P-1925

Incidence of Long COVID Symptoms During the Year Post Admission Among Patients Hospitalized for COVID-19

Mark Berry[1], Valentina Shvachko[1], Amanda Kong[2], Rohan Shah[2], Gina Brown[1], Anand Chokkalingam[1]

[1]Gilead Sciences, Inc., Foster City, CA, USA; [2]Aetion, Inc., New York, NY, USA

Mark Berry
mark.berry1@gilead.com

Copies of this poster obtained through QR (Quick Response) are for personal use only and may not be reproduced without written permission of the authors

Conclusions

- The incidence of Long COVID–associated symptoms/conditions tended to decline over time, including among patients who had none of the conditions at baseline, indicating that these outcomes may have resulted from COVID-19 infection
- The incidence of certain symptoms/conditions (hair loss and hypoxemia) declined more steeply than others, which could have been due to an improvement in symptoms or trends in patient symptom reporting over the yearlong period

Plain Language Summary

- People who have had COVID-19 can have symptoms that may continue or even begin after infection, a condition known as Long COVID
- There is no widely accepted definition of Long COVID, as patients may experience a variety of symptoms that can last for months or years
- To better understand Long COVID, more data are needed about how often symptoms occur and how long symptoms might last
- This study used insurance and hospital data to identify common Long COVID symptoms over the course of a year after COVID-19 hospitalization
- Long COVID symptoms generally declined over time, even in patients who did not have any pre-existing conditions, suggesting a connection to COVID-19 infection
- Symptoms like hair loss and hypoxemia decreased the most over time

Introduction

- COVID-19 symptoms can last for weeks to months after the initial infection period, or can develop later, with approximately 20% of patients with COVID-19 experiencing symptoms for ≥5 weeks and 10% for ≥12 weeks[1,2]
- Symptoms/conditions associated with post-COVID conditions, or Long COVID, are highly variable and can be an exacerbation of existing conditions or newly developed symptoms
- There is currently no standardized case definition for Long COVID,[3,4] and it remains unknown how long different symptoms and health conditions may persist after infection[5]
- This study used insurance claims and hospital chargemaster data to characterize the incidence of common symptoms/conditions that may be associated with Long COVID during the first year after admission among patients hospitalized with COVID-19

Methods

- Data for this retrospective, observational analysis were obtained from HealthVerity Real-Time Insights and Evidence, a unified database of medical and pharmacy claims and hospital chargemaster data for patients in the United States
- The analysis included patients aged ≥12 years with a recorded COVID-19 diagnosis during an inpatient hospitalization from May 1, 2020, to January 21, 2024, with ≥31 days of follow-up who had ≥365 days of baseline enrollment in closed medical claims prior to hospitalization; a 30-day gap in enrollment was permitted
- Patients with a hospitalization of <2 days, clinical trial participation, diagnosis related to pregnancy, or prior diagnosis of Long COVID were excluded
- The presence of pre-existing symptoms/conditions at baseline was assessed from 365 days through 15 days prior to the COVID-19 hospitalization date (index date)
- Patients were considered to have Long COVID or a Long COVID symptom if they had ≥1 of the following symptoms/conditions, derived from *International Classification of Diseases, Tenth Revision, Clinical Modification* diagnosis codes, recorded from 90 days to 270 days post hospital admission:
 — Arrhythmias, brain fog, cerebrovascular disease, chest pain, cognitive dysfunction, cough, diabetes, diarrhea, dysautonomia, dyspnea/breathlessness, fatigue, hair loss, headache, hyperlipidemia, hypoxemia, insomnia, ischemic heart disease, joint pain/arthralgia, the specific Long COVID diagnosis code (U09.9), muscle pain/myalgia, muscle weakness, nephropathy, neuropsychiatric features, obesity, smell disturbance/anosmia, tachycardia, taste disturbance/dysgeusia/ageusia, thromboembolic disease
- The incidence of Long COVID outcomes was assessed based on diagnosis codes during 2 periods: 31 to 197 days and 198 to 365 days post hospital admission
- Incidence rates per 1000 person-years with 95% CIs were calculated for all hospitalized patients, patients with no record of any of the symptoms/conditions in the year prior to admission, and patients with immunocompromising conditions[6]
- The percent change in incidence rates between time periods was calculated for each symptom

Results

Participants

- A total of 92,913 patients from the HealthVerity database met the study criteria and were included in the analysis (**Figure 1**)

