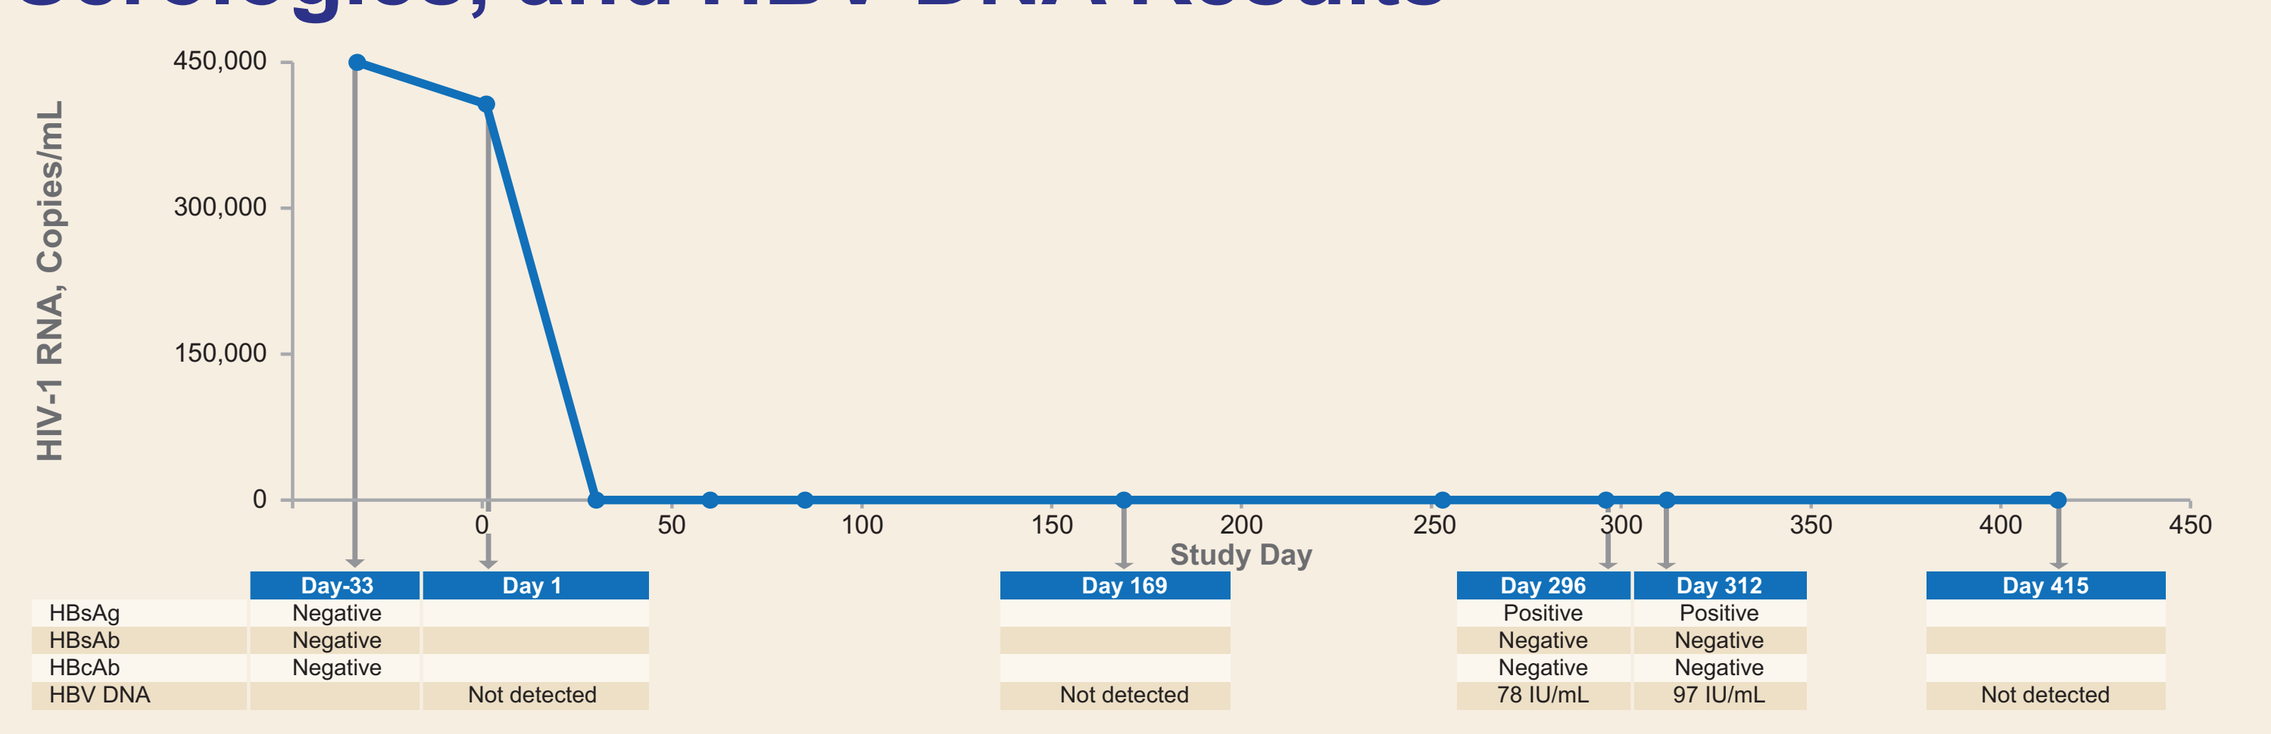
HIV-1 RNA, Copies/mL
450,000
300,000
150,000
0
0
50
100
150
200
250
300
350
400
450
Study Day
HBsAg
HBsAb
HBcAb
HBV DNA
Day-33
Negative
Negative
Negative
Day 1
Not detected
Day 169
Not detected
Day 296
Positive
Negative
Negative
78 IU/mL
Day 312
Positive
Negative
Negative
97 IU/mL
Day 415
Not detected