Figure 1. Patient Attrition

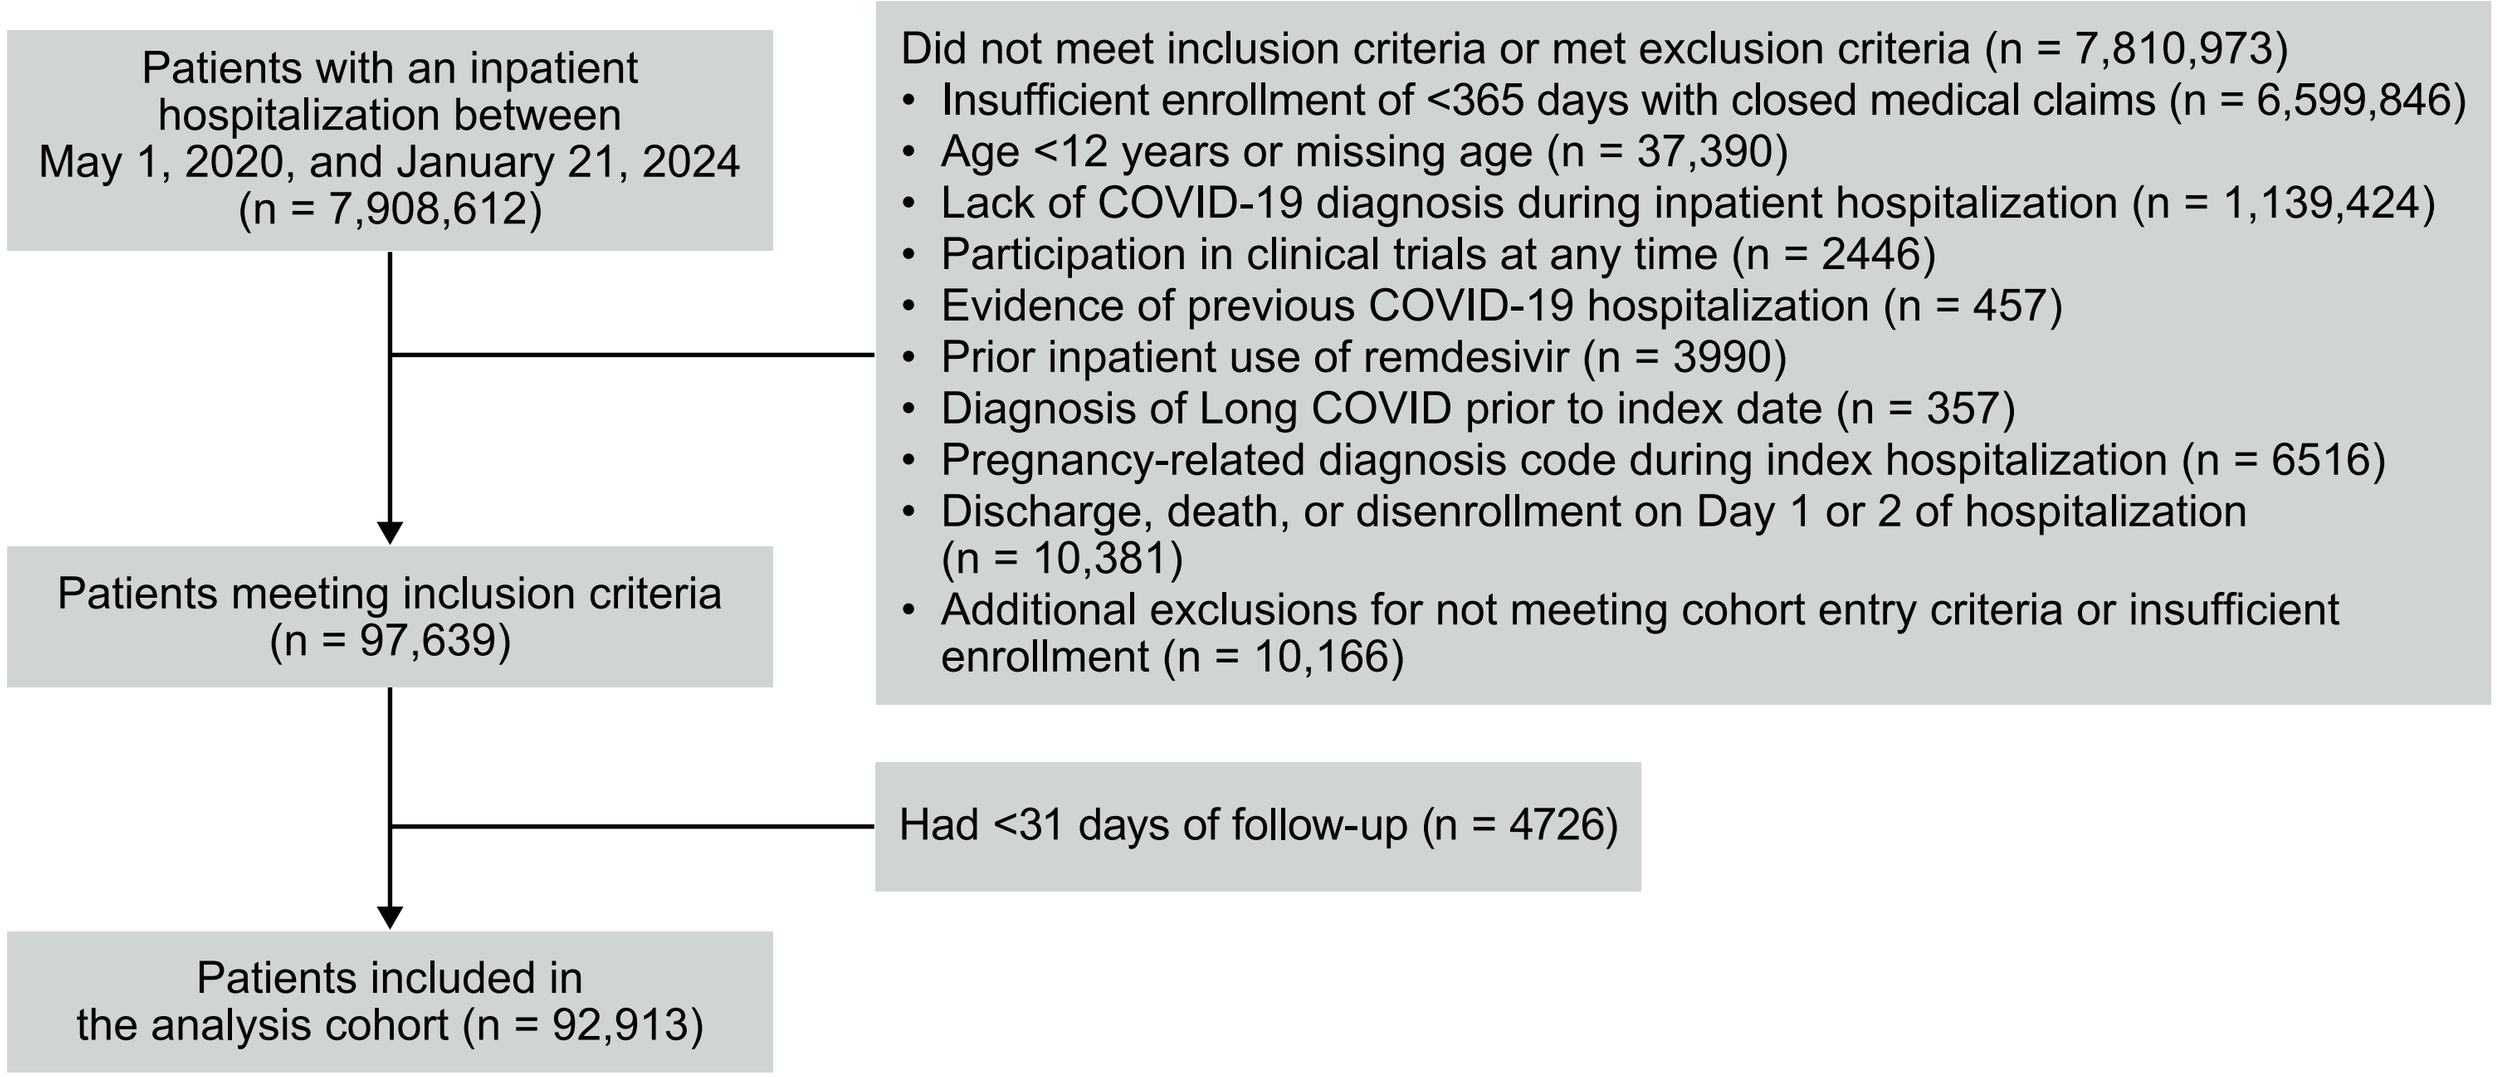

Patients with an inpatient hospitalization between May 1, 2020, and January 21, 2024 (n = 7,908,612)
Did not meet inclusion criteria or met exclusion criteria (n = 7,810,973)
• Insufficient enrollment of <365 days with closed medical claims (n = 6,599,846)
• Age <12 years or missing age (n = 37,390)
• Lack of COVID-19 diagnosis during inpatient hospitalization (n = 1,139,424)
• Participation in clinical trials at any time (n = 2446)
• Evidence of previous COVID-19 hospitalization (n = 457)
• Prior inpatient use of remdesivir (n = 3990)
• Diagnosis of Long COVID prior to index date (n = 357)
• Pregnancy-related diagnosis code during index hospitalization (n = 6516)
• Discharge, death, or disenrollment on Day 1 or 2 of hospitalization (n = 10,381)
• Additional exclusions for not meeting cohort entry criteria or insufficient enrollment (n = 10,166)
Patients meeting inclusion criteria (n = 97,639)
Had <31 days of follow-up (n = 4726)
Patients included in the analysis cohort (n = 92,913)

The index date was the date of COVID-19 hospitalization.
Closed payer claims data were derived from the insurance provider (or payer) and captured nearly all events that occurred during a patient's enrollment period, including medical and pharmacy visits and transactions for both retail and specialty settings.

- The most common Long COVID–associated conditions that were present at baseline were hyperlipidemia (51%), neuropsychiatric features (49%), and diabetes (41%; **Table 1**)

Table 1. Demographic Characteristics and Baseline Symptoms/Conditions Associated With Long COVID for All Hospitalized Patients

Characteristic	Patients (n = 92,913)
Age, years, median (IQR)	60.0 (47.0-71.0)
Length of stay, days, median (IQR)	7.0 (4.0-11.0)
Sex, n (%)	
Female	47,592 (51)
Male	45,321 (49)
Immunocompromised status,[a] n (%)	39,185 (42)
Baseline conditions associated with Long COVID,[b] n (%)	
Arrhythmias	21,498 (23)
Brain fog	19,839 (21)
Cerebrovascular disease	13,822 (15)
Chest pain	23,688 (26)
Cognitive dysfunction	20,104 (22)
Cough	19,180 (21)
Diabetes	37,664 (41)
Diarrhea	8334 (9)
Dysautonomia	182 (<1)
Dyspnea/breathlessness	23,518 (25)
Fatigue	24,824 (27)
Hair loss	531 (<1)
Headache	11,552 (12)
Hyperlipidemia	47,775 (51)
Hypoxemia	12,414 (13)
Insomnia	9743 (11)
Ischemic heart disease	8829 (10)
Joint pain/arthralgia	25,922 (28)
Muscle pain/myalgia	4536 (5)
Muscle weakness	12,852 (14)
Neuropsychiatric features	45,239 (49)
Obesity	32,105 (35)
Smell disturbance/anosmia	146 (<1)
Tachycardia	10,069 (11)
Taste disturbance/dysgeusia/ageusia	127 (<1)
Thromboembolic disease	6639 (7)