Conclusions

- B/F/TAF- and F/TAF-containing regimens produced robust HBV antiviral responses in treatment-naïve participants with HIV/HBV coinfection
 - 85% of participants (11/13) achieved HBV DNA <29 IU/mL at Week 48
 - No participant developed HBV resistance to FTC or TAF; the 2 without HBV DNA <29 IU/mL at Week 48 had decreases in HBV DNA from >170,000,000 IU/mL at baseline to <400 IU/mL at Week 48
 - 15% had HBsAg loss and seroconverted to HBsAb-positive status at Week 48
 - 25% of HBeAg-positive participants experienced HBeAg loss at Week 48
 - 100% had HIV-1 RNA <50 copies/mL at Week 48
- B/F/TAF- and F/TDF-containing regimens maintained HIV-1 virologic suppression in HIV/HBV-coinfected participants with HIV-1 suppression at study entry
 - 100% of participants with HBV suppression at baseline had HBV DNA <29 IU/mL at Week 48
 - 1/4 (25%) who were HBeAg positive at baseline had HBeAg conversion at Week 48
 - 100% with Week 48 HIV-1 RNA results had HIV-1 RNA <50 copies/mL at Week 48
- No participant treated with B/F/TAF, or an F/TAF- or F/TDF-containing regimen acquired HBV infection during the studies
 - 1 treated with ABC/3TC/DTG had incident HBV infection with confirmed HBV viremia at Week 48
- The results confirm findings from prior studies of ART with anti-HBV activity in patients with HIV/HBV coinfection:
 - Higher HBsAb seroconversion rates than in chronic HBV mono-infection[9]
 - Not all patients become undetectable after 48 wk in the setting of high HBV DNA at baseline[10]
 - To date, there is no evidence of HBV resistance to F/TAF-containing regimens
- B/F/TAF may be a treatment option for HIV-1–infected patients with HBV coinfection
 - Further studies of HBV treatment and prevention with B/F/TAF and other F/TAF-containing ART regimens are warranted in HIV/HBV-coinfected patients

References

1. Joint United Nations Programme on HIV/AIDS (UNAIDS). Global report: UNAIDS report on the global AIDS epidemic, 2013; http://files.unaids.org/en/media/unaids/contentassets/documents/epidemiology/2013/gr2013/UNAIDS_Global_Report_2013_en.pdf; 2. Vemlidy [package insert]. Foster City, CA: Gilead Sciences, Inc., 4/17; 3. Vemlidy [SMPC]. Cambridge, UK: Gilead Sciences International Ltd., 2/18; 4. AIDS info. http://www.aidsinfo.nih.gov/ContentFiles/Adul-tandAdolescentGL.pdf. Jan 2016; 5. European AIDS Clinical Society. Guidelines. Version 9.0, Oct 2017; www.eacsociety.org/files/guidelines_9.0-english.pdf; 6. Gunthard HF, et al. JAMA 2016;316:191-210; 7. Gallant J, et al Lancet 2017;390:2063-72; 8. Sax PE, et al. Lancet 2017;390:2073-82; 9. Boesecke C, et al. CROI 2017, abstr 580; 10. Price H, et al. PLoS One 2013;8:e68152.

Acknowledgments

We extend our thanks to the participants, their families, and all participating study investigators and staff. These studies were funded by Gilead Sciences, Inc.

Presented at Conference on Retroviruses and Opportunistic Infections, March 4–7, 2018, Boston, MA

©2018 Gilead Sciences, Inc. All rights reserved.