[a]Immunocompromised condition during the past 12 months (symptomatic HIV infection, hematologic and solid malignancy, organ transplant, rheumatologic/inflammatory, or other immune conditions).
[b]Measured 365 to 15 days prior to admission.

Long COVID Symptoms/Conditions Post Hospital Admission

- In the overall hospitalized population, the rate per 1000 person-years of any Long COVID symptom/condition was 7842 for Days 31 to 197 post hospitalization and 5239 for Days 198 to 365 post hospitalization, with a percent change between time periods of –33%
- For the overall population, the incidence of all outcomes decreased between the first and second time periods, and the rates of decrease varied by symptom/condition; those with the largest and smallest decreases over time are shown in **Figure 2**
 — Decreases in incidence over time for other symptoms/conditions included cognitive dysfunction (–29%), brain fog (–28%), arrhythmias (–26%), cerebrovascular disease (–23%), insomnia (–22%), neuropsychiatric features (–20%), chest pain (–19%), taste disturbance/dysgeusia/ageusia (–19%), diarrhea (–18%), hyperlipidemia (–17%), diabetes (–16%), cough (–15%), and obesity (–12%)

Figure 2. Rate of Long COVID Symptoms/Conditions per 1000 Person-years for Days 31 to 197 and Days 198 to 365 Post Hospital Admission Among All Hospitalized Patients (n = 92,913)

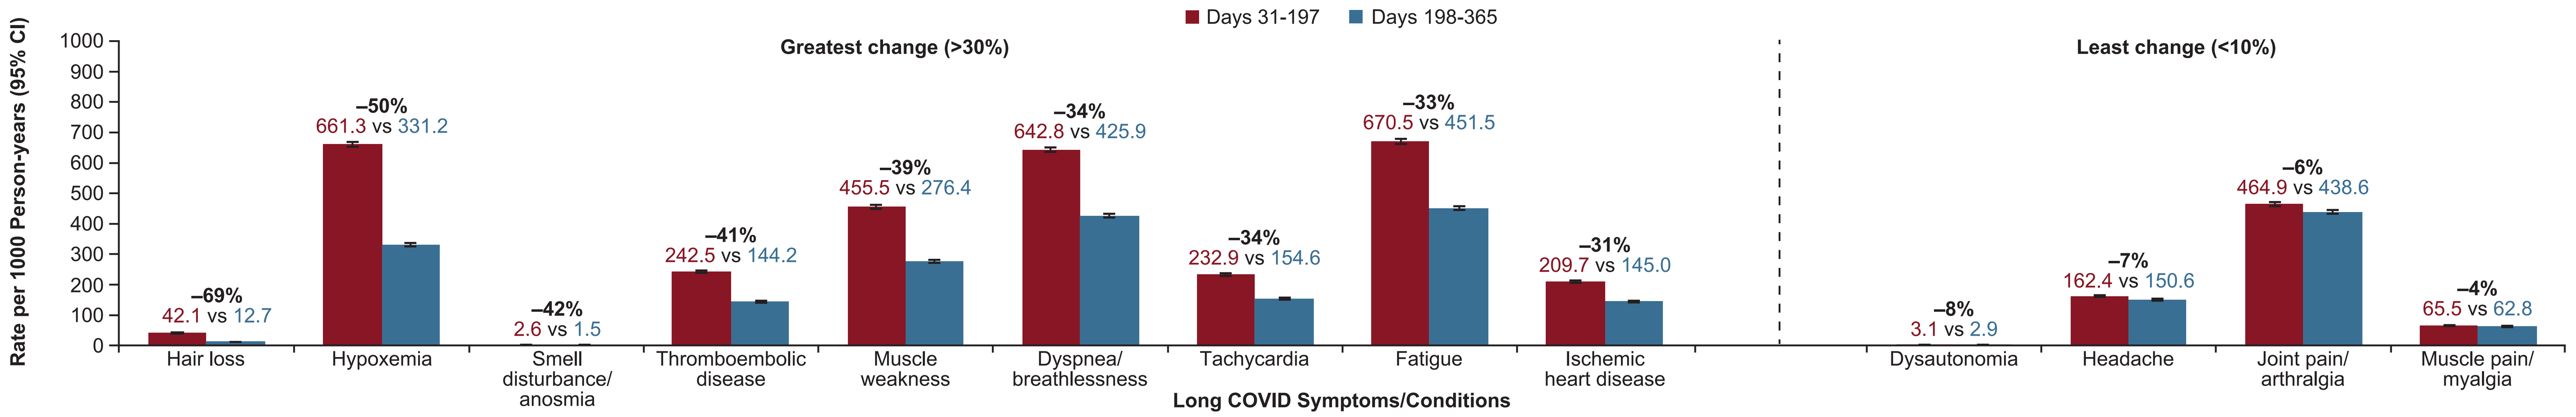

Days 31-197
Days 198-365
Greatest change (>30%)
Least change (<10%)
Rate per 1000 Person-years (95% CI)
0
100
200
300
400
500
600
700
800
900
1000
–69%
42.1 vs 12.7
Hair loss
–50%
661.3 vs 331.2
Hypoxemia
–42%
2.6 vs 1.5
Smell disturbance/ anosmia
–41%
242.5 vs 144.2
Thromboembolic disease
–39%
455.5 vs 276.4
Muscle weakness
–34%
642.8 vs 425.9
Dyspnea/ breathlessness
–34%
232.9 vs 154.6
Tachycardia
–33%
670.5 vs 451.5
Fatigue
–31%
209.7 vs 145.0
Ischemic heart disease
–8%
3.1 vs 2.9
Dysautonomia
–7%
162.4 vs 150.6
Headache
–6%
464.9 vs 438.6
Joint pain/ arthralgia
–4%
65.5 vs 62.8
Muscle pain/ myalgia
Long COVID Symptoms/Conditions

Symptoms/conditions with the greatest change over time (>30%) are shown on the left, and those with the least change over time (<10%) are shown on the right, with percent change indicated above the bars. Error bars represent 95% CI.

- Patients who had none of the symptoms/conditions at baseline (n = 12,100) had a percent change of –36% in the rate of any Long COVID symptoms/conditions over time, with a rate per 1000 person-years of 2348 for Days 31 to 197 versus 1513 for Days 198 to 365
 — Symptoms/conditions with the largest and smallest decreases over time are shown in **Figure 3**; decreases were also observed for neuropsychiatric features (–29%), chest pain (–27%), obesity (–24%), diabetes (–23%), diarrhea (–19%), cough (–18%), hyperlipidemia (–18%), and muscle pain/myalgia (–12%)

Figure 3. Rate of Long COVID Symptoms/Conditions per 1000 Person-years for Days 31 to 197 and Days 198 to 365 Post Hospital Admission Among Hospitalized Patients With None of the Symptoms/Conditions at Baseline (n = 12,100)

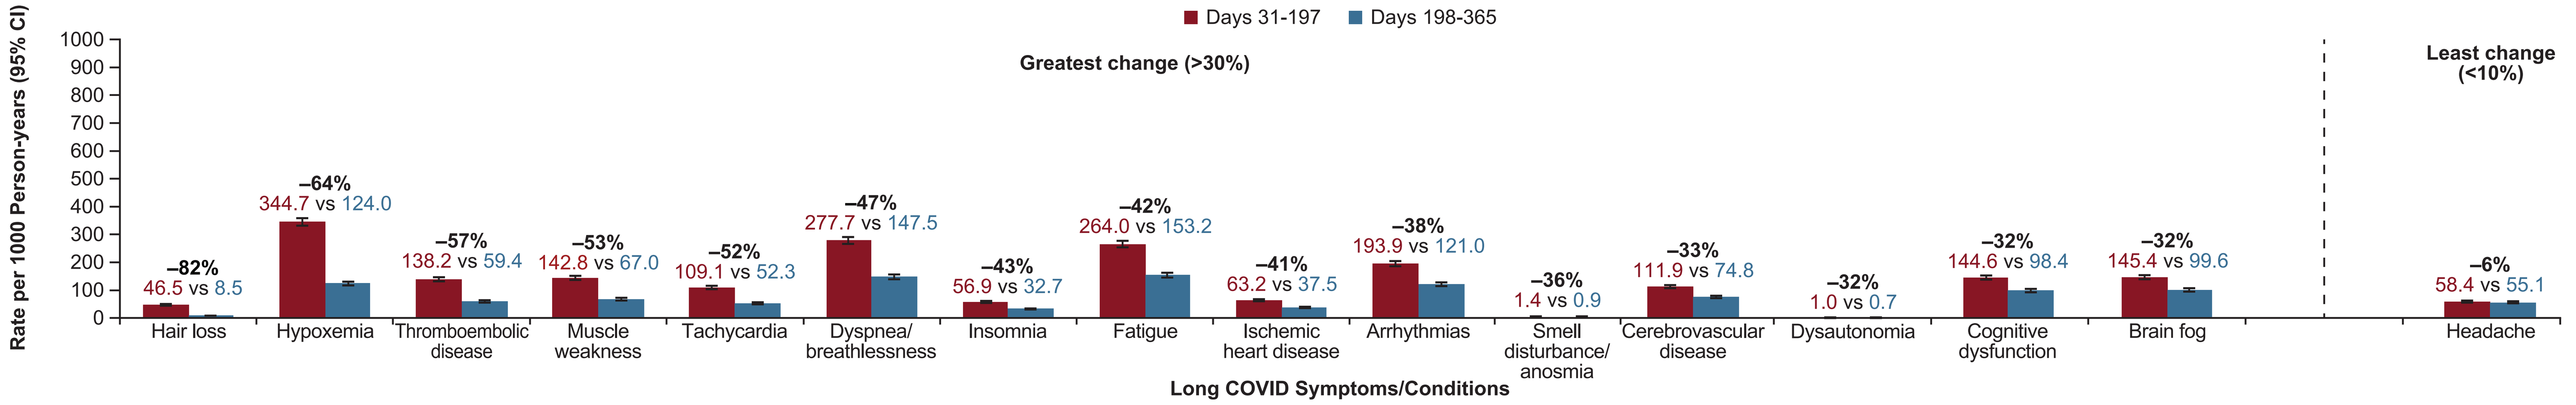

Days 31-197
Days 198-365
Greatest change (>30%)
Least change (<10%)
Rate per 1000 Person-years (95% CI)
0
100
200
300
400
500
600
700
800
900
1000
–82%
46.5 vs 8.5
Hair loss
–64%
344.7 vs 124.0
Hypoxemia
–57%
138.2 vs 59.4
Thromboembolic disease
–53%
142.8 vs 67.0
Muscle weakness
–52%
109.1 vs 52.3
Tachycardia
–47%
277.7 vs 147.5
Dyspnea/ breathlessness
–43%
56.9 vs 32.7
Insomnia
–42%
264.0 vs 153.2
Fatigue
–41%
63.2 vs 37.5
Ischemic heart disease
–38%
193.9 vs 121.0
Arrhythmias
–36%
1.4 vs 0.9
Smell disturbance/ anosmia
–33%
111.9 vs 74.8
Cerebrovascular disease
–32%
1.0 vs 0.7
Dysautonomia
–32%
144.6 vs 98.4
Cognitive dysfunction
–32%
145.4 vs 99.6
Brain fog
–6%
58.4 vs 55.1
Headache
Long COVID Symptoms/Conditions

Symptoms/conditions with the greatest change over time (>30%) are shown on the left, and those with the least change over time (<10%) are shown on the right, with percent change indicated above the bars. Error bars represent 95% CI.

- For patients with immunocompromising conditions (n = 39,185), the rate per 1000 person-years of any Long COVID symptoms/conditions was 11,543 for Days 31 to 197 and 7356 for Days 198 to 365; rates were higher compared to those of the overall population but had a similar percent change (–36%) over time
 — Symptoms/conditions with the largest and smallest decreases over time are shown in **Figure 4**; decreases were also observed for ischemic heart disease (–29%), arrhythmia (–25%), taste disturbance/dysgeusia/ageusia (–25%), cerebrovascular disease (–23%), insomnia (–22%), neuropsychiatric features (–21%), diarrhea (–20%), chest pain (–20%), hyperlipidemia (–19%), cough (–15%), diabetes (–15%), and obesity (–10%)

Figure 4. Rate of Long COVID Symptoms/Conditions per 1000 Person-years for Days 31 to 197 and Days 198 to 365 Post Hospital Admission Among Hospitalized Patients With Immunocompromising Conditions (n = 39,185)

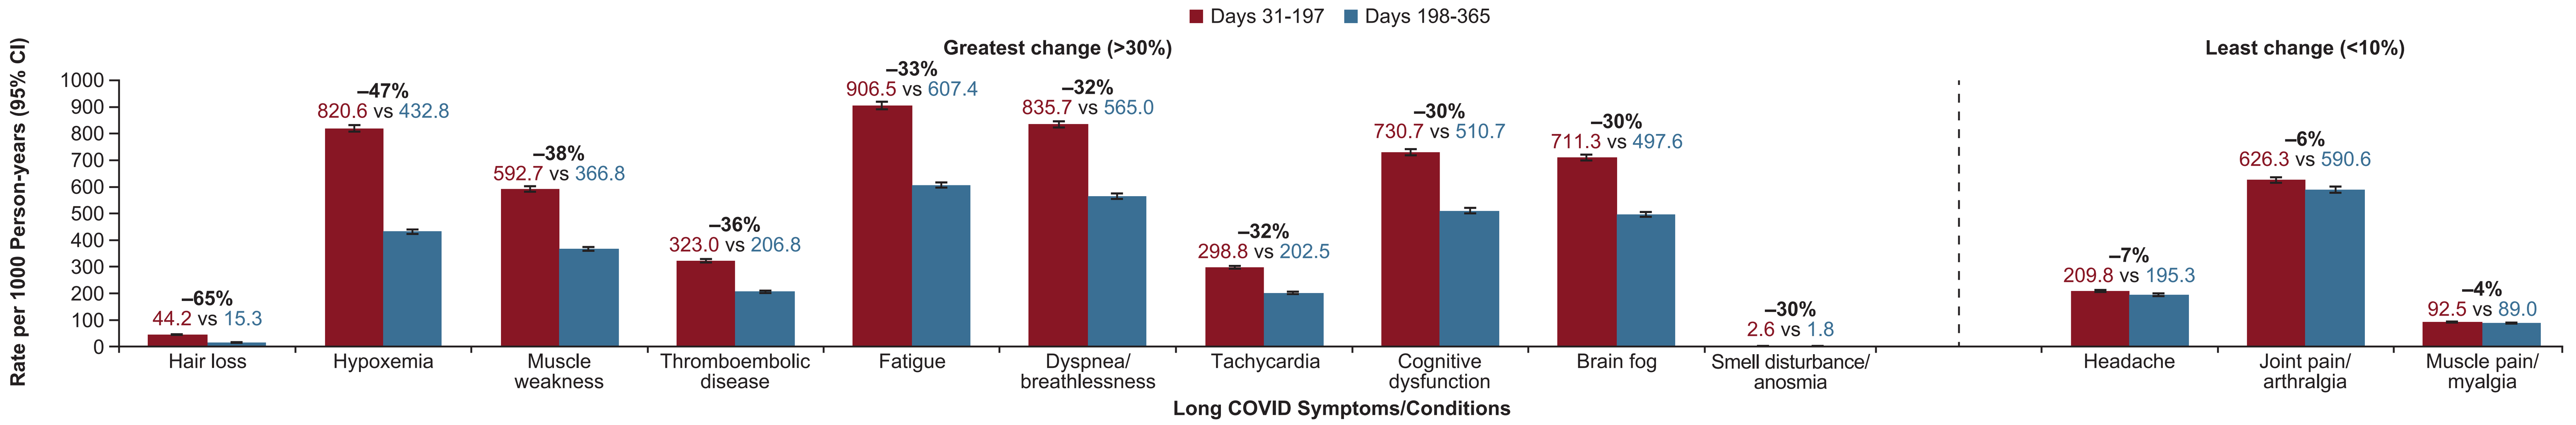

Days 31-197
Days 198-365
Greatest change (>30%)
Least change (<10%)
Rate per 1000 Person-years (95% CI)
0
100
200
300
400
500
600
700
800
900
1000
–65%
44.2 vs 15.3
Hair loss
–47%
820.6 vs 432.8
Hypoxemia
–38%
592.7 vs 366.8
Muscle weakness
–36%
323.0 vs 206.8
Thromboembolic disease
–33%
906.5 vs 607.4
Fatigue
–32%
835.7 vs 565.0
Dyspnea/ breathlessness
–32%
298.8 vs 202.5
Tachycardia
–30%
730.7 vs 510.7
Cognitive dysfunction
–30%
711.3 vs 497.6
Brain fog
–30%
2.6 vs 1.8
Smell disturbance/ anosmia
–7%
209.8 vs 195.3
Headache
–6%
626.3 vs 590.6
Joint pain/ arthralgia
–4%
92.5 vs 89.0
Muscle pain/ myalgia
Long COVID Symptoms/Conditions

Symptoms/conditions with the greatest change over time (>30%) are shown on the left, and those with the least change over time (<10%) are shown on the right, with percent change indicated above the bars. Error bars represent 95% CI.

- A few symptoms/conditions had a nominal increase over time, although this may not be clinically meaningful due to small sample sizes, including taste disturbance/dysgeusia/ageusia (+127% [95% CI, 127-127]) and joint pain/arthralgia (+1% [95% CI, 0-3]) for patients who had none of the symptoms/conditions at baseline and dysautonomia (+11% [95% CI, 10-11]) for patients with immunocompromising conditions

References: **1.** Burn E, et al. *Nat Commun*. 2021;12:777. **2.** Davis HE, et al. *Nat Rev Microbiol*. 2023;21:133-46. **3.** O'Mahoney LL, et al. *EClinicalMedicine*. 2023;55:101762. **4.** Soriano JB, et al. *Lancet Infect Dis*. 2022;22:e102-7. **5.** Bowe B, et al. *Nat Med*. 2023;29:2347-57. **6.** Patel M, et al. *Emerg Infect Dis*. 2020;26:1720-30.

Acknowledgments: This study was funded by Gilead Sciences, Inc. Medical writing and editorial support were provided by Katherine Townsend, PhD, of Lumanity Communications Inc., and were funded by Gilead Sciences, Inc.

Disclosures: **MB**, **VS**, **GB**, and **AC** are employees of and may own stock or stock options in Gilead Sciences, Inc. **AK** and **RS** are employees of Aetion, Inc.

IDWeek; October 16-19, 2024; Los Angeles, CA, USA